1773

PRE-CLINICAL CHARACTERIZATION OF ALLOGENEIC Vγ9Vδ2 HER2-TAC T CELLS FOR THE TREATMENT OF HER2-POSITIVE SOLID TUMORS

triumvira

Suzanna L. Prosser, Stacey X. Xu, Ling Wang, Ritu R. Randhawa, Sailaja Pirati, Deepika Bhemarasetty, Seung Mi Yoo, Miyoung Jung, Laurentia Gheorghiu, Chris Ayers, Donna Rill, Christopher W. Helsen and Andreas G. Bader

Triumvira Immunologics, 9433 Bee Caves Rd Building 1, Suite 240, Austin, TX 78733, USA (Headquarters), 270 Longwood Road South, Hamilton, Ontario L8P 0A6, Canada (Research Division)

ABSTRACT

Background

The T cell antigen coupler (TAC) is a novel, proprietary chimeric receptor that facilitates the redirection of T cells to tumor cells, and activates T cells by co-opting the endogenous T cell receptor complex with the goal to elicit safe and durable anti-tumor responses. TAC01-HER2, a first-in-class, autologous TAC T cell product targeting HER2 (ERBB2), has entered a phase I/II clinical trial in patients with HER2-positive solid tumors. Here, we describe the development of an allogeneic HER2-TAC T cell product based on Vγ9Vδ2 (γδ) T cells which belong to a subset of T cells that recognize target cells in a human leukocyte antigen (HLA) independent manner. Thus, γδ T cells do not cause GvHD and have the potential for allogeneic cell therapy applications.

Materials and Methods

A variety of in vitro and in vivo assays were used to evaluate the potency and safety of HER2-TAC γδ T cells generated from multiple donors. In vitro assays included flow cytometric analysis determining the γδ T cell phenotype, intracellular cytokines, CD69 upregulation and T cell proliferation. Anti-tumor cytotoxicity was assessed via real-time microscopy-based co-culture assays. Mixed lymphocyte reactions (MLR) were performed to measure cytokine production and proliferation of HER2-TAC γδ T cells in response to HLA mismatches between unrelated donors. In vivo studies examined the anti-tumor effect of HER2-TAC γδ T cells against established solid HER2-expressing tumors.

Results

HER2-TAC γδ T cells selectively reacted to HER2-expressing tumor cells in co-culture, as demonstrated by CD69 upregulation, intracellular cytokine production, increase in proliferation, and cytotoxicity. In contrast, HER2-TAC γδ T cells failed to show activity in MLR assays, potentially indicating that HER2-TAC γδ T cells are free of GvH reactivity. These MLR assays comprised dendritic cells that represent the major HLA subtypes found in North America. In addition, HER2-TAC γδ T cells showed strong anti-tumor efficacy in HER2-positive human tumor xenograph mouse models, without signs of toxicity.

Conclusions

The in vitro and in vivo data confirms strong and specific activity of HER2-targeted TAC γδ T cells against HER2-expressing tumor models, and highlights the potential of the TAC platform in the development of an allogeneic product for therapeutic applications in solid tumors.

TAC SCIENCE

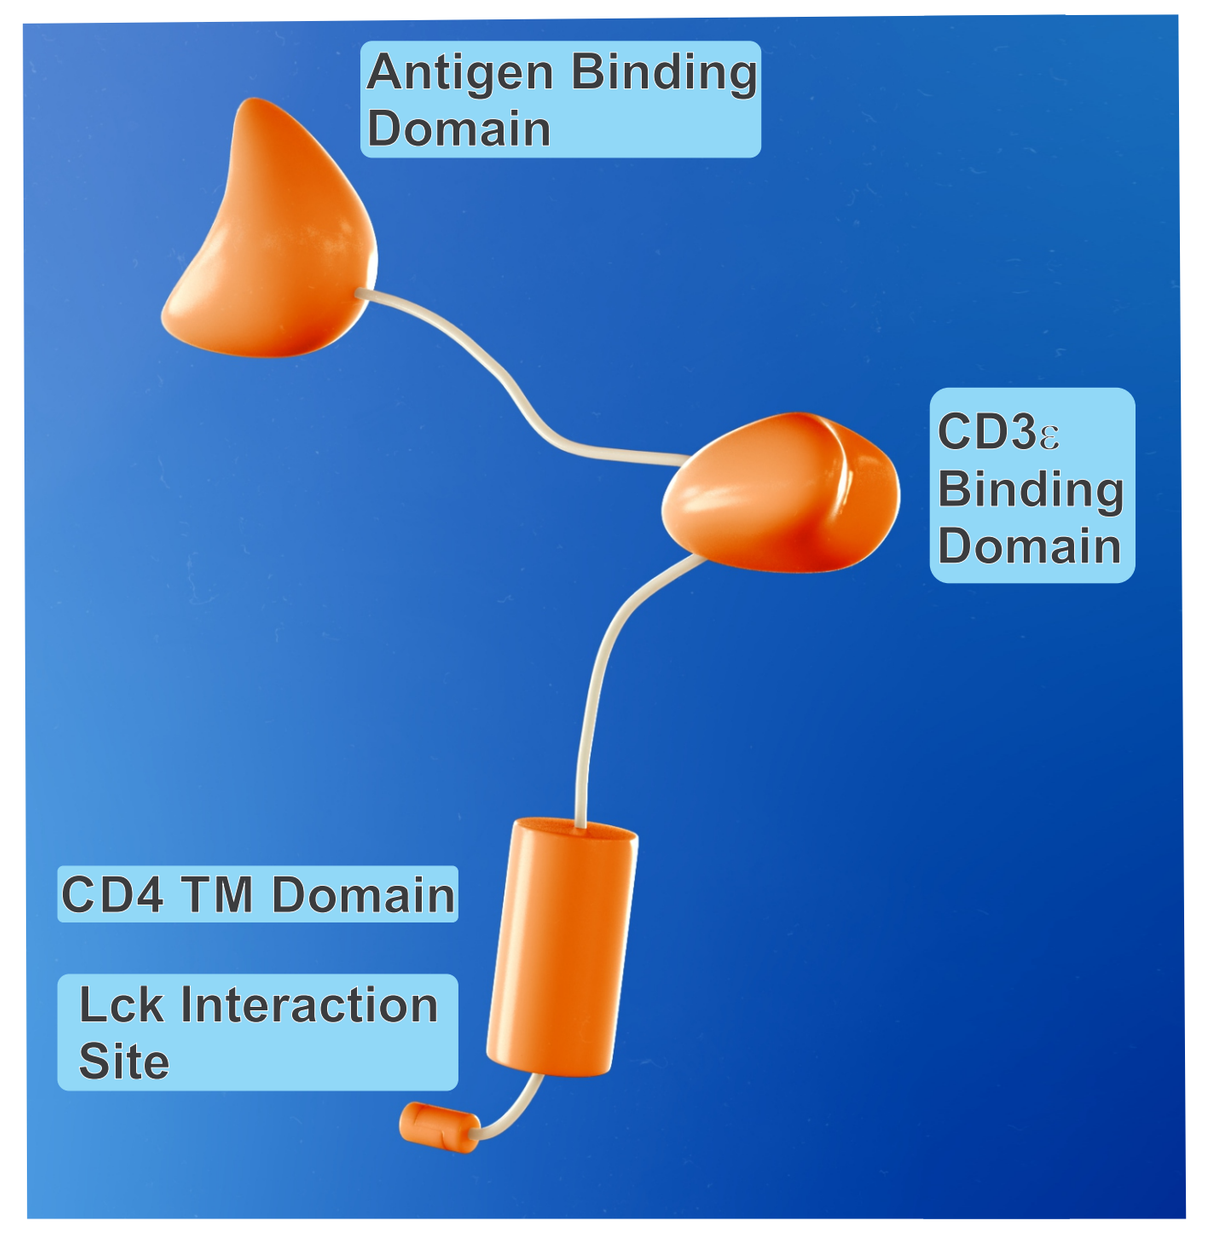
Antigen Binding Domain
CD3ε Binding Domain
CD4 TM Domain
Lck Interaction Site

