A phase 1/2 study evaluating the safety and efficacy of autologous TAC T cells in subjects with claudin 18.2+ advanced solid tumors

Ecaterina E. Dumbrava[1], Syma Iqbal[2], Simon Turcotte[3], Gregory Botta[4], Benjamin Schlechter[5], Geoffrey Ku[6], Peter Hosein[7], Sam Saibil[8], Miriam Gavriliuc[1], Maria Apostolopoulou[9], Mobolaji Giwa[9], Heather MacGregor[9], Kara Moss[9], Eli Panna[9], Swaminathan Murugappan[9], Davendra Sohal[10]

[1] The University of Texas MD Anderson Cancer Center, Houston, Texas, USA; [2] University of Southern California Health Sciences, Los Angeles, California, USA; [3] Centre Hospitalier de l'Université de Montréal (CHUM), Montreal, Quebec, Canada; [4] University of California, San Diego Health, San Diego, California, USA; [5] Dana Farber Cancer Institute, Boston, Massachusetts, USA; [6] Memorial Sloan Kettering Cancer Center, New York, New York, USA; [7] University of Miami, Coral Gables, Florida, USA; [8] Princess Margaret/University Health Network, Toronto, Ontario, CA; [9] Triumvira Immunologics, Inc., Austin, Texas, USA; [10] University of Cincinnati, Cincinnati, Ohio, USA

INTRODUCTION

- The T cell antigen coupler (TAC) is a novel, proprietary chimeric receptor that facilitates the re-direction of T cells to tumor cells and activates T cells by co-opting the endogenous T cell receptor complex, with the goal to elicit a safe and durable anti-tumor response. In preclinical models, TAC-engineered T cells effectively eradicate tumor cells *in vitro* and *in vivo* without toxicities typically associated with engineered T cell products. TAC01-CLDN18.2 is an autologous T cell product comprising T cells expressing the CLDN18.2 TAC, which specifically recognize CLDN18.2+ cells.
- TACTIC-3 (NCT05862324) is an open-label, multicenter phase I/II study that aims to establish safety, maximum tolerated dose (MTD), recommended phase 2 dose (RP2D), pharmacokinetic profile, and efficacy of TAC01-CLDN18.2 in patients with CLDN18.2 positive solid tumors by immunohistochemistry (i.e. gastric, GEJ, esophageal adenocarcinoma, PDAC, colorectal cancer, cholangiocarcinoma, ovarian mucinous cancer, gallbladder cancer and NSCLC) who have been exposed to at least 2 prior anti-cancer therapies.

TAC SCIENCE

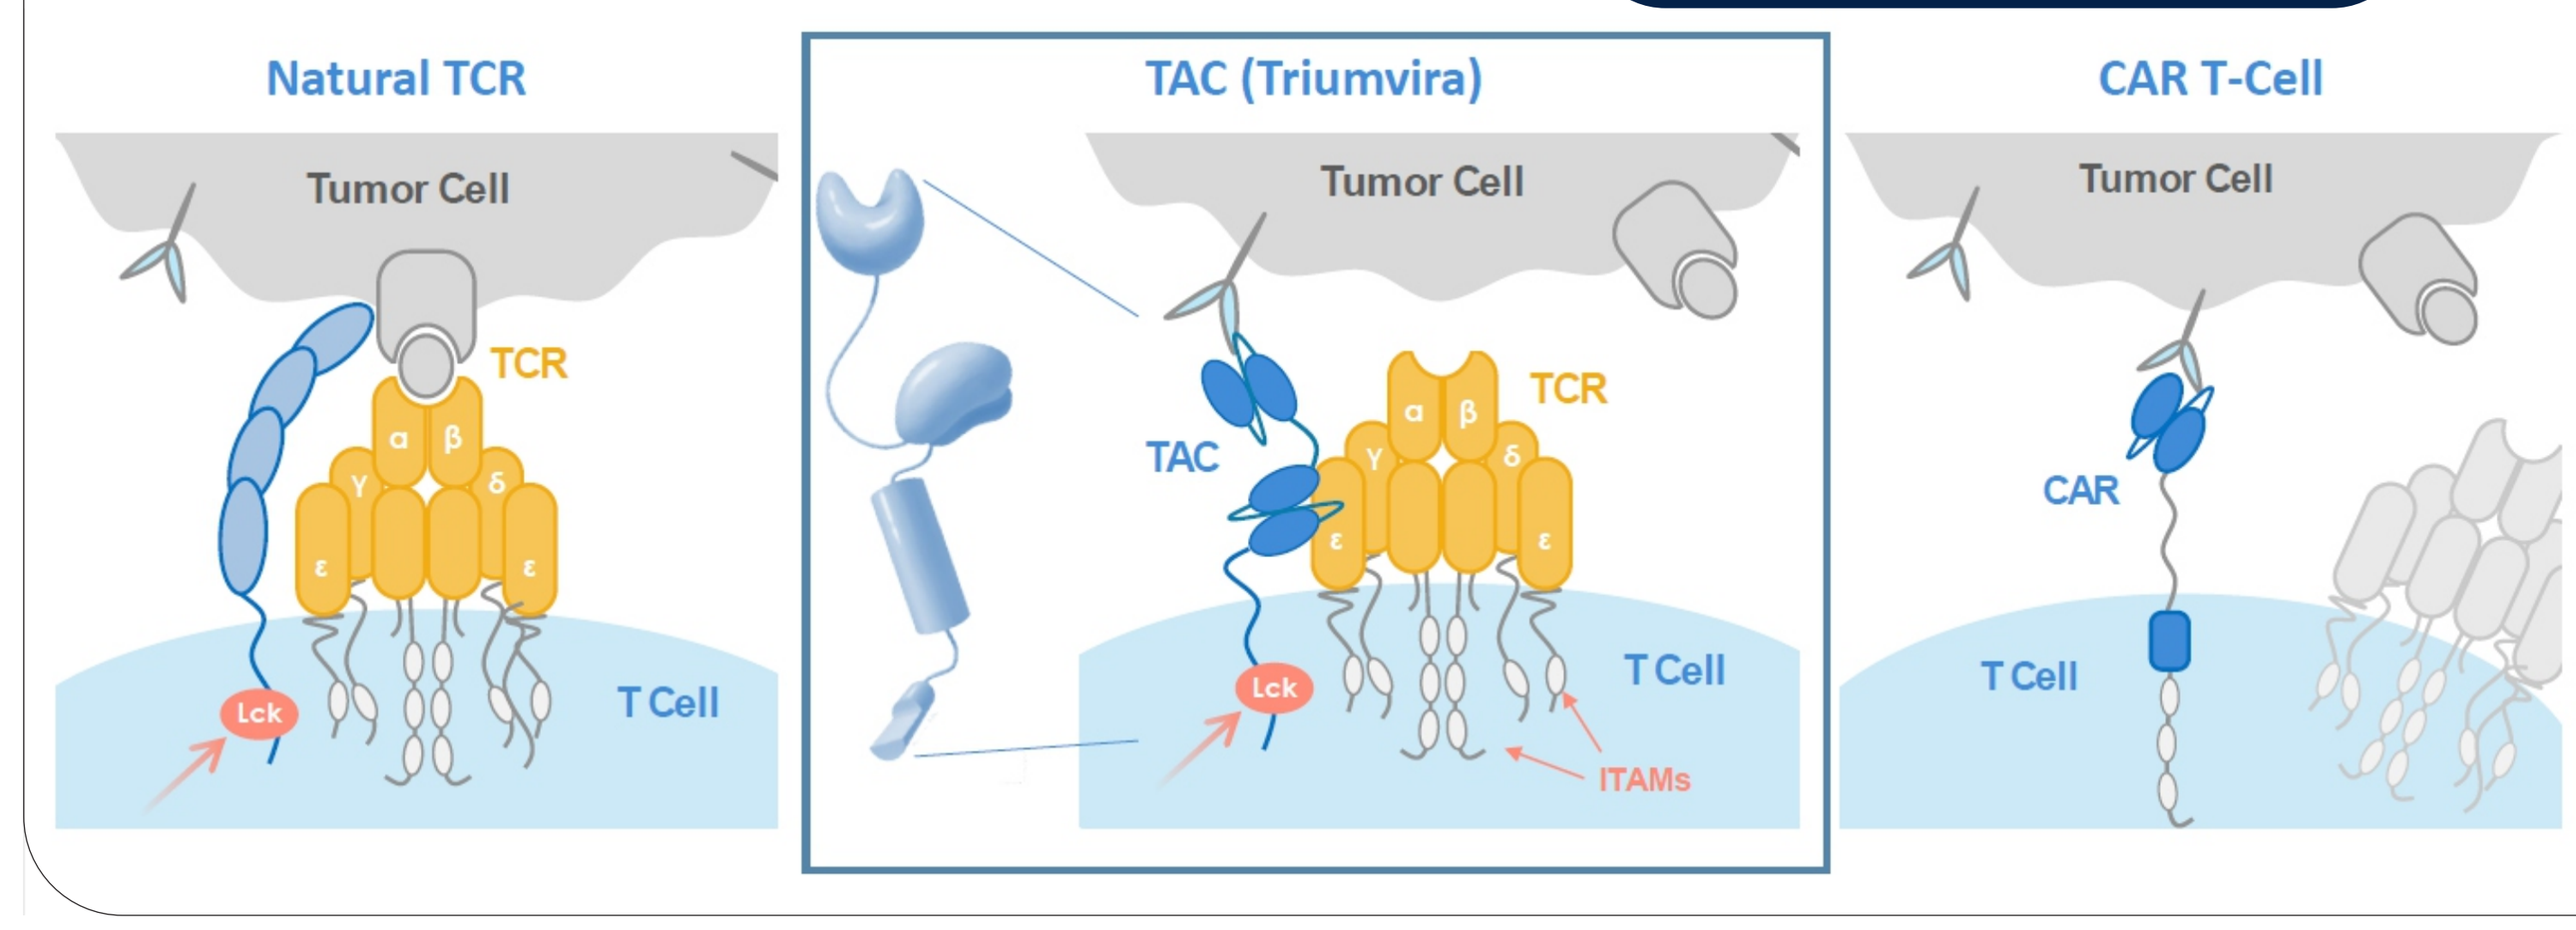
Natural TCR
Tumor Cell
TCR
T Cell
Lck
TAC (Triumvira)
Tumor Cell
TCR
TAC
T Cell
Lck
ITAMs
CAR T-Cell
Tumor Cell
CAR
T Cell