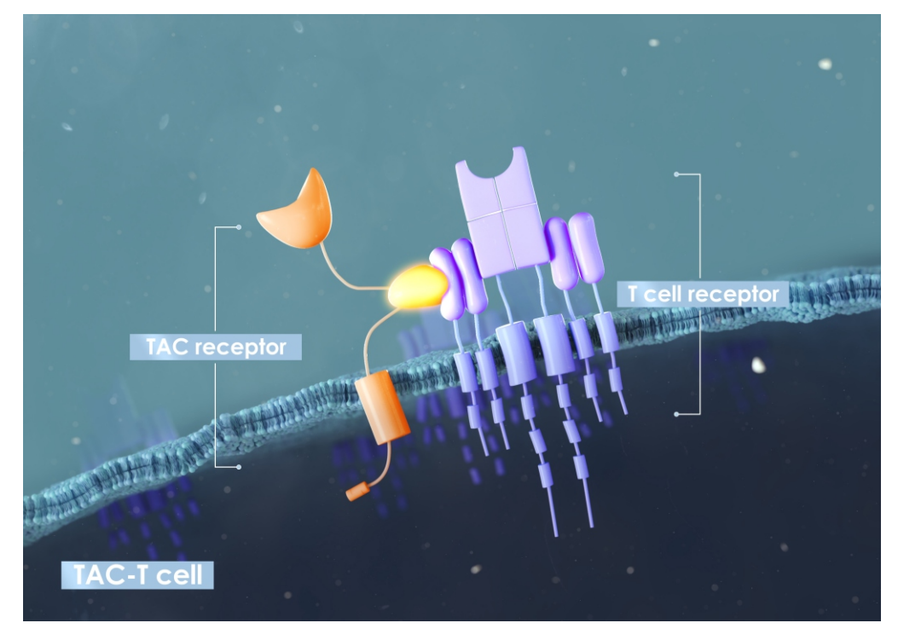
TAC receptor
T cell receptor
TAC-T cell

The membrane-bound TAC receptor interacts directly with the TCR-CD3 epsilon domain and...

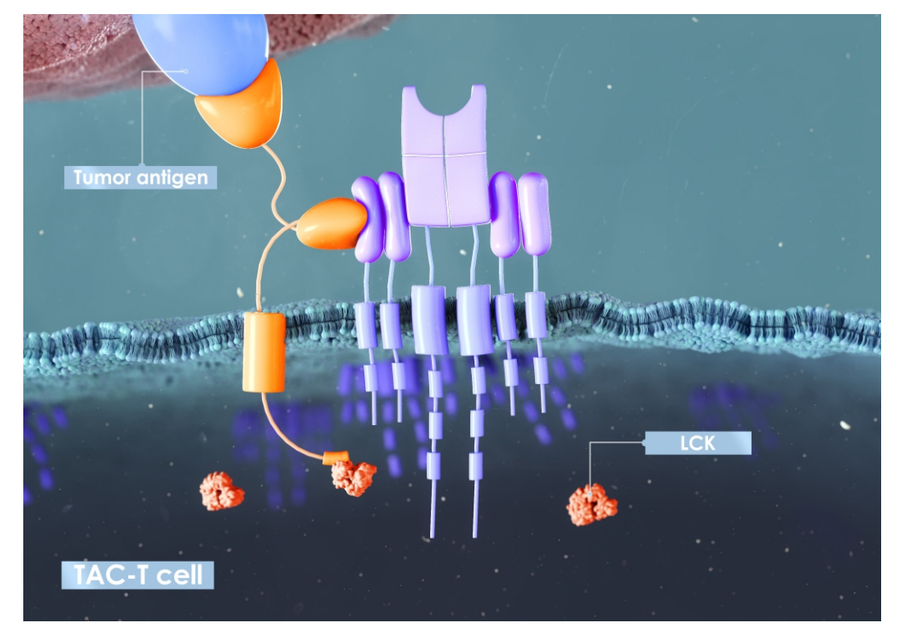
Tumor antigen
LCK
TAC-T cell

...binds directly to the targeted tumor antigen. Clustering of TAC-TCR complexes leads to recruitment of kinases (Lck) via the cytoplasmic co-receptor domain and...

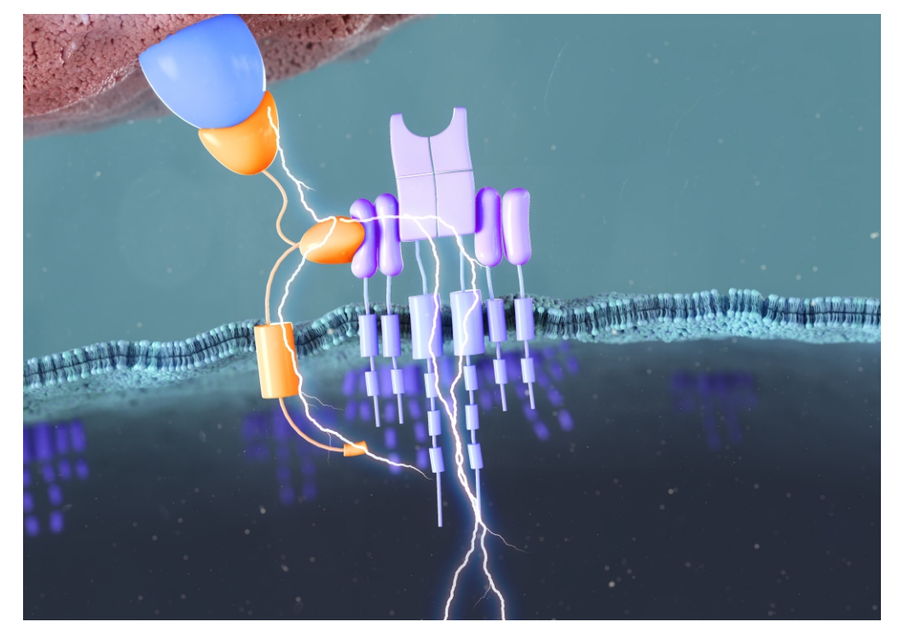

...initiates T cell activation via the endogenous CD3-TCR complex.

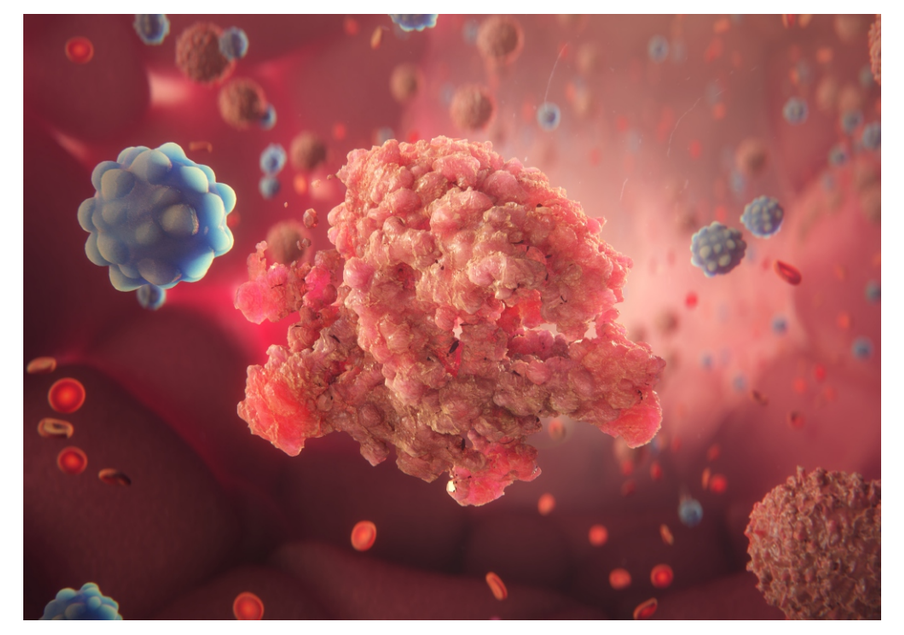

This results in effective cell lysis of multiple tumor cells during multiple killing events.

Watch a short animation to understand the TAC mechanism

Key features of TAC technology:

- TAC functions independently of MHC
- TAC activates T cells via the endogenous TCR
- TAC incorporates the co-receptor and recruits the TCR complex, mimicking natural TCR activation

triumvira

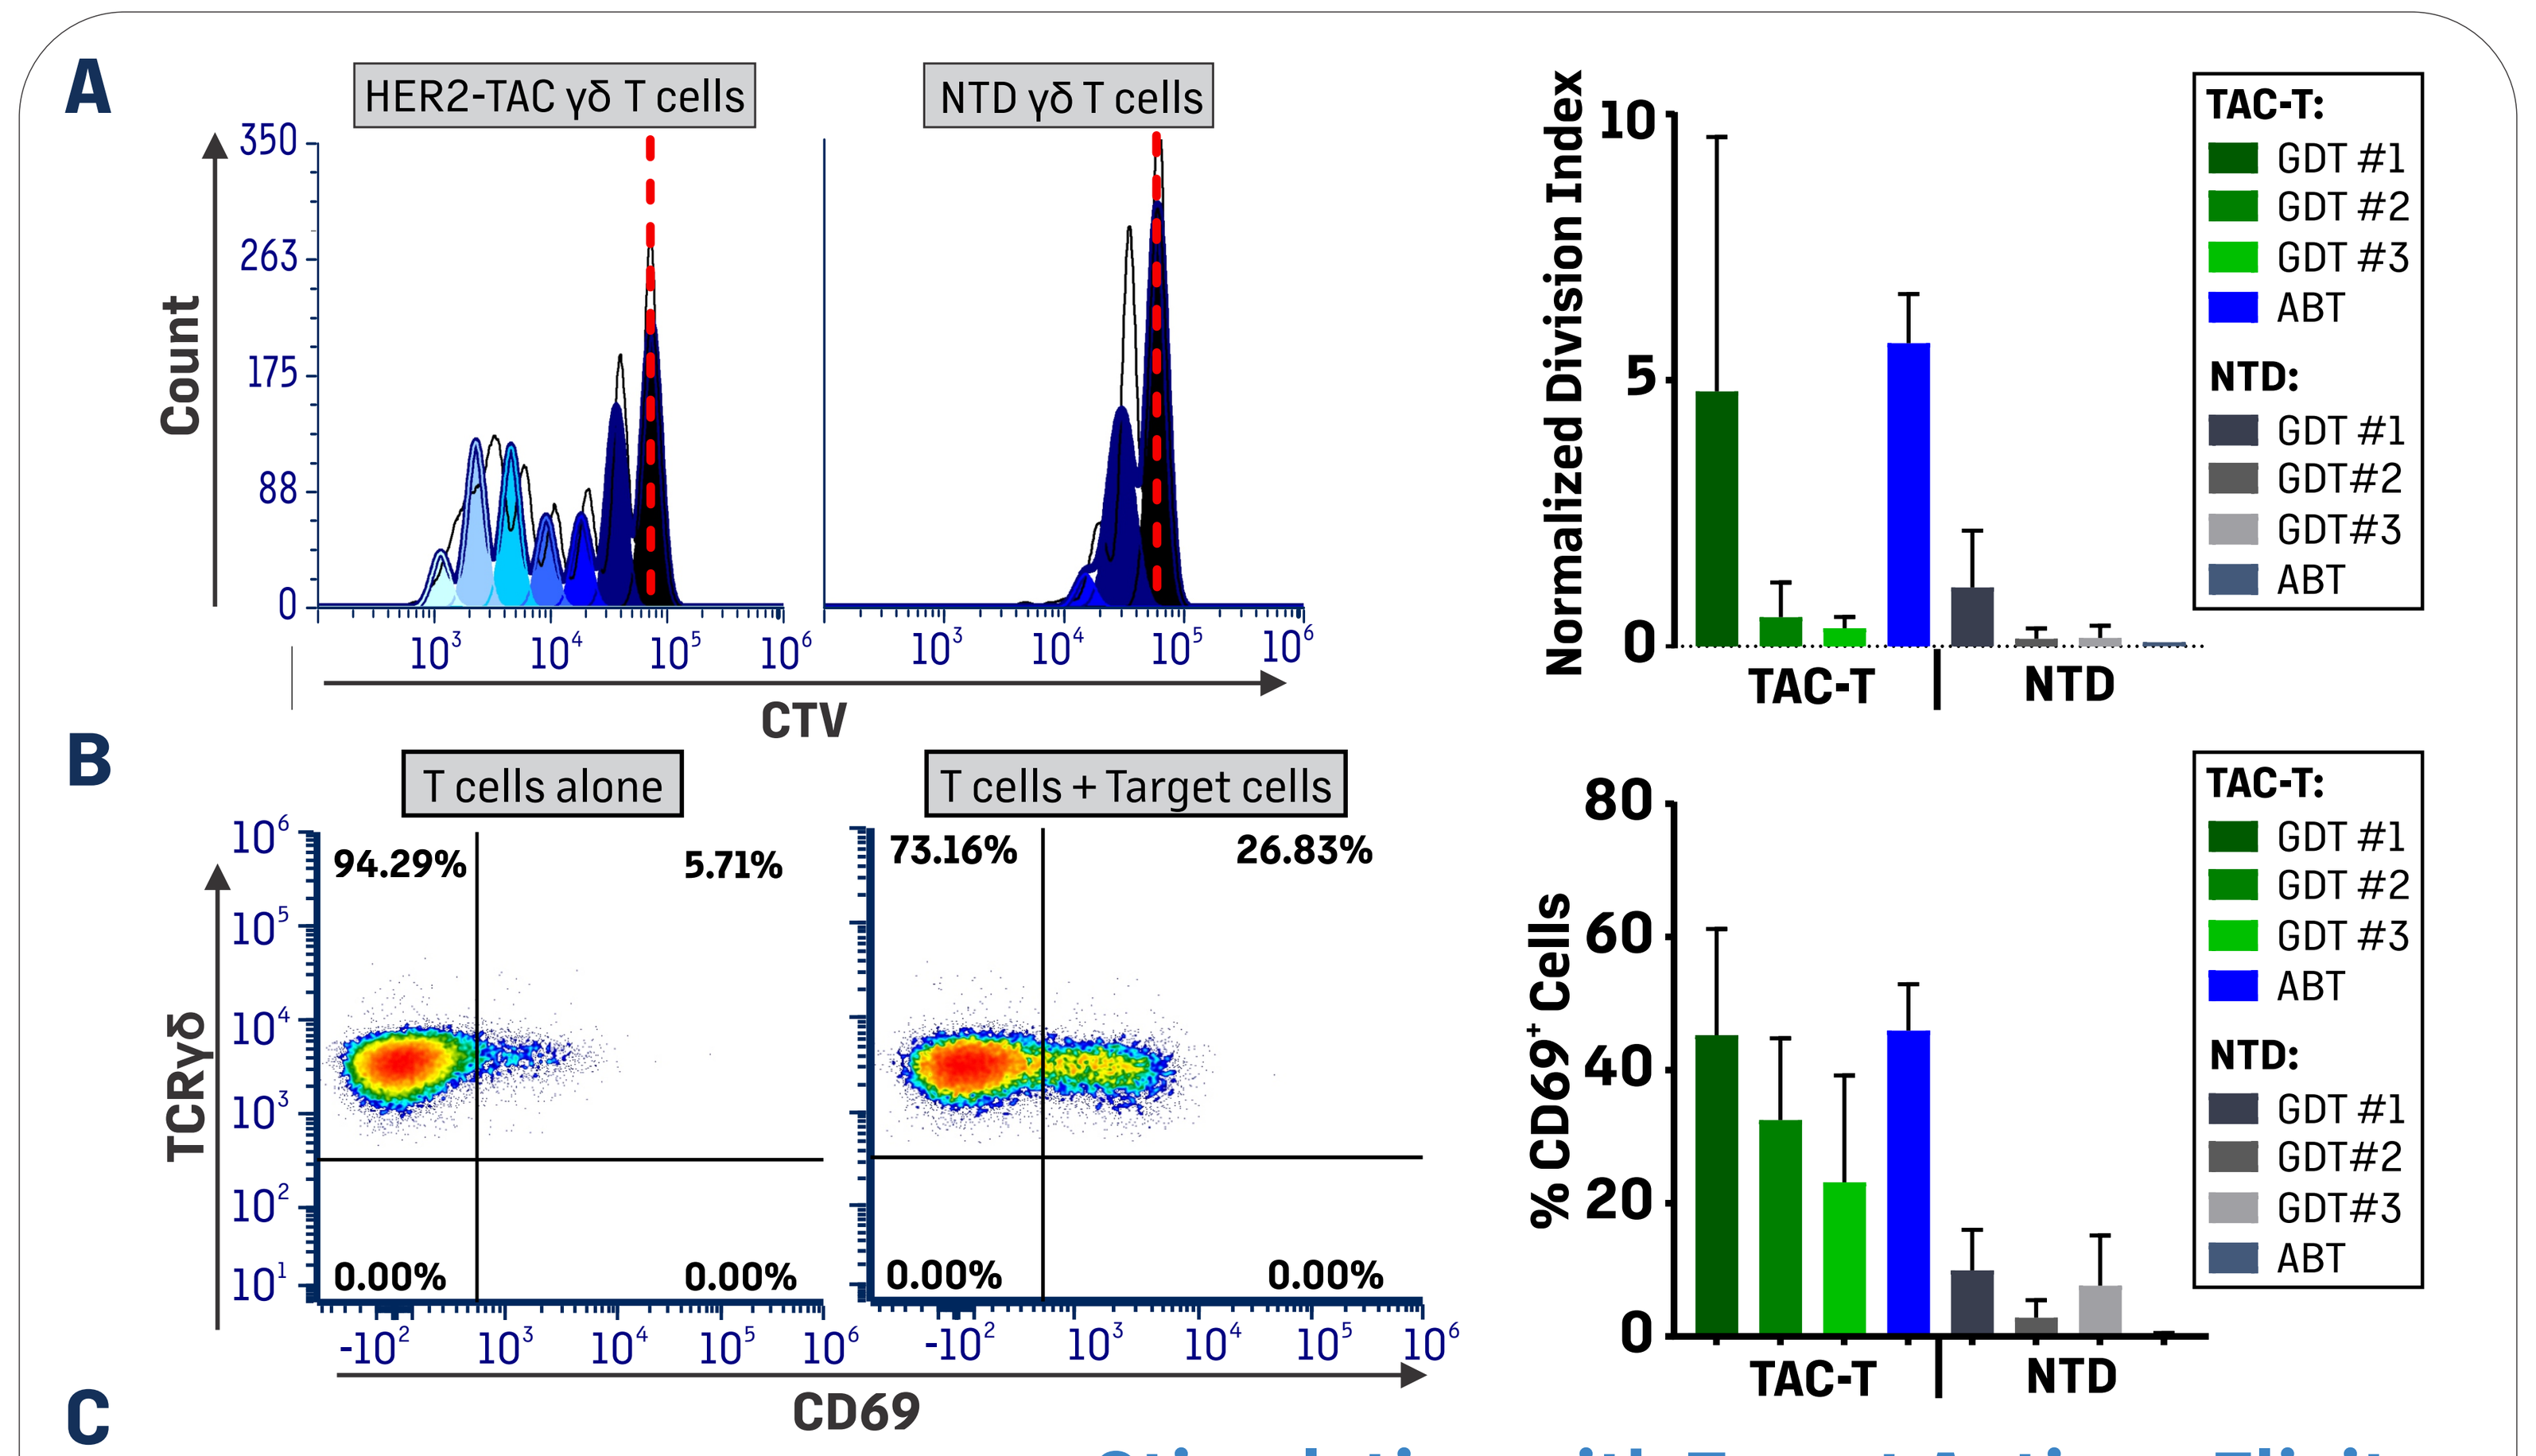
A
HER2-TAC γδ T cells
NTD γδ T cells
Count
350
263
175
88
0
10^3 10^4 10^5 10^6
CTV
Normalized Division Index
TAC-T
NTD
TAC-T: GDT #1 GDT #2 GDT #3 ABT
NTD: GDT #1 GDT#2 GDT#3 ABT
B
T cells alone
T cells + Target cells
94.29%
5.71%
0.00%
0.00%
73.16%
26.83%
0.00%
0.00%
TCRγδ
CD69
% CD69$^+$ Cells