TAC achieves TCR activation via a CD3 binding domain while tightly binding the target of interest, CLDN18.2. TAC thus co-opts natural TCR function and provides intracellular co-receptor signaling via Lymphocyte-specific Protein Tyrosine Kinase (LCK), mimicking physiologic TCR activation.

TRIAL DESIGN

Phase I (Dose Escalation)

The classic 3+3 dose escalation design will be employed to efficiently determine the maximum tolerated dose (MTD) and RP2D using well-defined DLT criteria.

N = 9-24 subjects

Phase II (Dose Expansion)

Groups A and C:
Approach: Simon 2-stage design.
Objective: Assess efficacy (ORR).

Group B:
Initial Approach: Evaluate ORR after 10 PDAC treatments.
Future: Adopt Simon 2-stage based on outcomes.

DL (-1)	DL 1	DL 2	DL 3	Group A	Group B	Group C
0-6 Subjects	3-6 Subjects	3-6 Subjects	3-6 Subjects	(Gastric and Esophageal AC)	(PDAC)	(Ovarian and NSCLC cancers)
1-3 x 10^5 Cells/kg	6-8 x 10^5 Cells/kg	1-3 x 10^6 Cells/kg	6-8 x 10^6 Cells/kg	Planned Enrollment: ≤ 57 Subjects	Planned Enrollment ≤10 Subjects	Planned Enrollment: ≤22 Subjects

In both phases, a second dose is allowed if subjects meet predefined clinical and safety criteria, with modified or no lymphodepleting chemotherapy (LDC).

Preferred LDC:
3 consecutive days of fludarabine (Flu) IV (30 mg/m^2) and cyclophosphamide (Cy) IV (300 mg/m^2) +/- Mesna IV, and a single dose of nab-paclitaxel (100mg/m^2) on the second day of LDC.

PHASE I TRIAL PROGRESS

Primary Endpoints
Safety: DLTs, AEs

Secondary Endpoints
RP2D, PK, Efficacy (ORR, DoR, OS, TCR, PFS)

DL1 and 2 have been completed. DL3 has been deemed safe by the DSMC and has been expanded to 7 subjects.

Eligibility Criteria
Patients with advanced, metastatic, unresectable solid tumors which express CLDN18.2 after at least 2 lines of therapy (1 for PDAC), at least 1 measurable lesion per RECIST version 1.1, ECOG performance score 0-1, grade 1 or baseline for any prior treatment related toxicities.

Note: Required specific tumor types for phase 1 include: gastric, GEJ, esophageal adenocarcinoma, PDAC, colorectal cancer, cholangiocarcinoma, ovarian mucinous cancer, gallbladder cancer and NSCLC.

Demographics and Tumor Intrinsic Characteristics (n=13)

Sex: Male/Female, n (%)	M	9 (69.2)	CLDN18.2 expression, n (%)	High	10 (76.9)
	F	4 (30.8)		Low	3 (23.1)
Race, n (%)	Hispanic	5 (38.5)	Age, Median (Range)		61 (43-75)
	White	4 (30.8)	Previous Anti-Cancer Therapy, Median (Range)		2.5 (2-8)
	Asian	3 (23.1)	Tumor Type, n (%)	Pancreatic	6 (46.2)
	Not Reported	1 (7.7)		Gastric	3 (23.1)
ECOG PS, n (%)	0	3 (38.5)		Esophageal	2 (15.4)
	1	8 (61.5)		Colorectal	1 (7.7)
				GEJ	1 (7.7)

High (H) CLDN18.2 expression subjects in this study are those with samples where 50% or more of tumor cells express CLDN18.2 with intensity of 2+ or 3+. All others are termed low (L) expression.

PHASE I CLINICAL SAFETY DATA

TAC01-CLDN18.2-Related Adverse Events by Preferred Terms and Grade

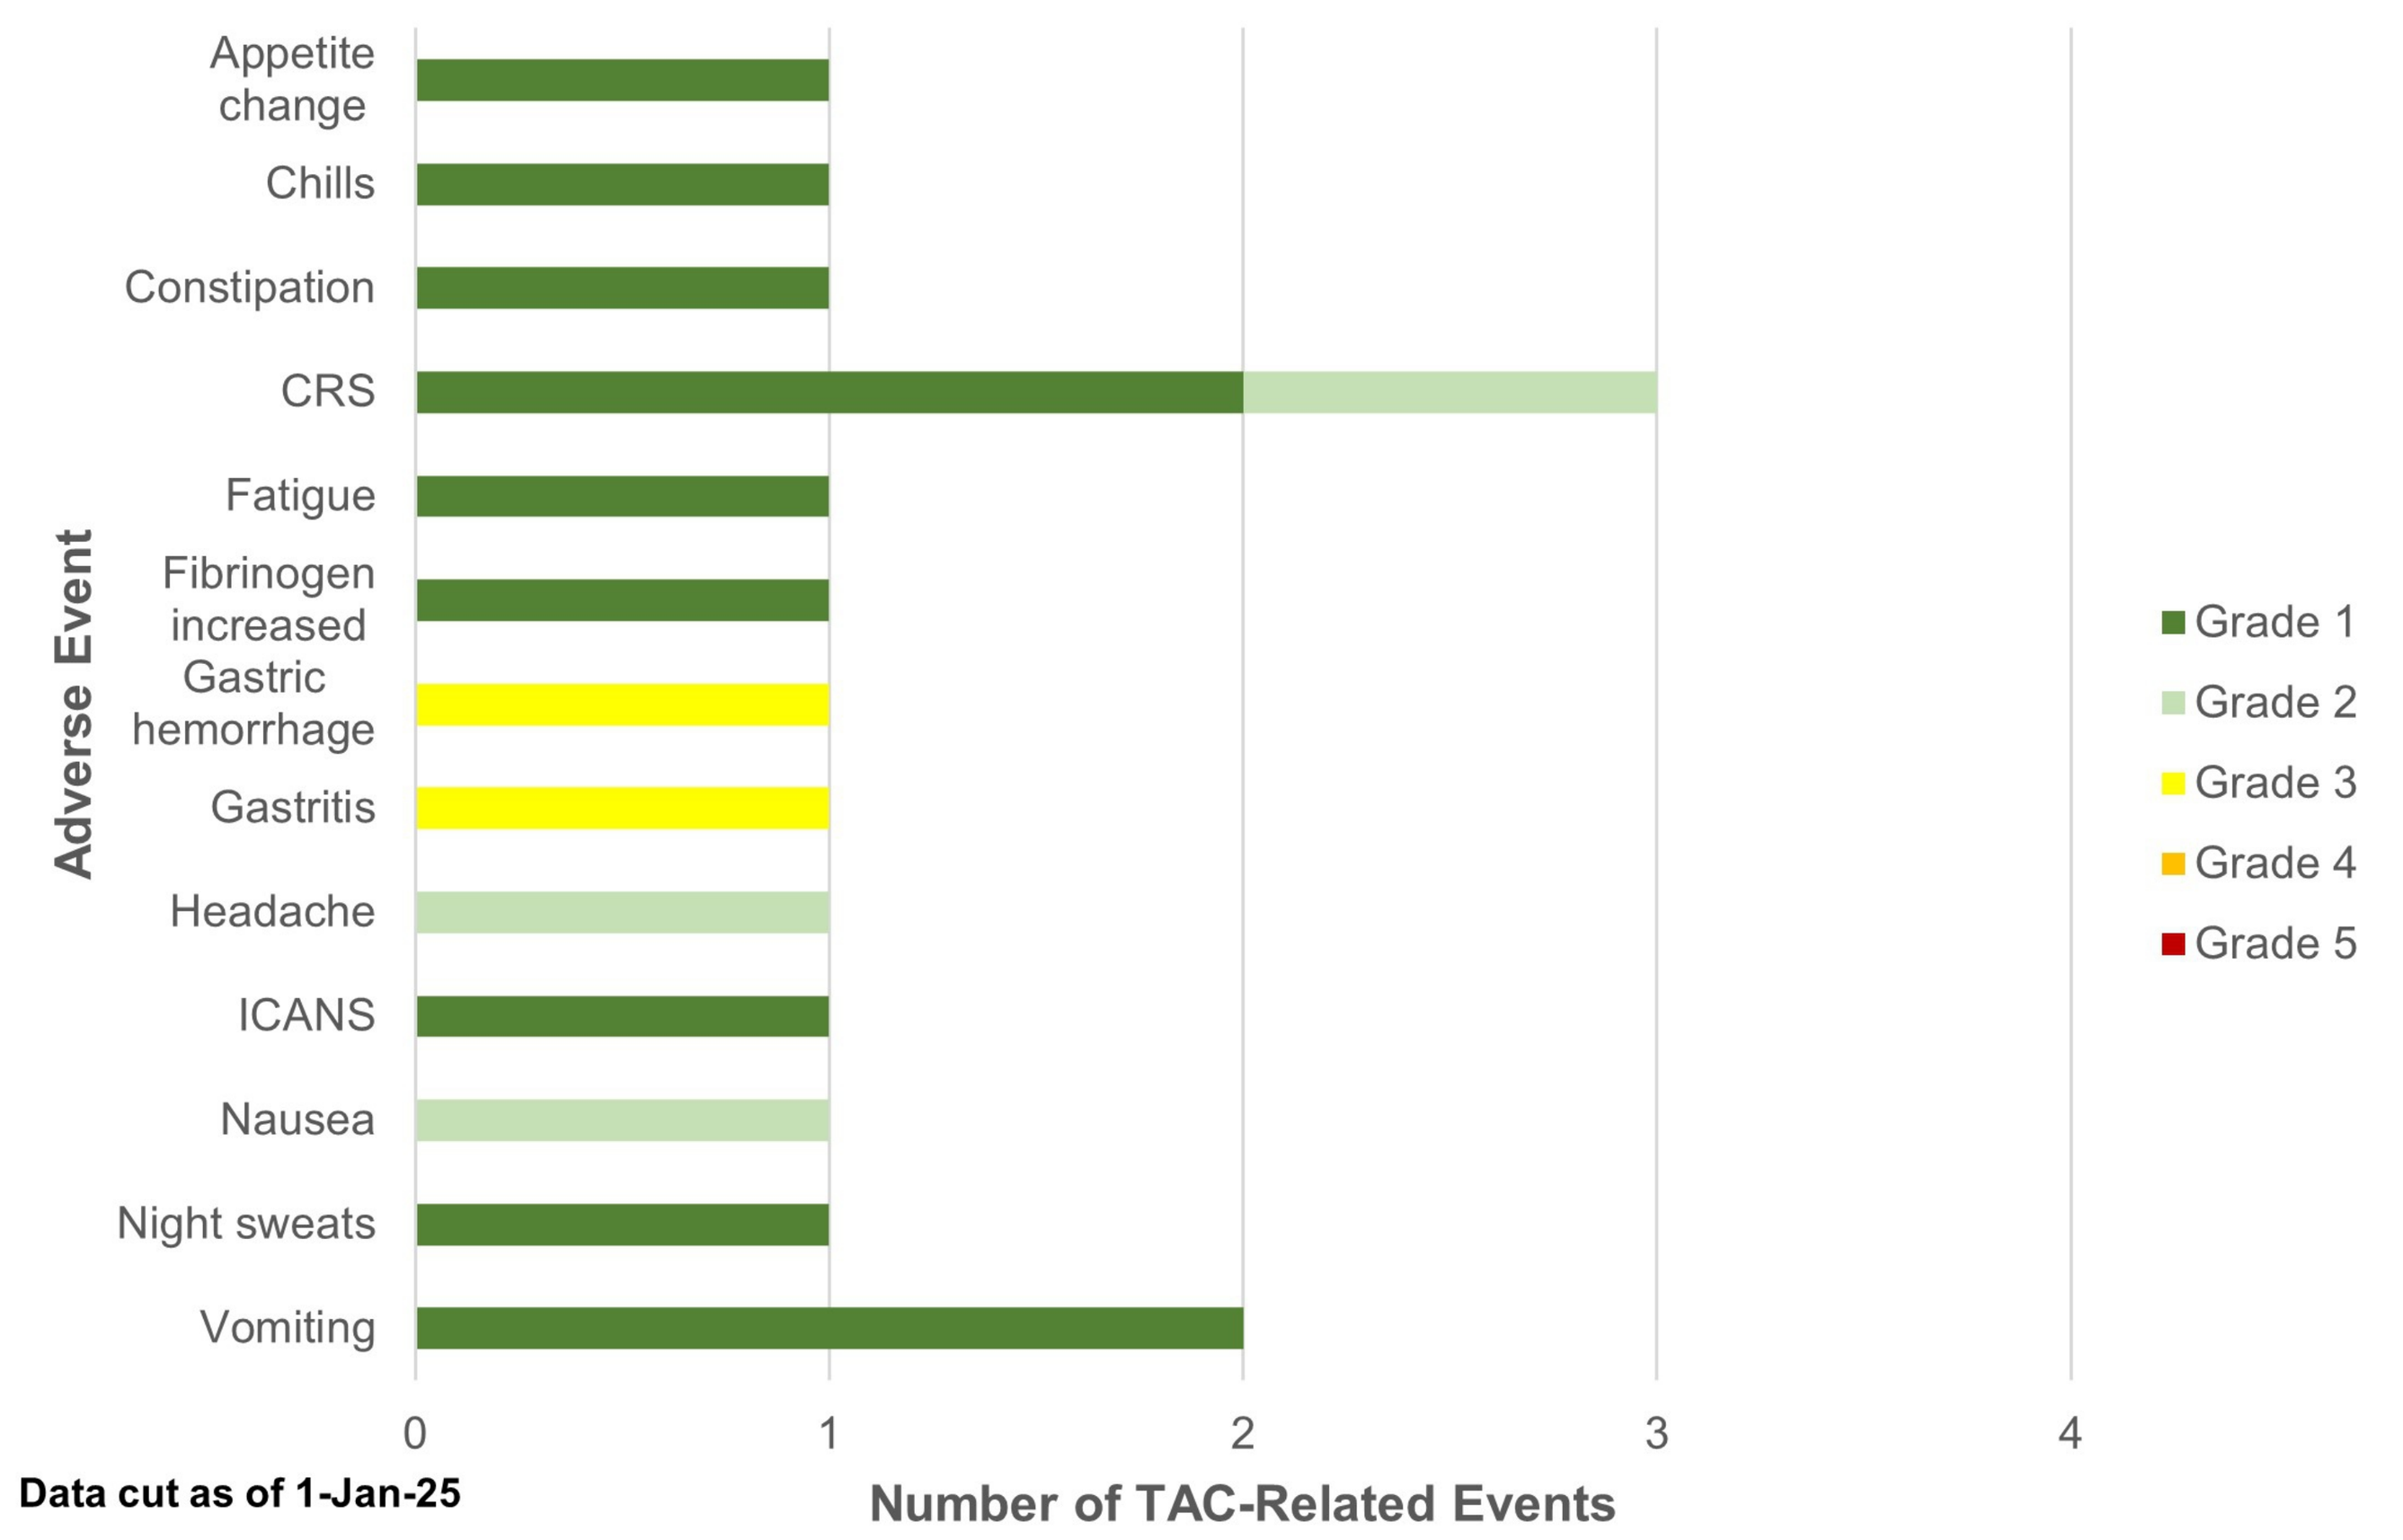
Appetite change
Chills
Constipation
CRS
Fatigue
Fibrinogen increased
Gastric hemorrhage
Gastritis
Headache
ICANS
Nausea
Night sweats
Vomiting
Adverse Event
0
1
2
3
4
Number of TAC-Related Events
Data cut as of 1-Jan-25
Grade 1
Grade 2
Grade 3
Grade 4
Grade 5

Safety Summary

- One subject in Dose Level 2 (DL 2) experienced a Grade 1 ICAN (Immune-effector Cell Associated Neurotoxicity) event that resolved within 24 hours without intervention.
- Three subjects in DL 3 experienced Grade 1 or 2 CRS (Cytokine Release Syndrome) that resolved with standard interventions.
- One subject experienced Grade 3 gastritis and gastric hemorrhage, both resolving within 4 days.