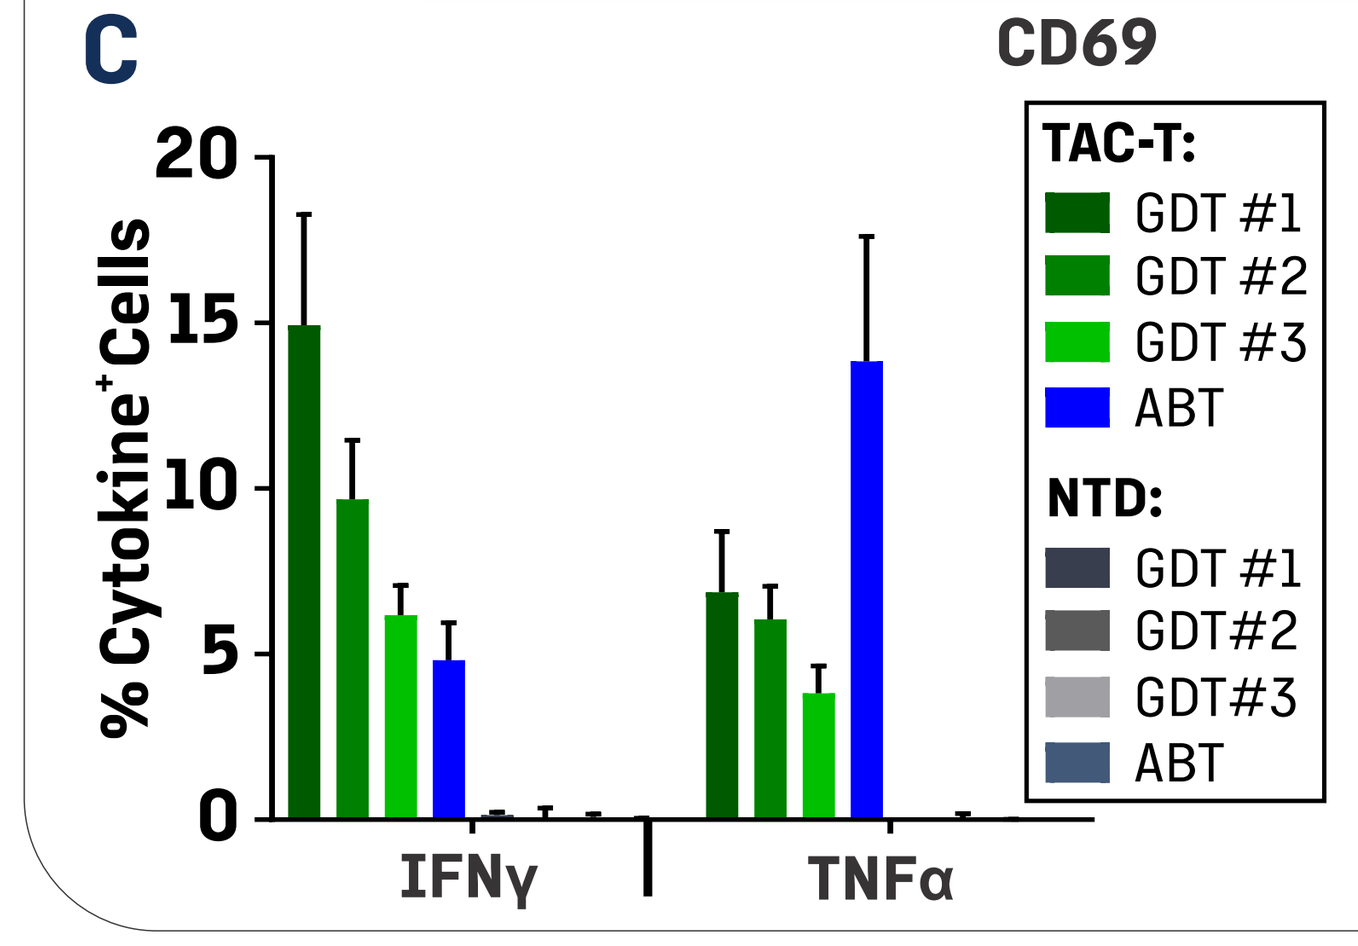
C
% Cytokine$^+$ Cells
IFNγ
TNFα
TAC-T: GDT #1 GDT #2 GDT #3 ABT
NTD: GDT #1 GDT#2 GDT#3 ABT