BEST RESPONSE (tumor measurements)

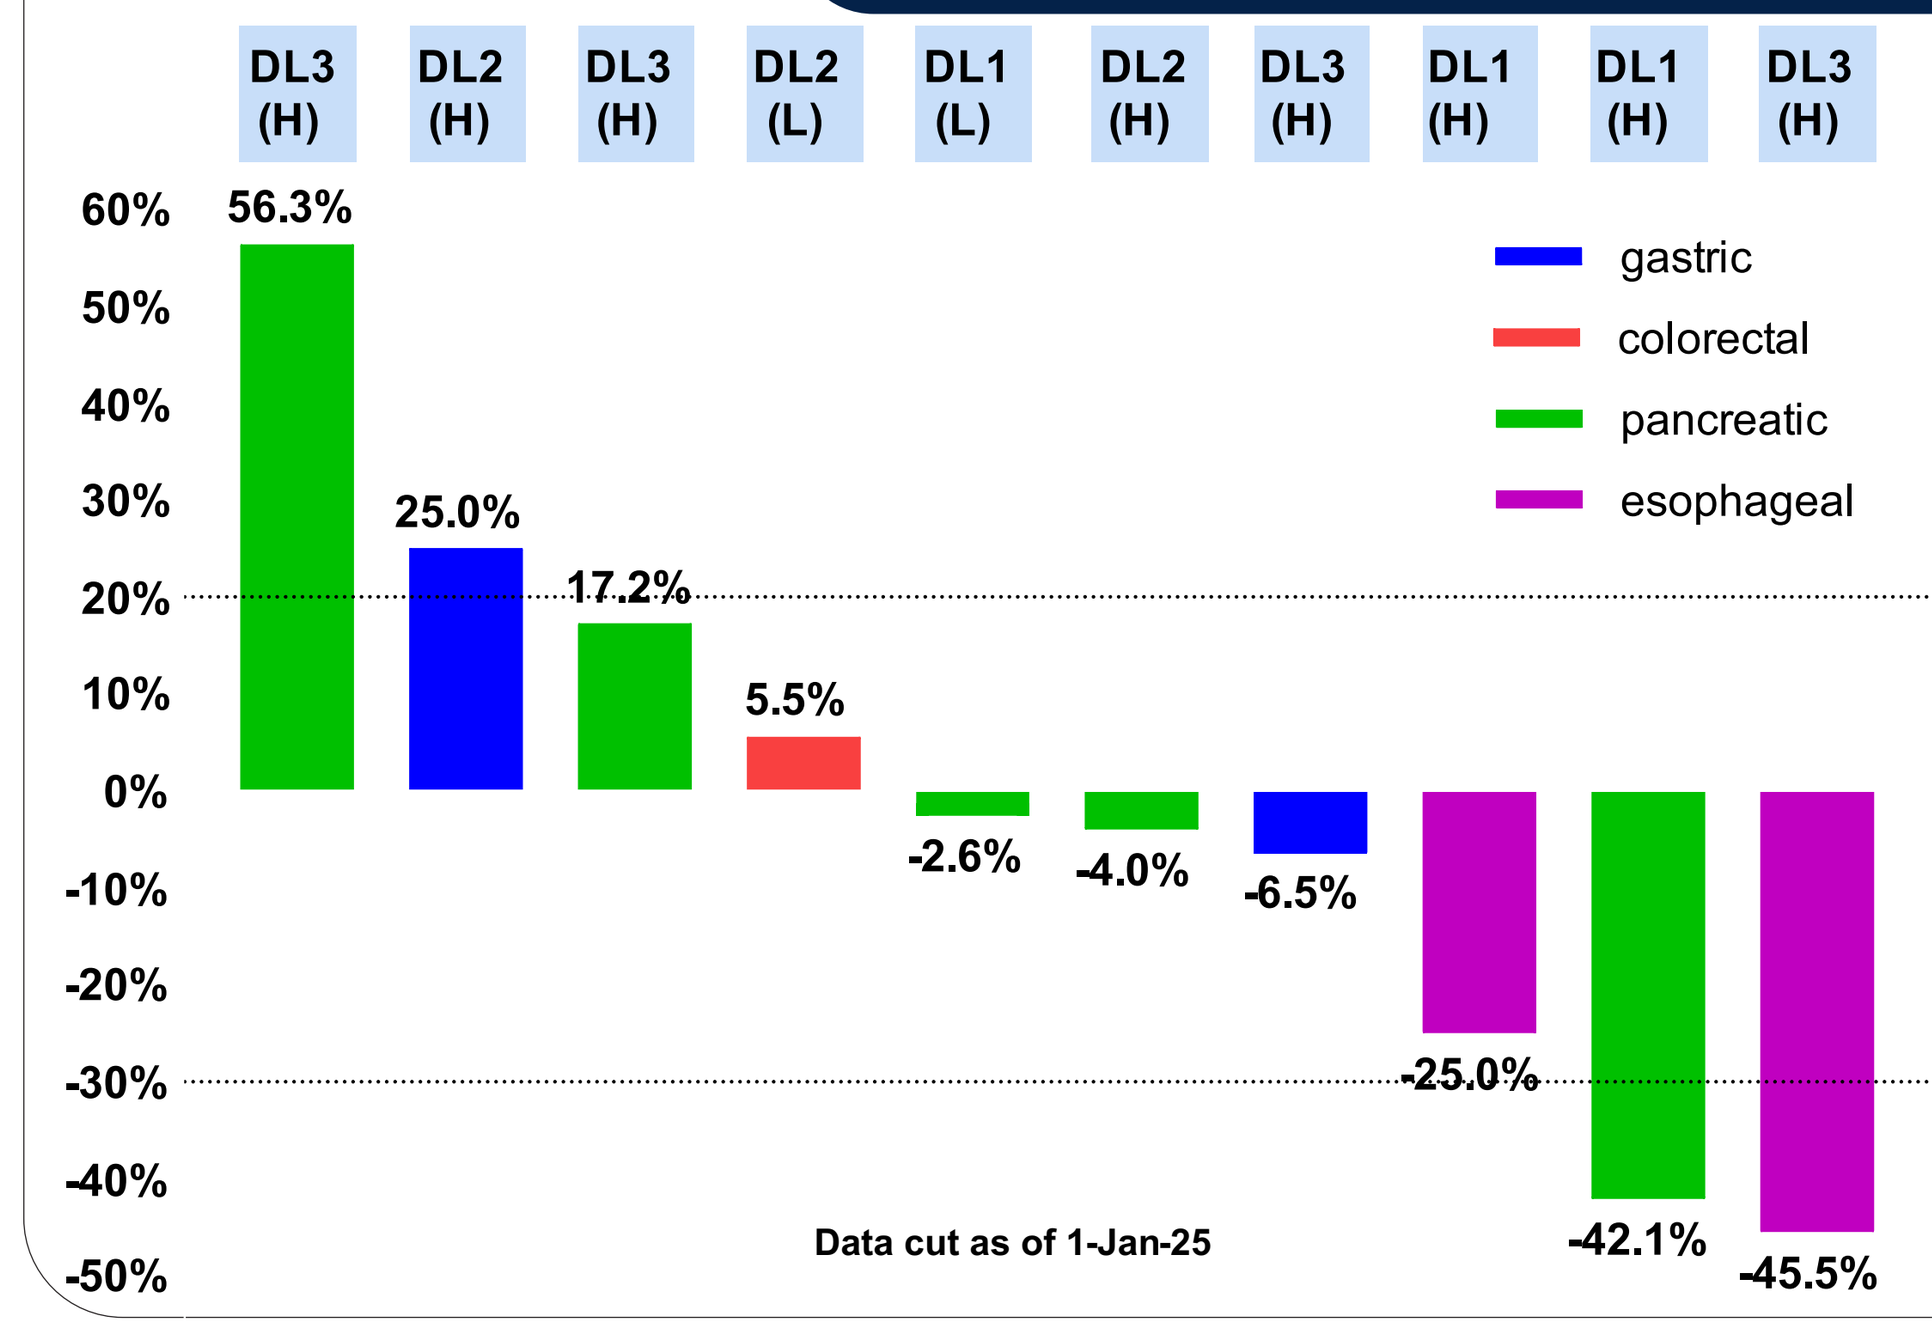
DL3 (H)
DL2 (H)
DL3 (H)
DL2 (L)
DL1 (L)
DL2 (H)
DL3 (H)
DL1 (H)
DL1 (H)
DL3 (H)
56.3%
25.0%
17.2%
5.5%
-2.6%
-4.0%
-6.5%
-25.0%
-42.1%
-45.5%
gastric
colorectal
pancreatic
esophageal
Data cut as of 1-Jan-25

Ten subjects have efficacy assessments available:

- **DL1:** One subject with Partial Response (PR) and 2 subjects with Stable Disease (SD).
- **DL2:** Two subjects with SD and one subject with Progressive Disease (PD).
- **DL3:** One subject with PR, one subject with SD and two subjects with PD.

Disease Control Rate (DCR) of 70% and Objective Response Rate (ORR) of 20% in heavily pre-treated subjects with aggressive malignancies.

SUBJECT RESPONSE AND FOLLOW-UP

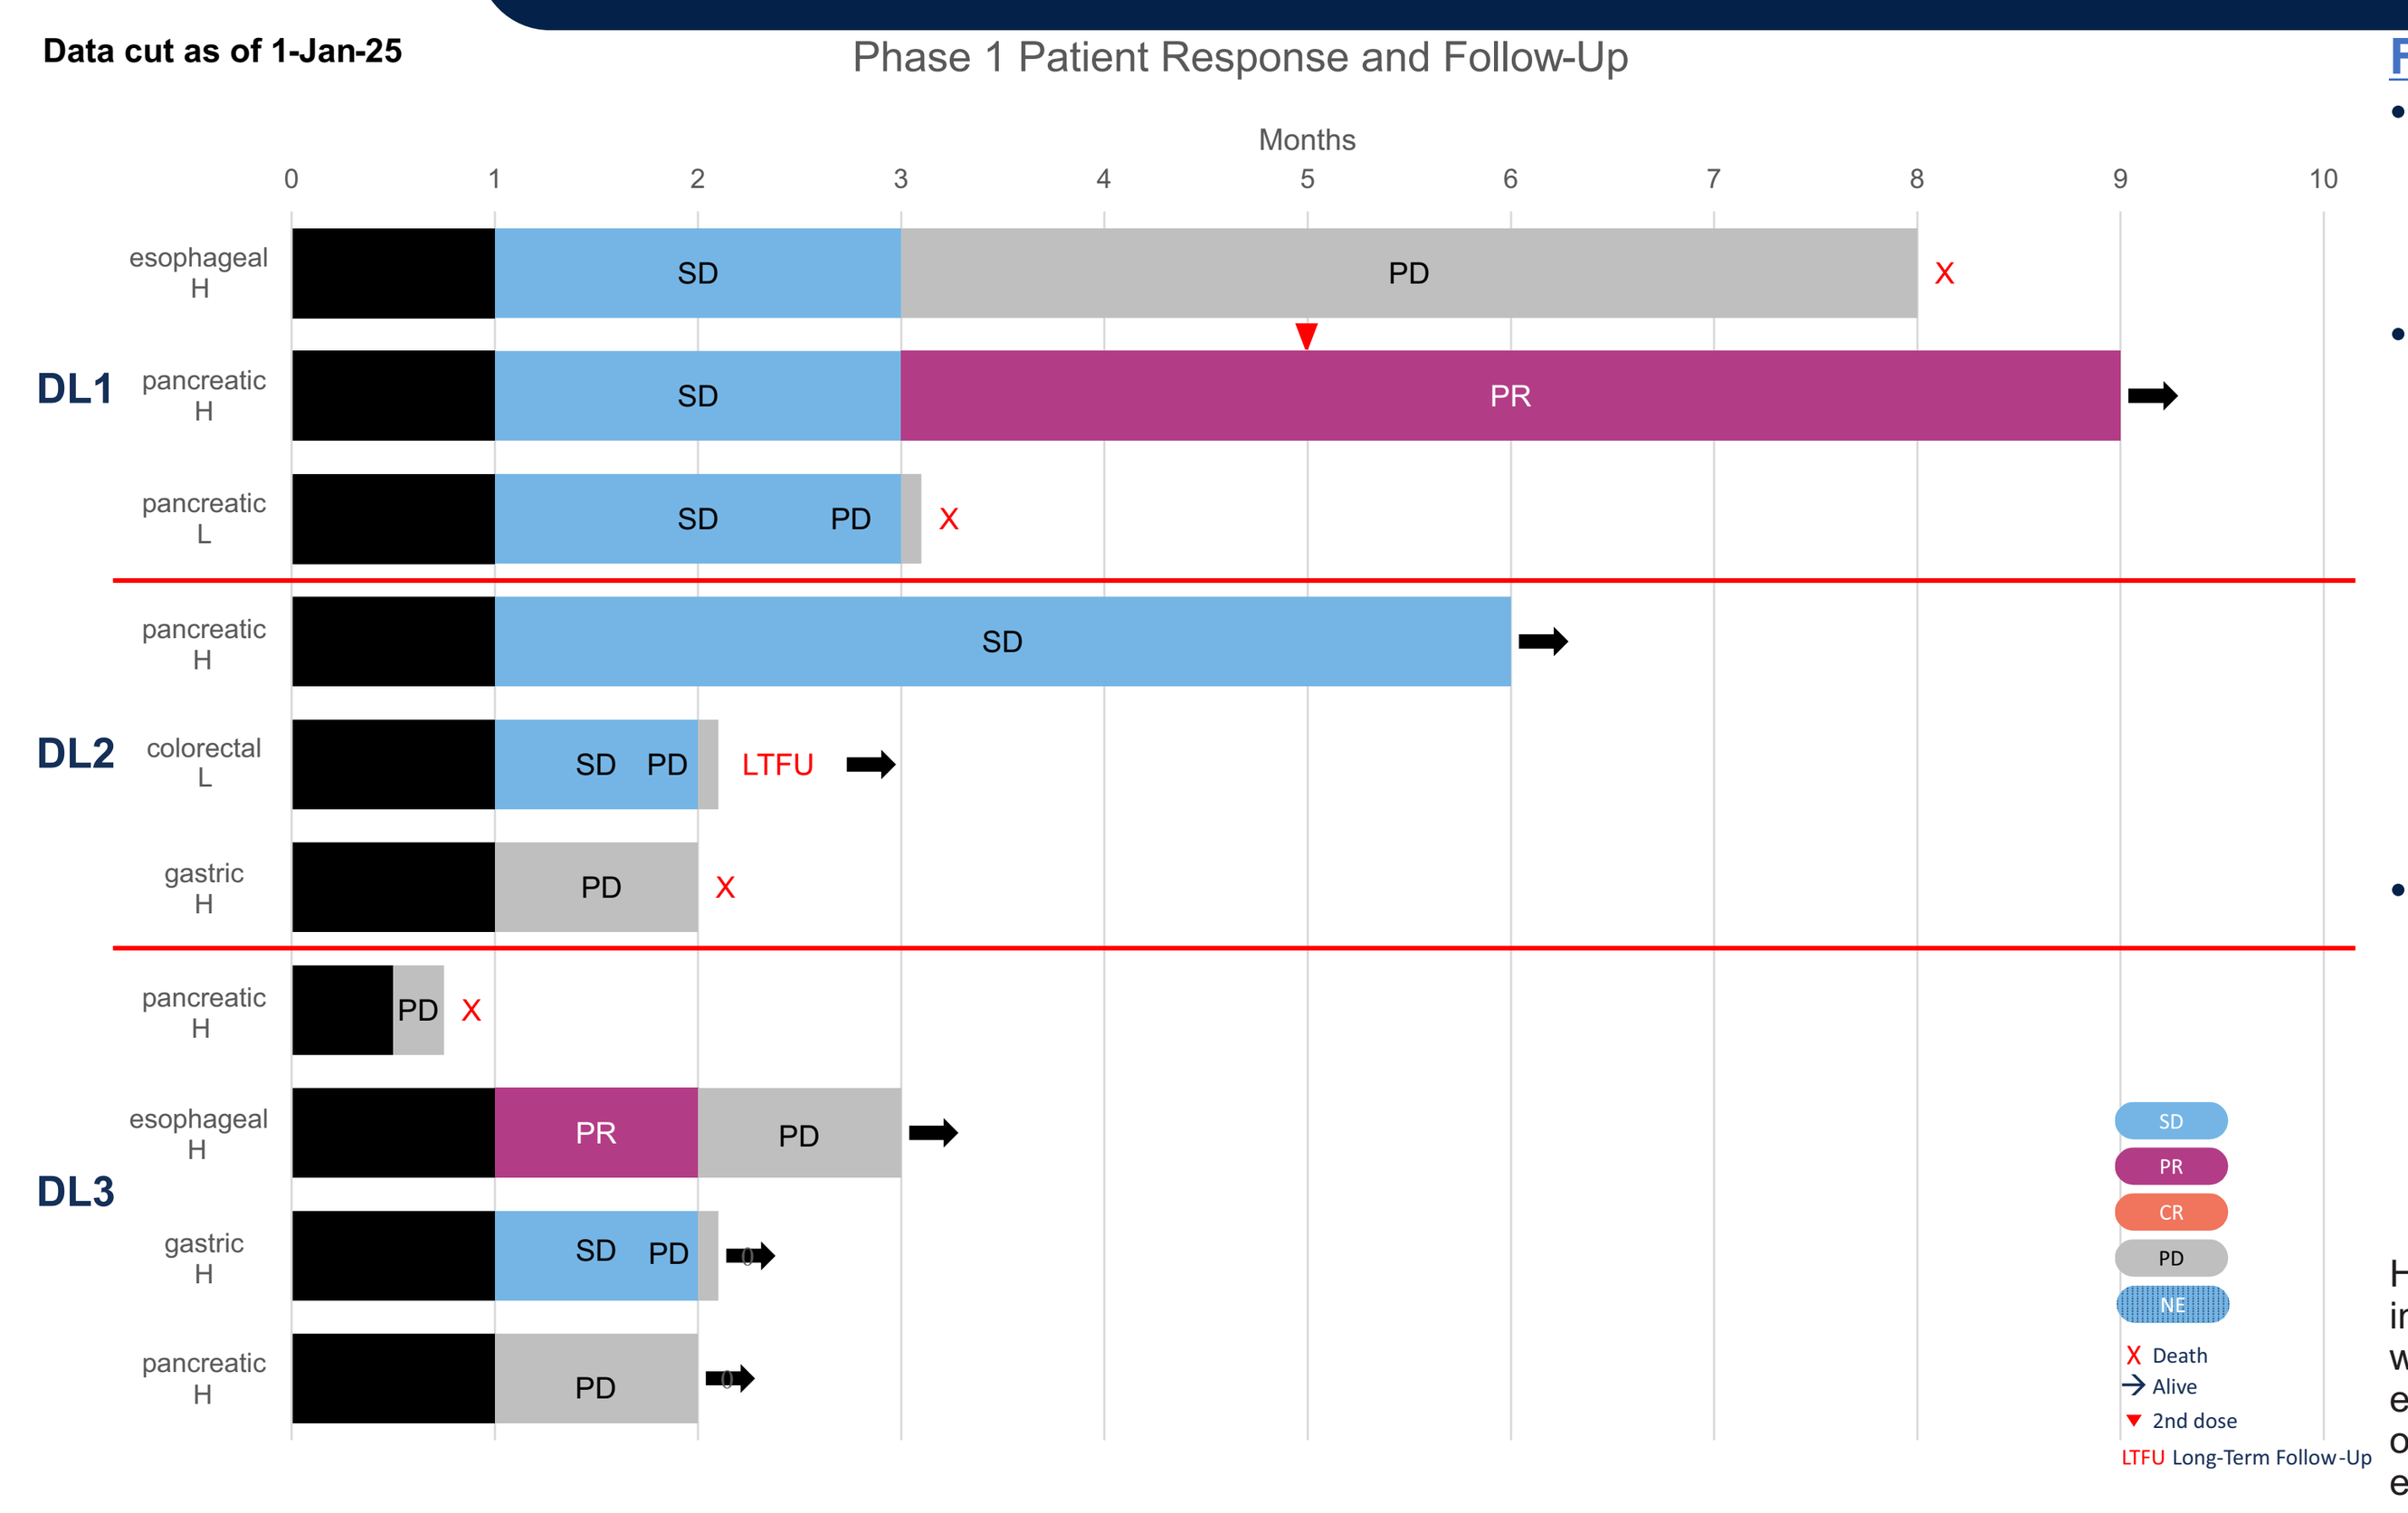
Data cut as of 1-Jan-25
Phase 1 Patient Response and Follow-Up
Months
DL1
DL2
DL3
esophageal H
pancreatic H
pancreatic L
pancreatic H
colorectal L
gastric H
pancreatic H
esophageal H
gastric H
pancreatic H
SD
PD
PR
LTFU
SD
PR
CR
PD
NE
Death
Alive
2nd dose
LTFU Long-Term Follow-Up