Stimulation with Target Antigen Elicits a Strong HER2-TAC γδ T Cell Response

HER2-TAC γδ T cells engineered from 3 healthy donors (GDT #1-3) were tested in a range of in vitro assays. TAC01-HER2 αβ T cells (ABT) served as a positive control. Proliferation: **A.** 4-day co-culture with HT1080 target cells. Early Activation: **B.** 4-hour co-culture with N87 target cells. Intracellular Cytokines: **C.** 4-hour co-culture with N87 target cells. All data shown as mean ± SD.

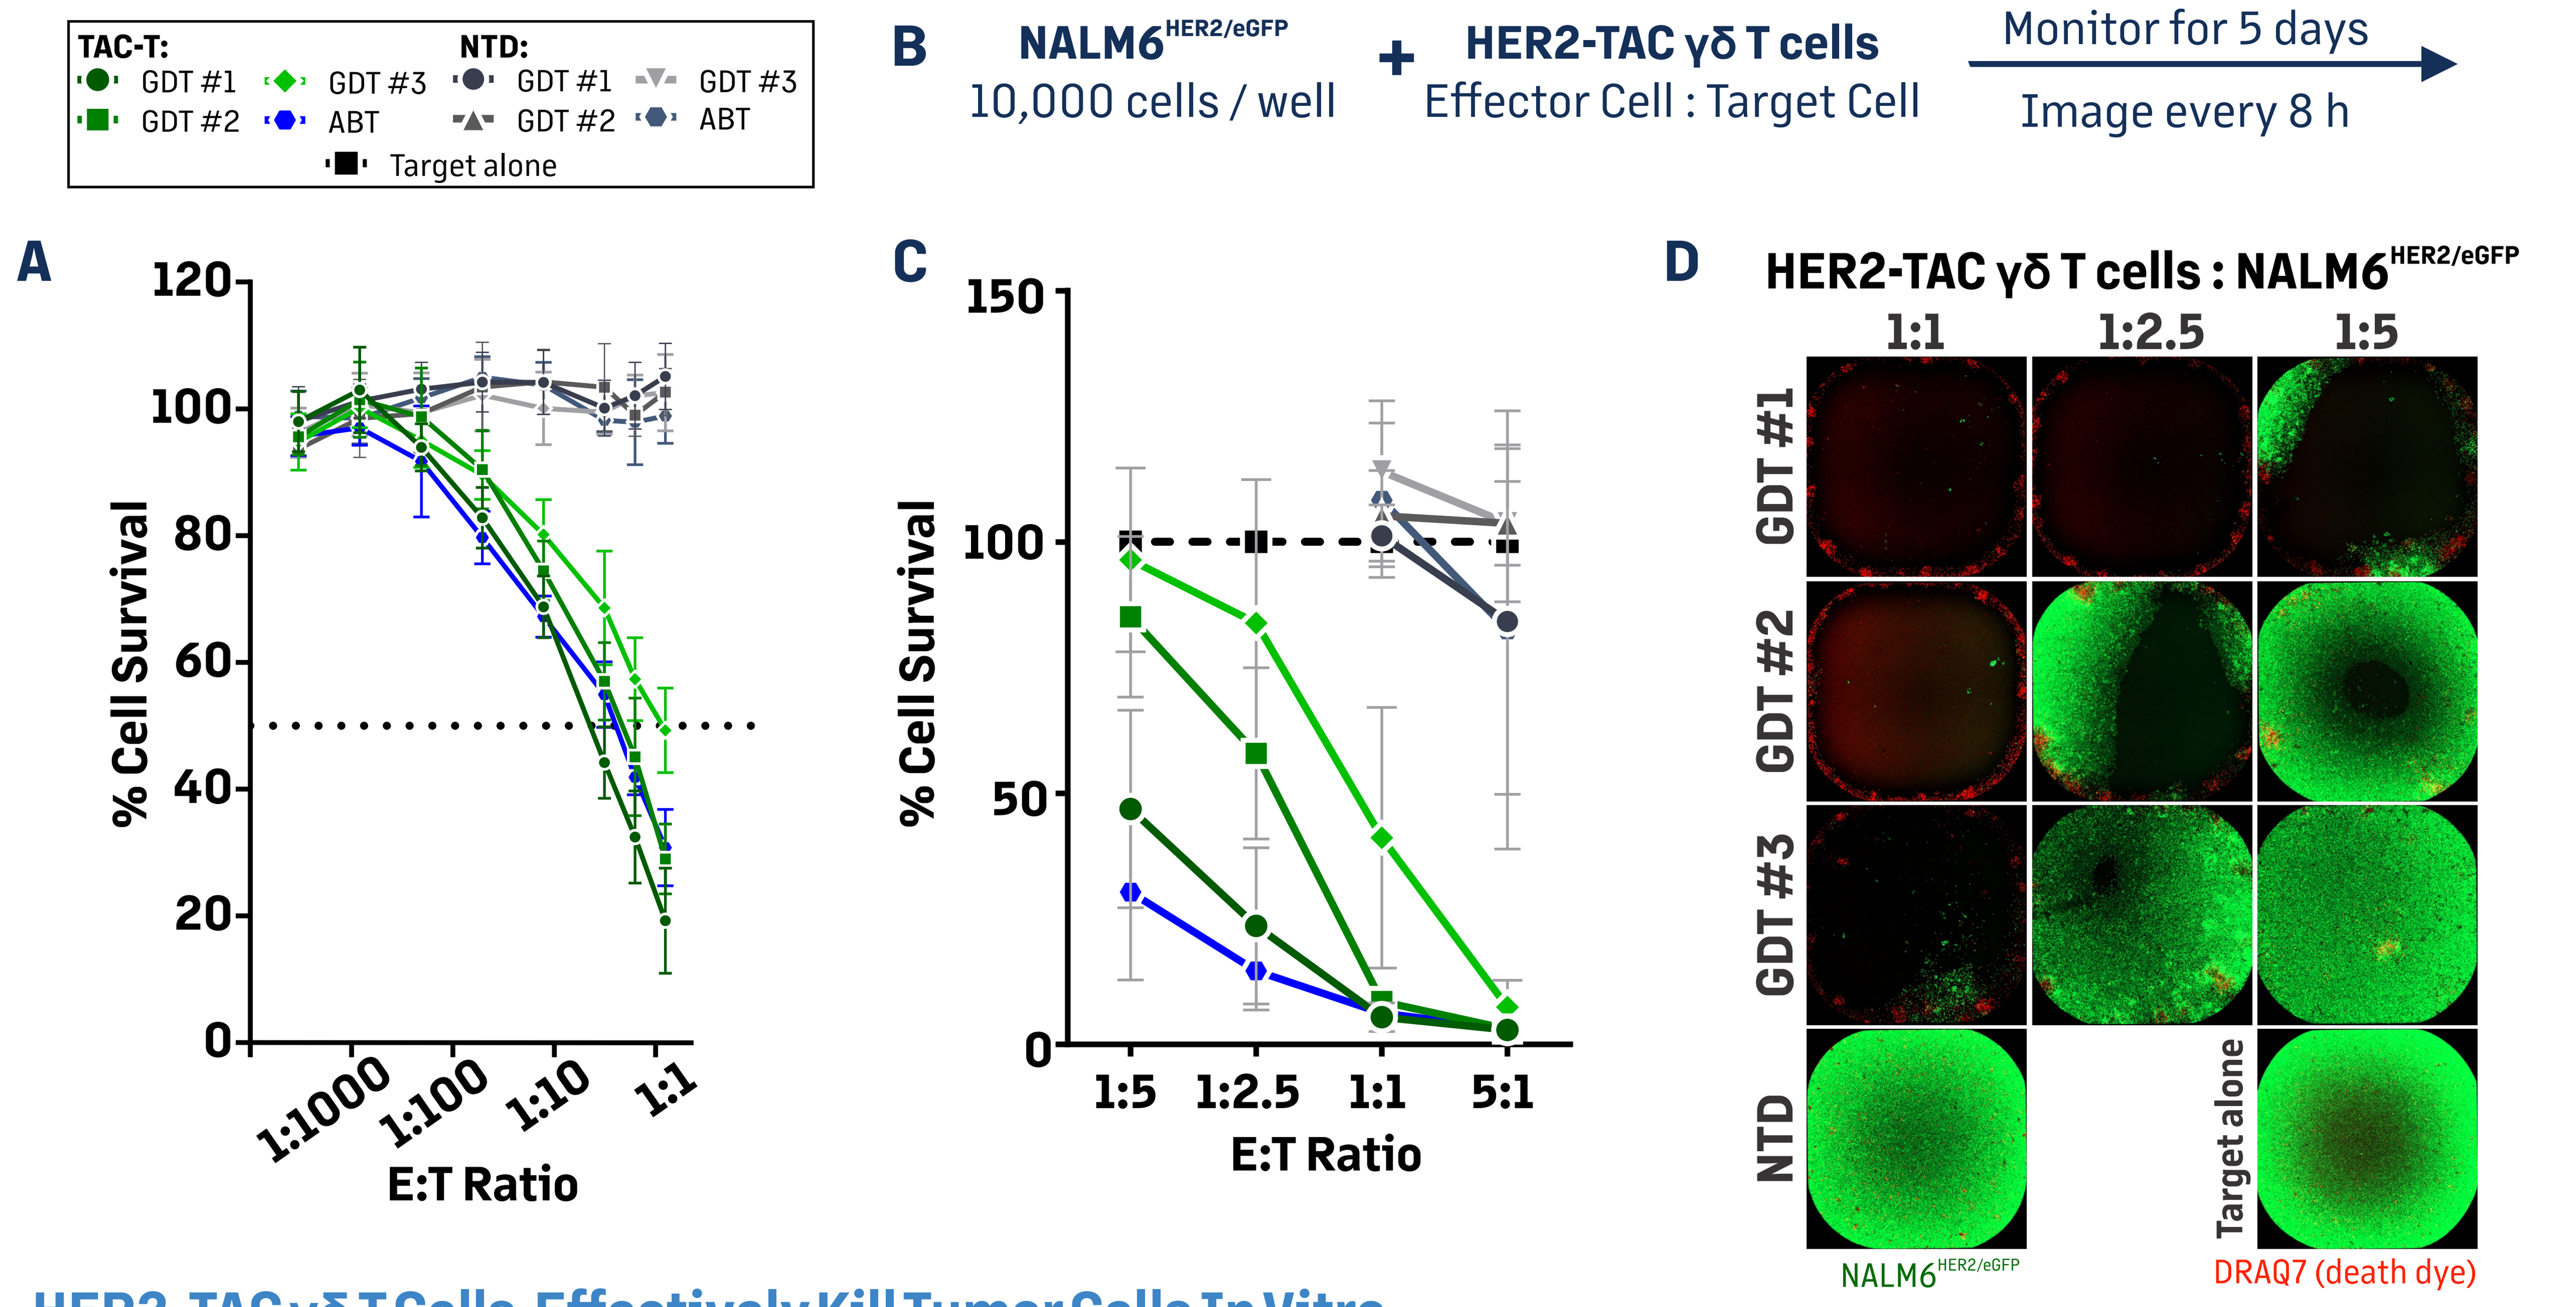
TAC-T: GDT #1 GDT #2 GDT #3 ABT
NTD: GDT #1 GDT #2 GDT #3 ABT
Target alone
A
% Cell Survival
E:T Ratio
1:1000
1:100
1:10
1:1
B
NALM6$^{HER2/eGFP}$
10,000 cells / well
+
HER2-TAC γδ T cells
Effector Cell : Target Cell
Monitor for 5 days
Image every 8 h
C
% Cell Survival
1:5
1:2.5
1:1
5:1
E:T Ratio
D
HER2-TAC γδ T cells : NALM6$^{HER2/eGFP}$
1:1
1:2.5
1:5
GDT #1
GDT #2
GDT #3
NTD
Target alone
NALM6$^{HER2/eGFP}$
DRAQ7 (death dye)