Follow-up

- All subjects are still in follow-up per protocol, except for four subjects who died due to progressive disease.
- One subject in DL1 with heavily pre-treated pancreatic adenocarcinoma (3 prior lines of therapy) and high CLDN18.2 expression has an ongoing, confirmed PR, lasting 6 months at the date of the data cut. This subject received a second dose.
- One subject in DL 3 with heavily pre-treated esophageal adenocarcinoma (2 prior lines of therapy) and high CLDN18.2 expression had an unconfirmed PR.

High (H) CLDN18.2 expression subjects in this study are those with samples where 50% or more of tumor cells express CLDN18.2 with intensity of 2+ or 3+. All others are termed low (L) expression.

TUMOR ASSESSMENT: SUBJECT RESPONSE

Subject 0112-0040

- 57 year old female with CLDN18.2+ (high expression) stage IV metastatic pancreatic cancer.
- Previously treted with 3 lines of therapy.
- The subject did not receive bridging therapy.
- The subject had stable disease at first tumor assessment, which turned to a partial response 3 months after TAC infusion. The partial response has been confirmed and is ongoing (for 6 months as of data cut date). The subject received a second TAC infusion 5 months after the first and is still on treatment (for 9 months as of data cut date). Tthe subject's non-target lesions met complete response crteria during their last scan.

RECIST 1.1 Tumor Response Assessments (Measurable Disease)

Baseline SoD (mm)	D29 SoD (mm, % change)	M3 SoD (mm, % change)	M5 SoD (mm, % change)	M6 SoD (mm, % change)	M7 SoD (mm, % change)
19	17 (-10.5)	11 (-42.1)	11 (-42.1)	11 (-42.1)	12 (-36.8)

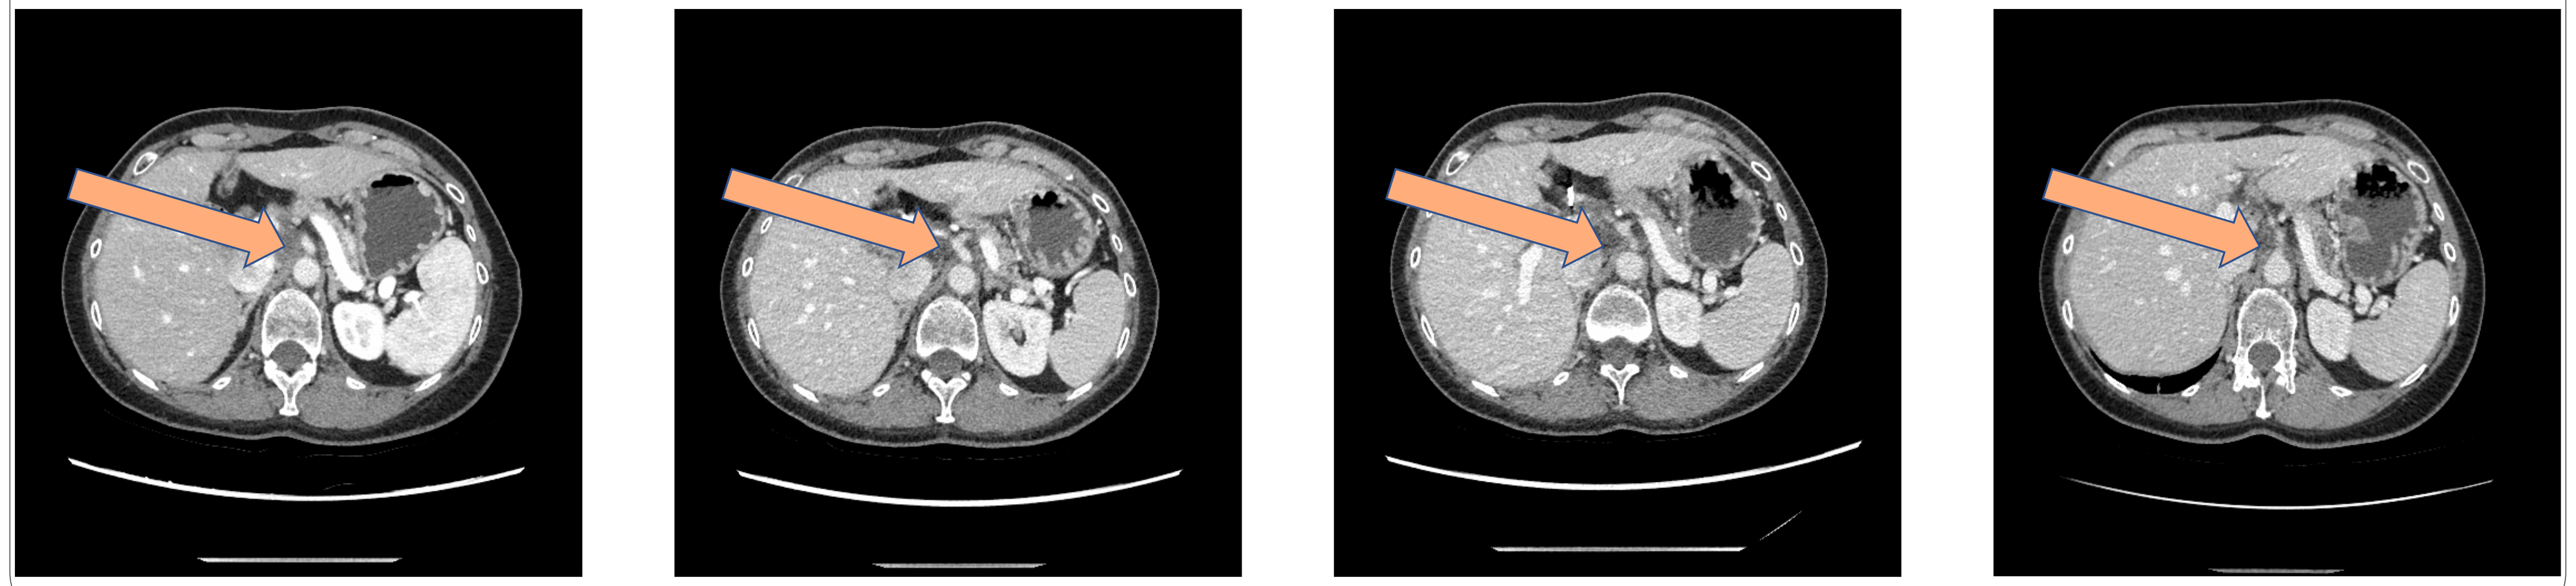

Baseline (19 mm) 3M (11 mm) 5M (11 mm) 7M (12 mm)

TAC01-CLDN18.2 PK AND CYTOKINE ANALYSIS

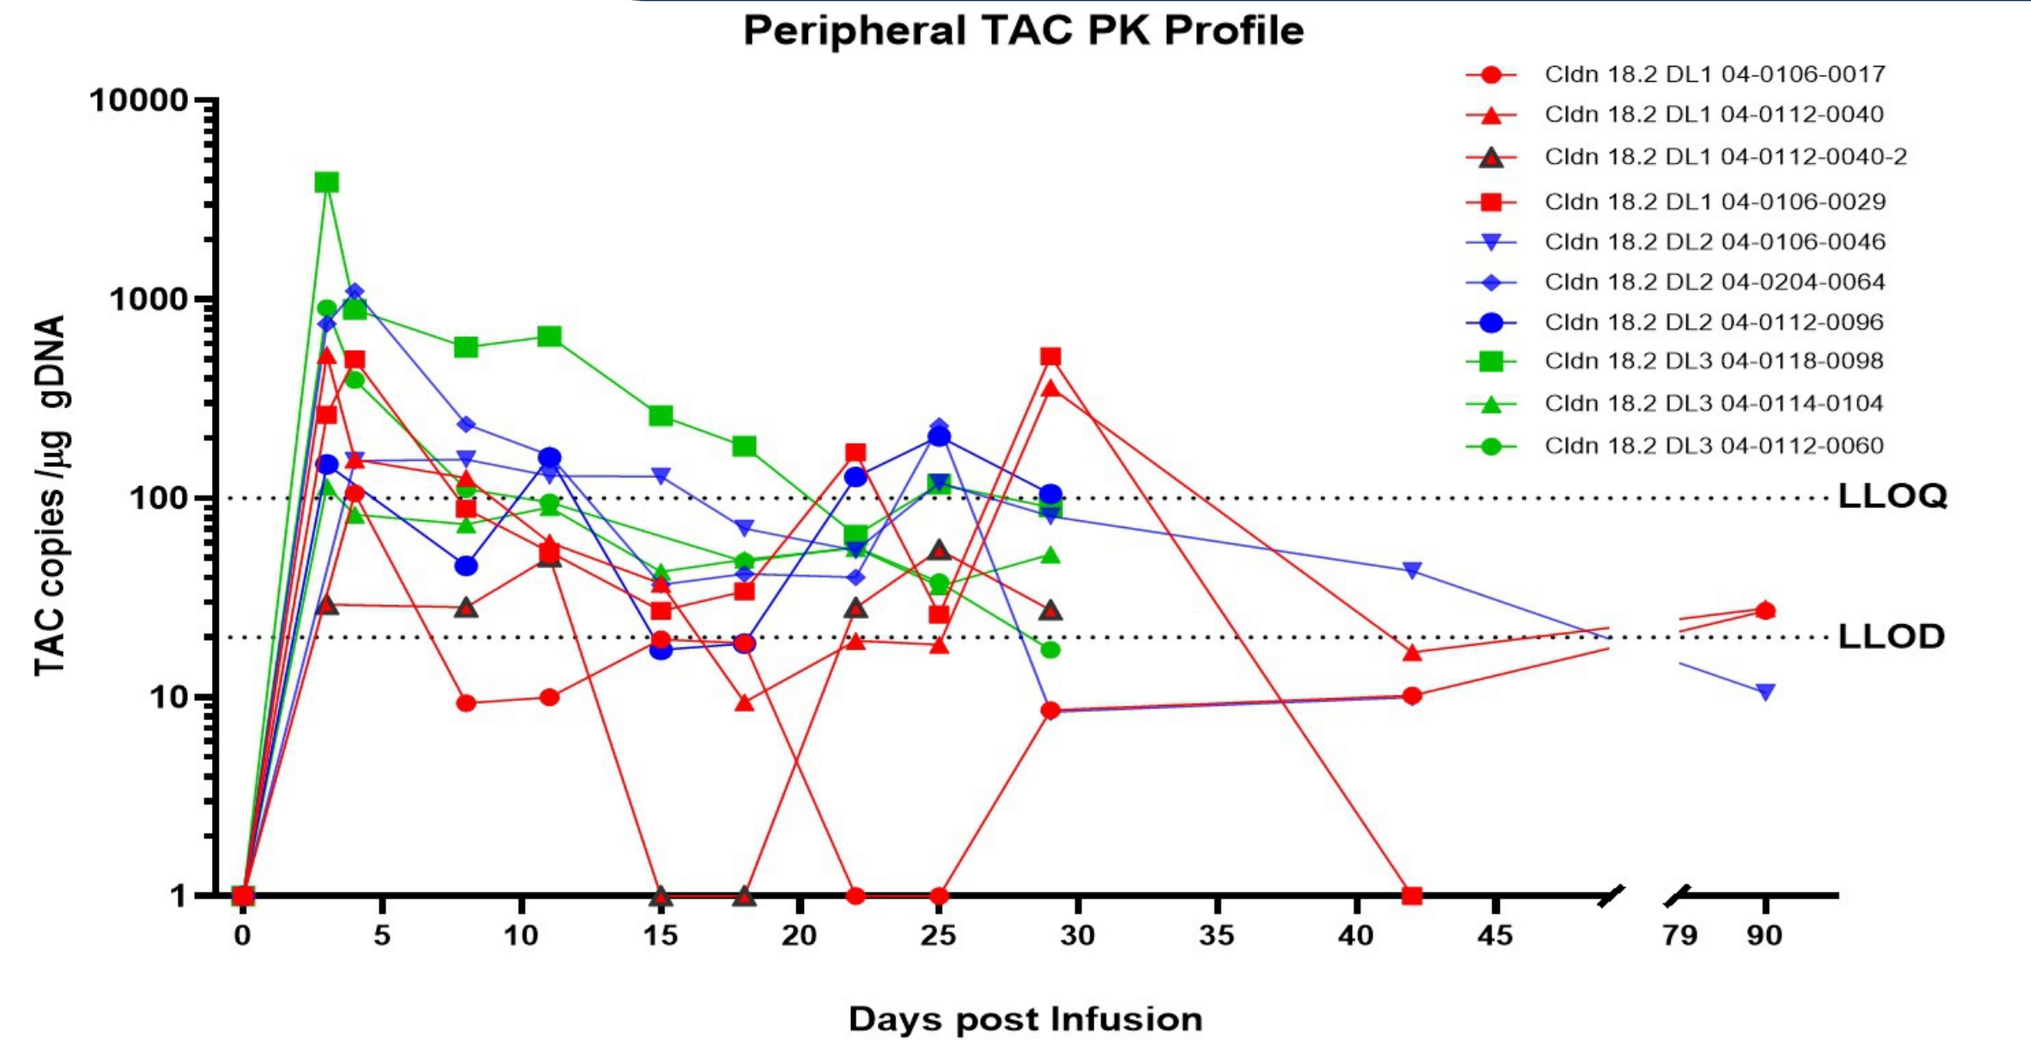
Peripheral TAC PK Profile
TAC copies /µg gDNA
Days post Infusion
LLOQ
LLOD
Cldn 18.2 DL1 04-0106-0017
Cldn 18.2 DL1 04-0112-0040
Cldn 18.2 DL1 04-0112-0040-2
Cldn 18.2 DL1 04-0106-0029
Cldn 18.2 DL2 04-0106-0046
Cldn 18.2 DL2 04-0204-0064
Cldn 18.2 DL2 04-0112-0096
Cldn 18.2 DL3 04-0118-0098
Cldn 18.2 DL3 04-0114-0104
Cldn 18.2 DL3 04-0112-0060

Subject / Dose Level	Biopsy Day	TAC copies /µg gDNA Blood	TAC copies /µg gDNA Biopsy
0064 / 1-3e^6 TAC+/ kg	Day 11 (A)	165	338
0064 / 1-3e^6 TAC+/ kg	Day 11 (B)	165	304
0064 / 1-3e^6 TAC+/ kg	Day 29 (A)	BLOD	64.44
0064 / 1-3e^6 TAC+/ kg	Day 29 (B)	BLOD	139.9
0060 / 6-8e^6 TAC+/kg	Day 29	17.3	218.5

TAC copies detected in the blood of subjects at the indicated days post-treatment. Red, blue and green lines indicate DL1, DL2 and DL3 subjects, respectively. TAC concentration after the second dose (black outline) was lower than similar timepoints after the first dose. TAC T cells still persist in the circulation of most subjects at D29, and even 3 months after infusion (D90; DL1 and DL2 data only available). Biopsies, where available, indicate a higher concentration of TAC cells in the tumor bed compared to blood samples taken at the same time. BLOD=below lower level of detection.