HER2-TAC γδ T Cells Effectively Kill Tumor Cells In Vitro

18-hour Cytotoxicity: **A.** Luciferase-based killing assay following co-culture with HT1080^{eLuc} target cells. 5-day Cytotoxicity: **B.** HER2-TAC γδ T cells were co-cultured with NALM6$^{HER2/eGFP}$ target cells at different E:T ratios. Tumor cell growth was monitored by GFP fluorescence. **C.** Mean percentage tumor cell survival is shown ± SD. **D.** Example images of 5-day co-cultures.

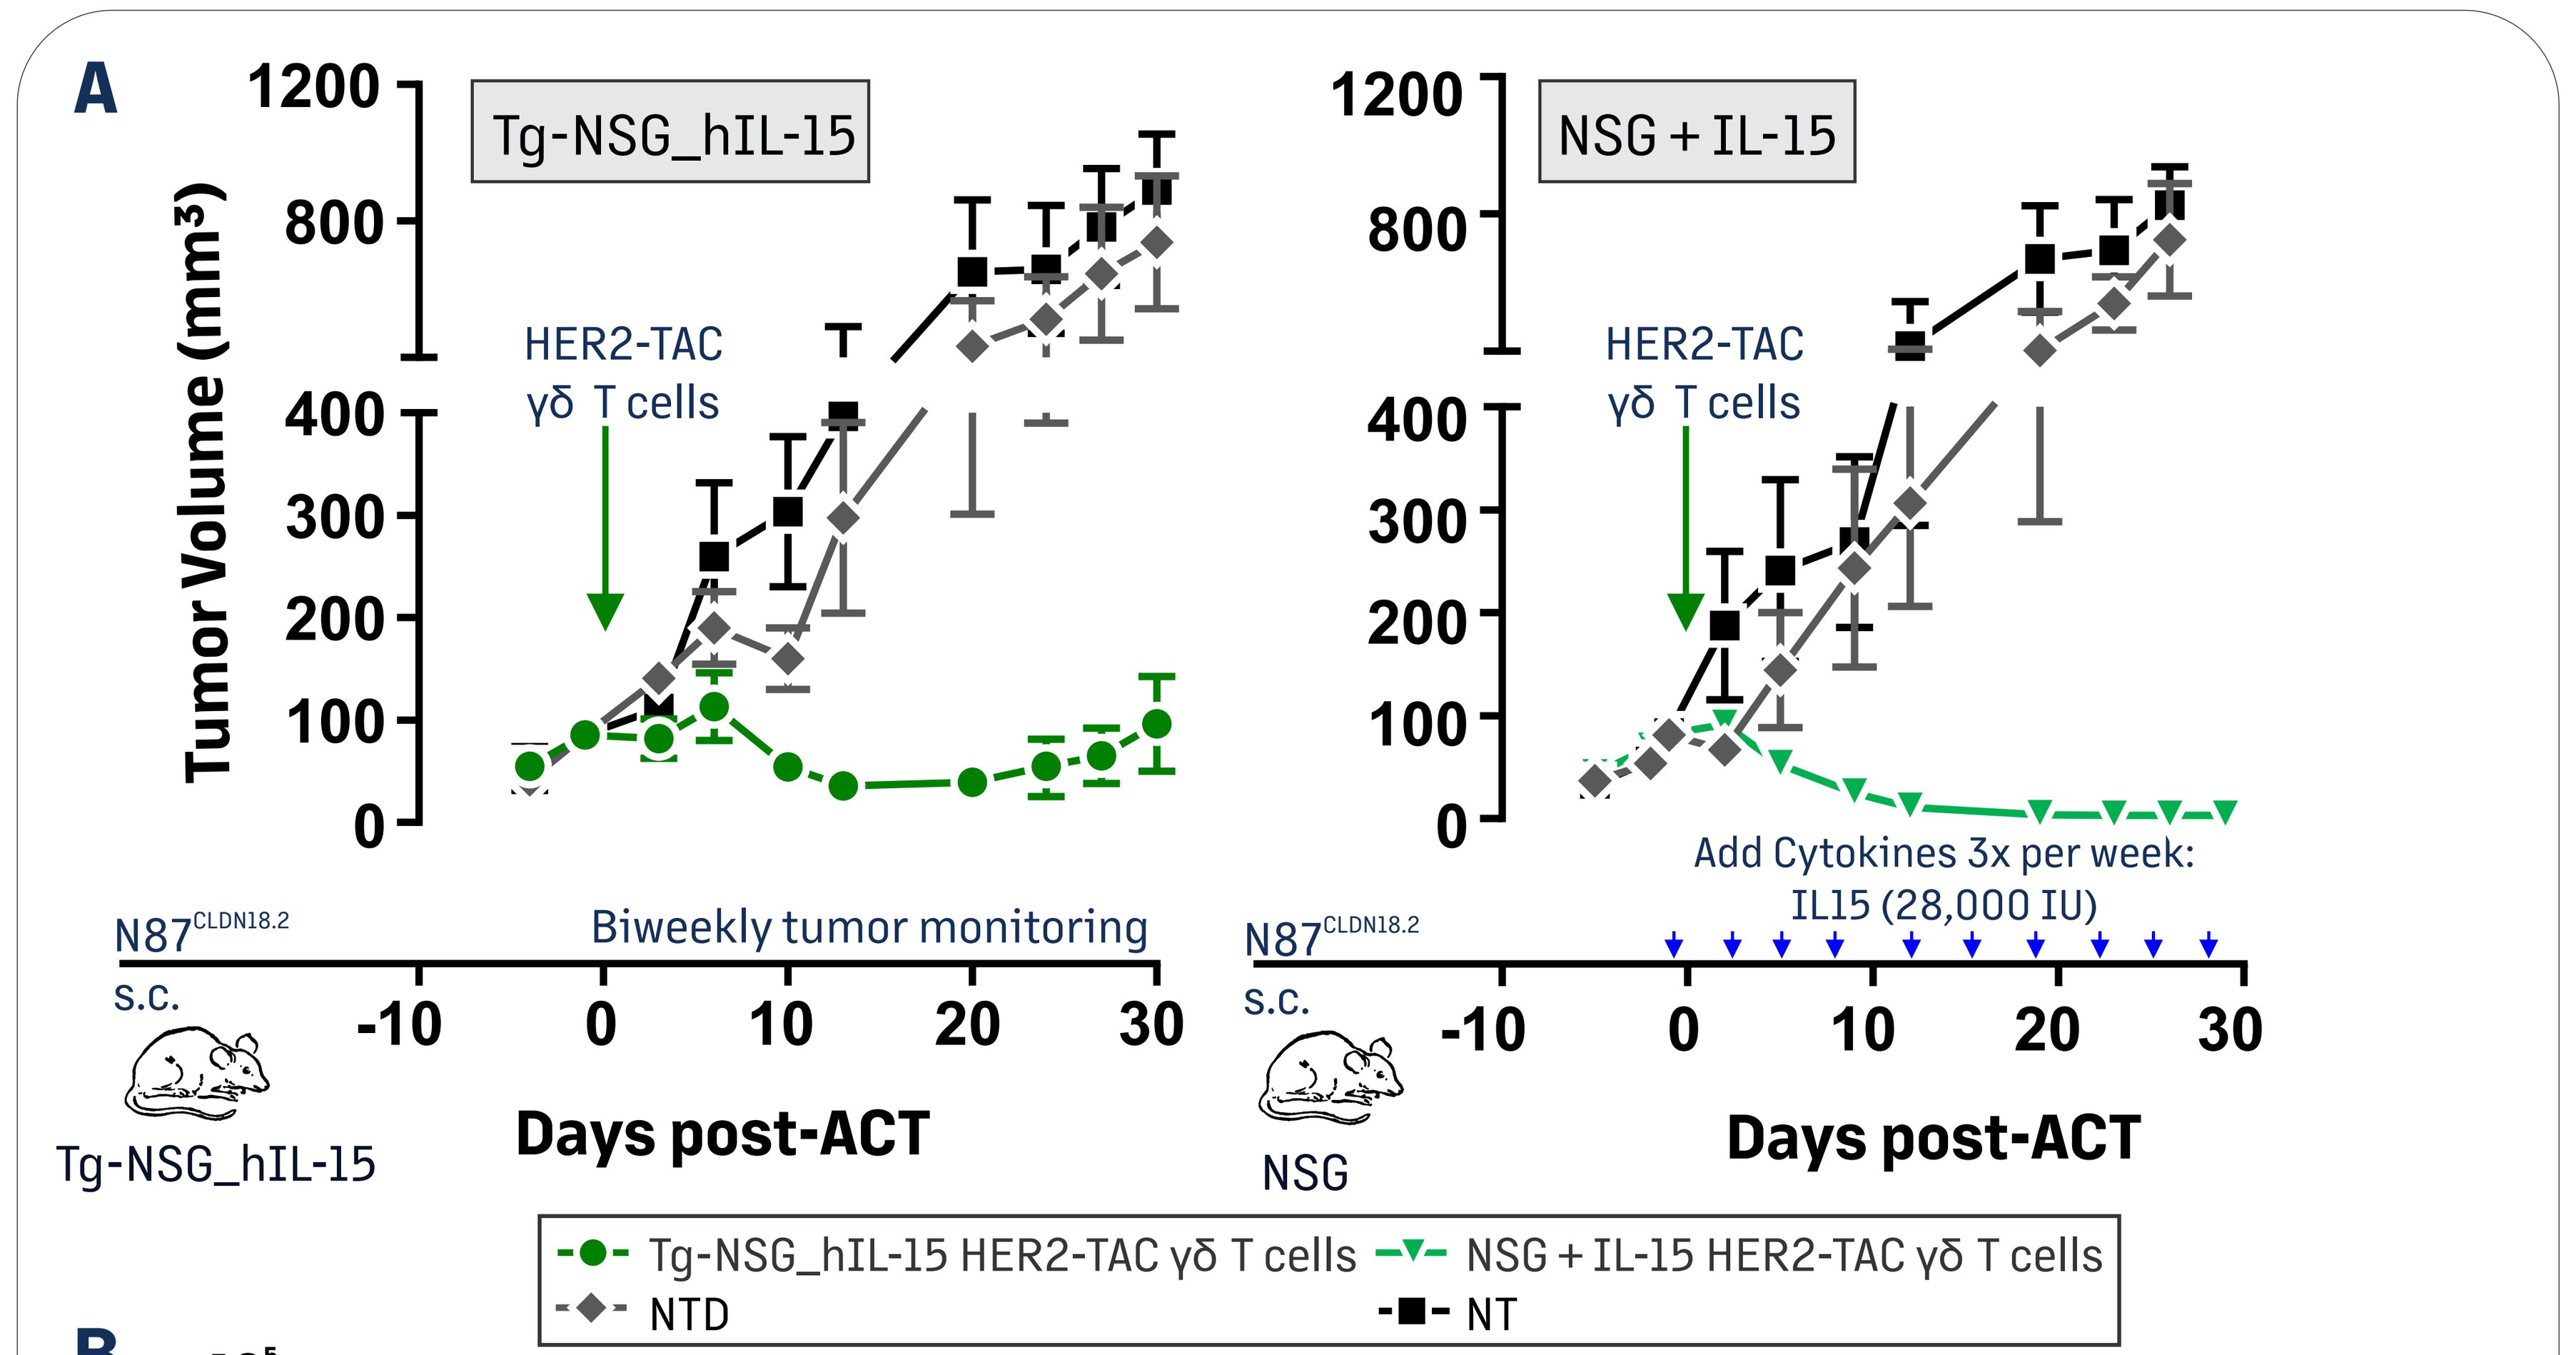
A
Tg-NSG_hIL-15
NSG + IL-15
Tumor Volume (mm³)
HER2-TAC γδ T cells
Biweekly tumor monitoring
Add Cytokines 3x per week:
IL15 (28,000 IU)
N87$^{CLDN18.2}$
s.c.
Days post-ACT
Tg-NSG_hIL-15
NSG
Tg-NSG_hIL-15 HER2-TAC γδ T cells
NSG + IL-15 HER2-TAC γδ T cells
NTD
NT