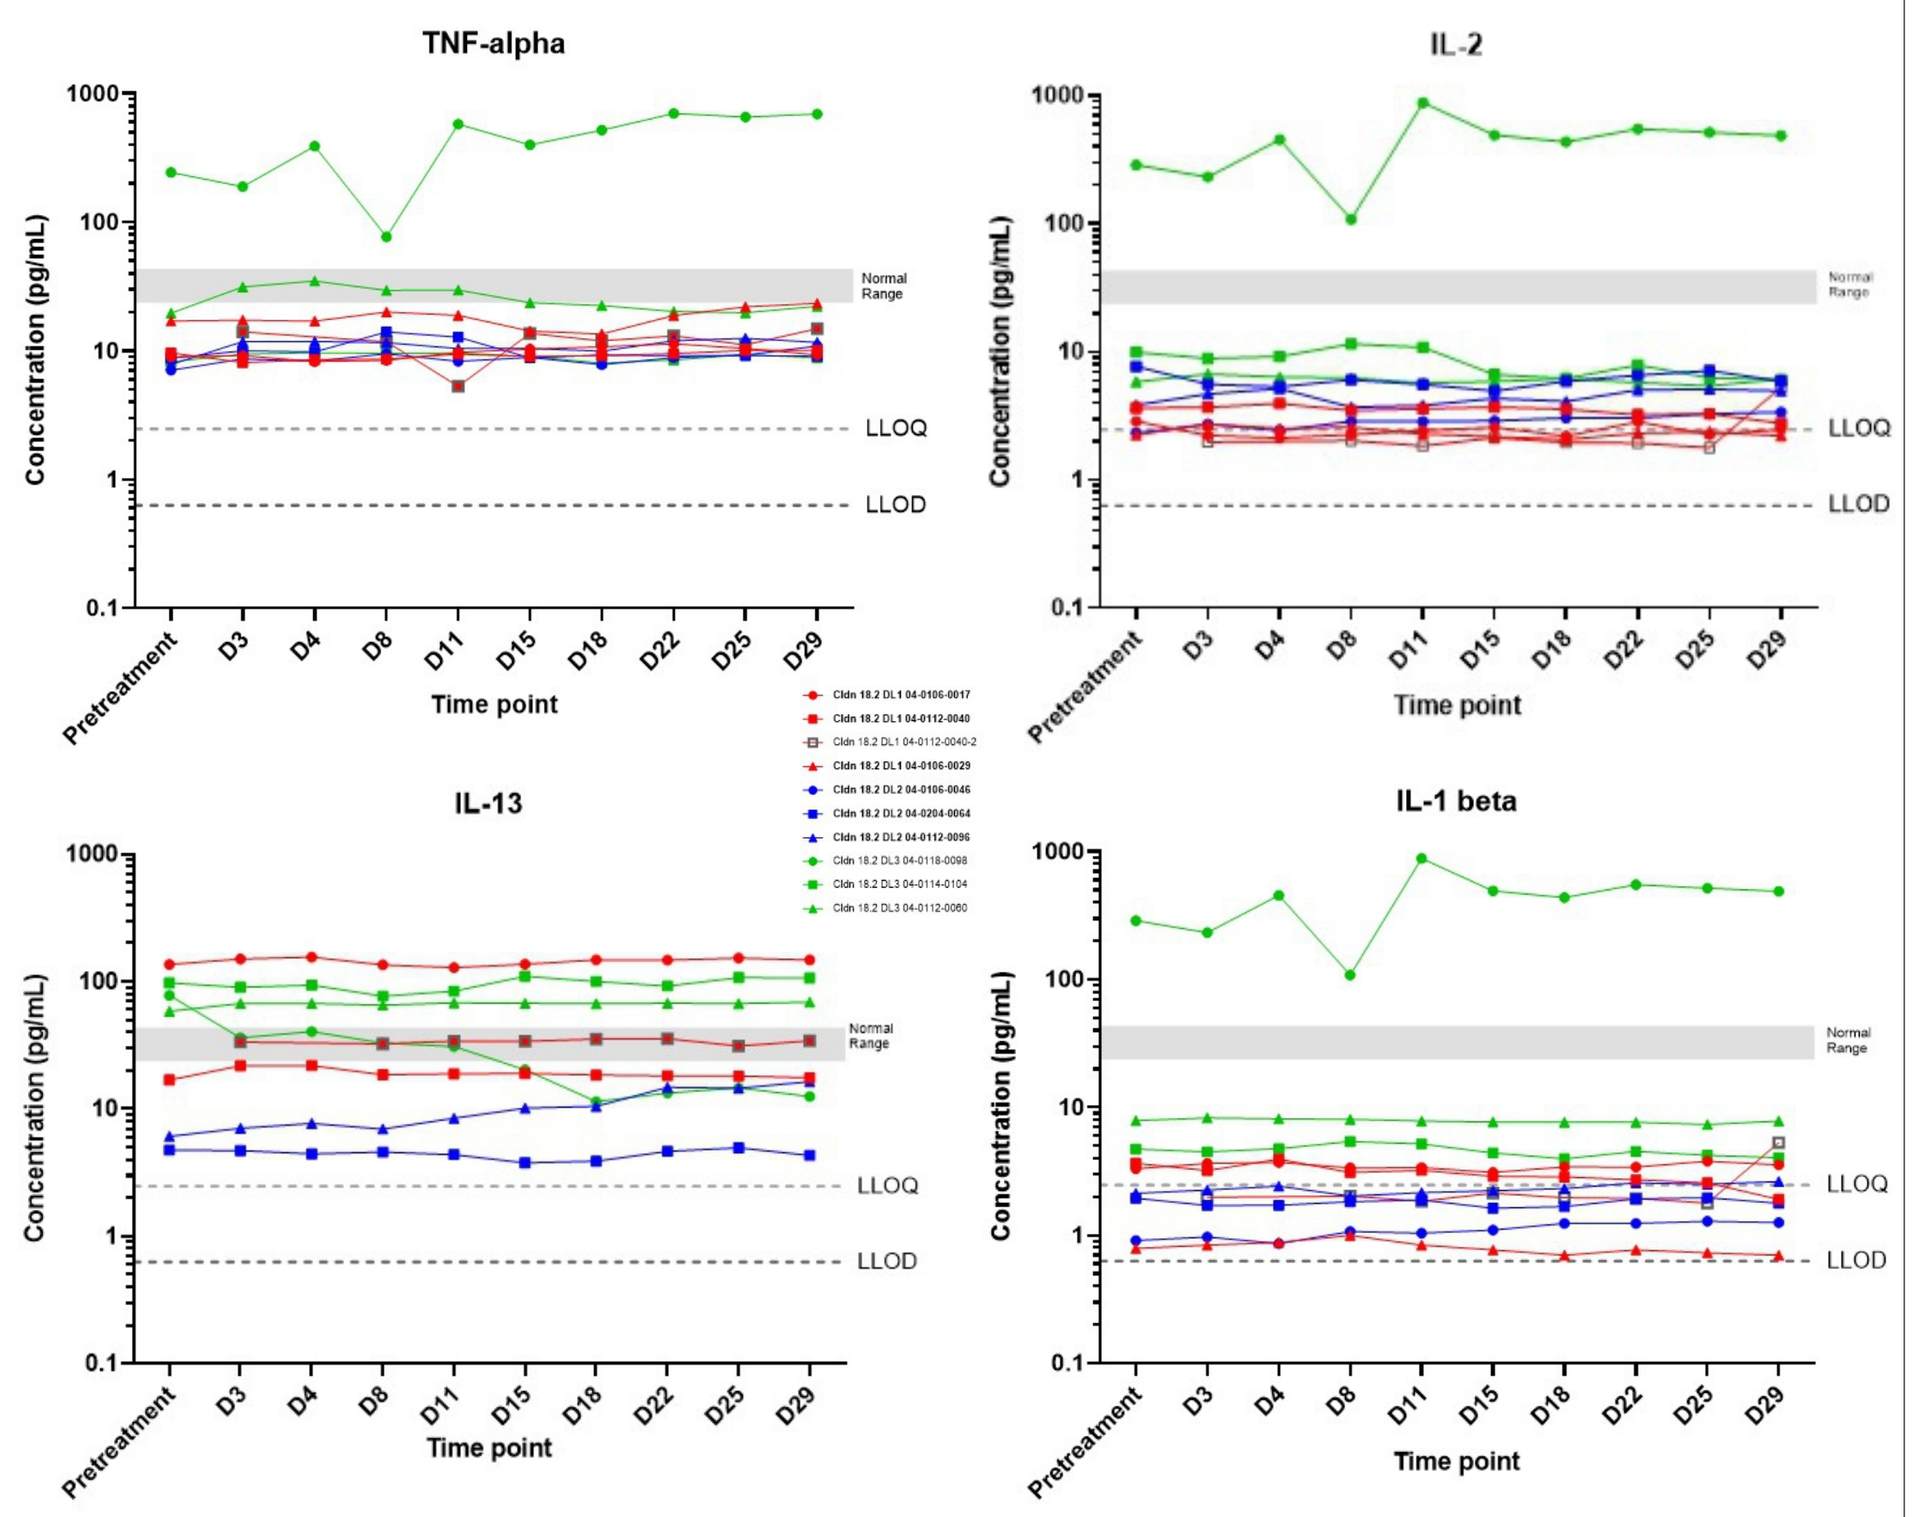
TNF-alpha
IL-2
IL-13
IL-1 beta
Concentration (pg/mL)
Time point
Normal Range
LLOQ
LLOD
Pretreatment
D3
D4
D8
D11
D15
D18
D22
D25
D29

Selected cytokines (TNFα, IL-2, IL-13 and IL-1β) were detected in the blood of subjects at the indicated days post-treatment. Most cytokine (TNFα, IL-2 and IL-1β) concentrations were below or within physiological levels (gray rectangle) with the notable exception of the subject experiencing the Grade 3 TAC-related adverse events (0118-0098).

SUMMARY & CONCLUSIONS

Safety

Interim results from the Phase I TACTIC-3 study suggests that TAC01-CLDN18.2 has manageable toxicity. Two Grade 3 treatment-related events of gastritis and gastric hemorrhage were observed, but resolved within 4 days. One Grade 1 ICAN event was observed, which resolved without intervention within 24 hours. Three low-grade CRS events were reported, which resolved with standard interventions..

Preliminary Efficacy

Demonstrated early signs of clinical activity, highlighting a confirmed, ongoing partial response (in a stage IV pancreatic cancer subject) at DL1, and an unconfirmed partial response at DL3. Overall DCR of 70% and ORR of 20% at 1st assessment across all DLs.

TRIAL PROGRESS: DL1-3 deemed safe. DL3 expanded.

Acknowledgements: Clinical Trial Sites and Apheresis Unit staffs, as well as the patients and their families.

Sponsorship: This Phase I/II clinical trial is fully funded by Triumvira Immunologics Inc.
Contact: MApostolopoulou@triumvira.com, eeileana@mdanderson.org