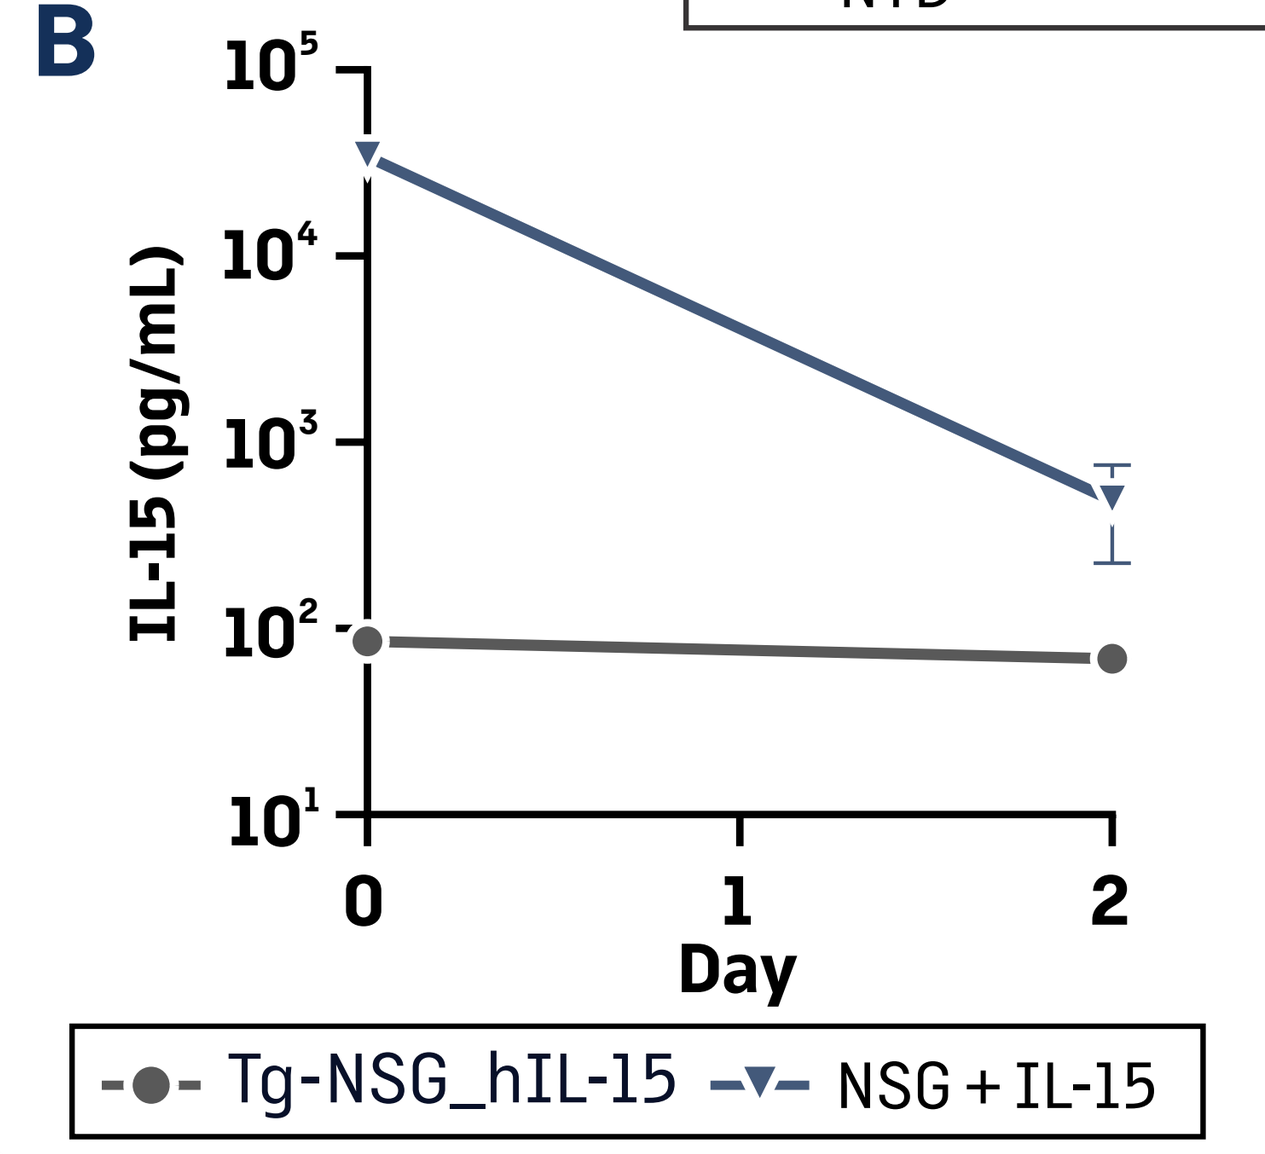
B
IL-15 (pg/mL)
Day
Tg-NSG_hIL-15
NSG + IL-15

HER2-TAC γδ T Cells Demonstrate Efficacy in Mice Expressing Human IL-15

A. NSG and transgenic Tg-NSG_hIL-15 mice inoculated with N87$^{CLDN18.2}$ tumor cells were treated with 12 x 10^6 HER2-TAC γδ T cells or NTD control. A no treatment (NT) group was included as a negative control. NSG mice were supplemented with IL-15 starting 1-hour pre-ACT, then 3 times per week thereafter. Mean tumor volumes are shown ± SEM. **B.** IL-15 serum levels were measured for Tg-NSG_hIL-15 mice, and 1-hour and 2-days post-cytokine injection for NSG mice. All mice were tumor bearing, but received no ACT. Data shown as mean ± SEM.

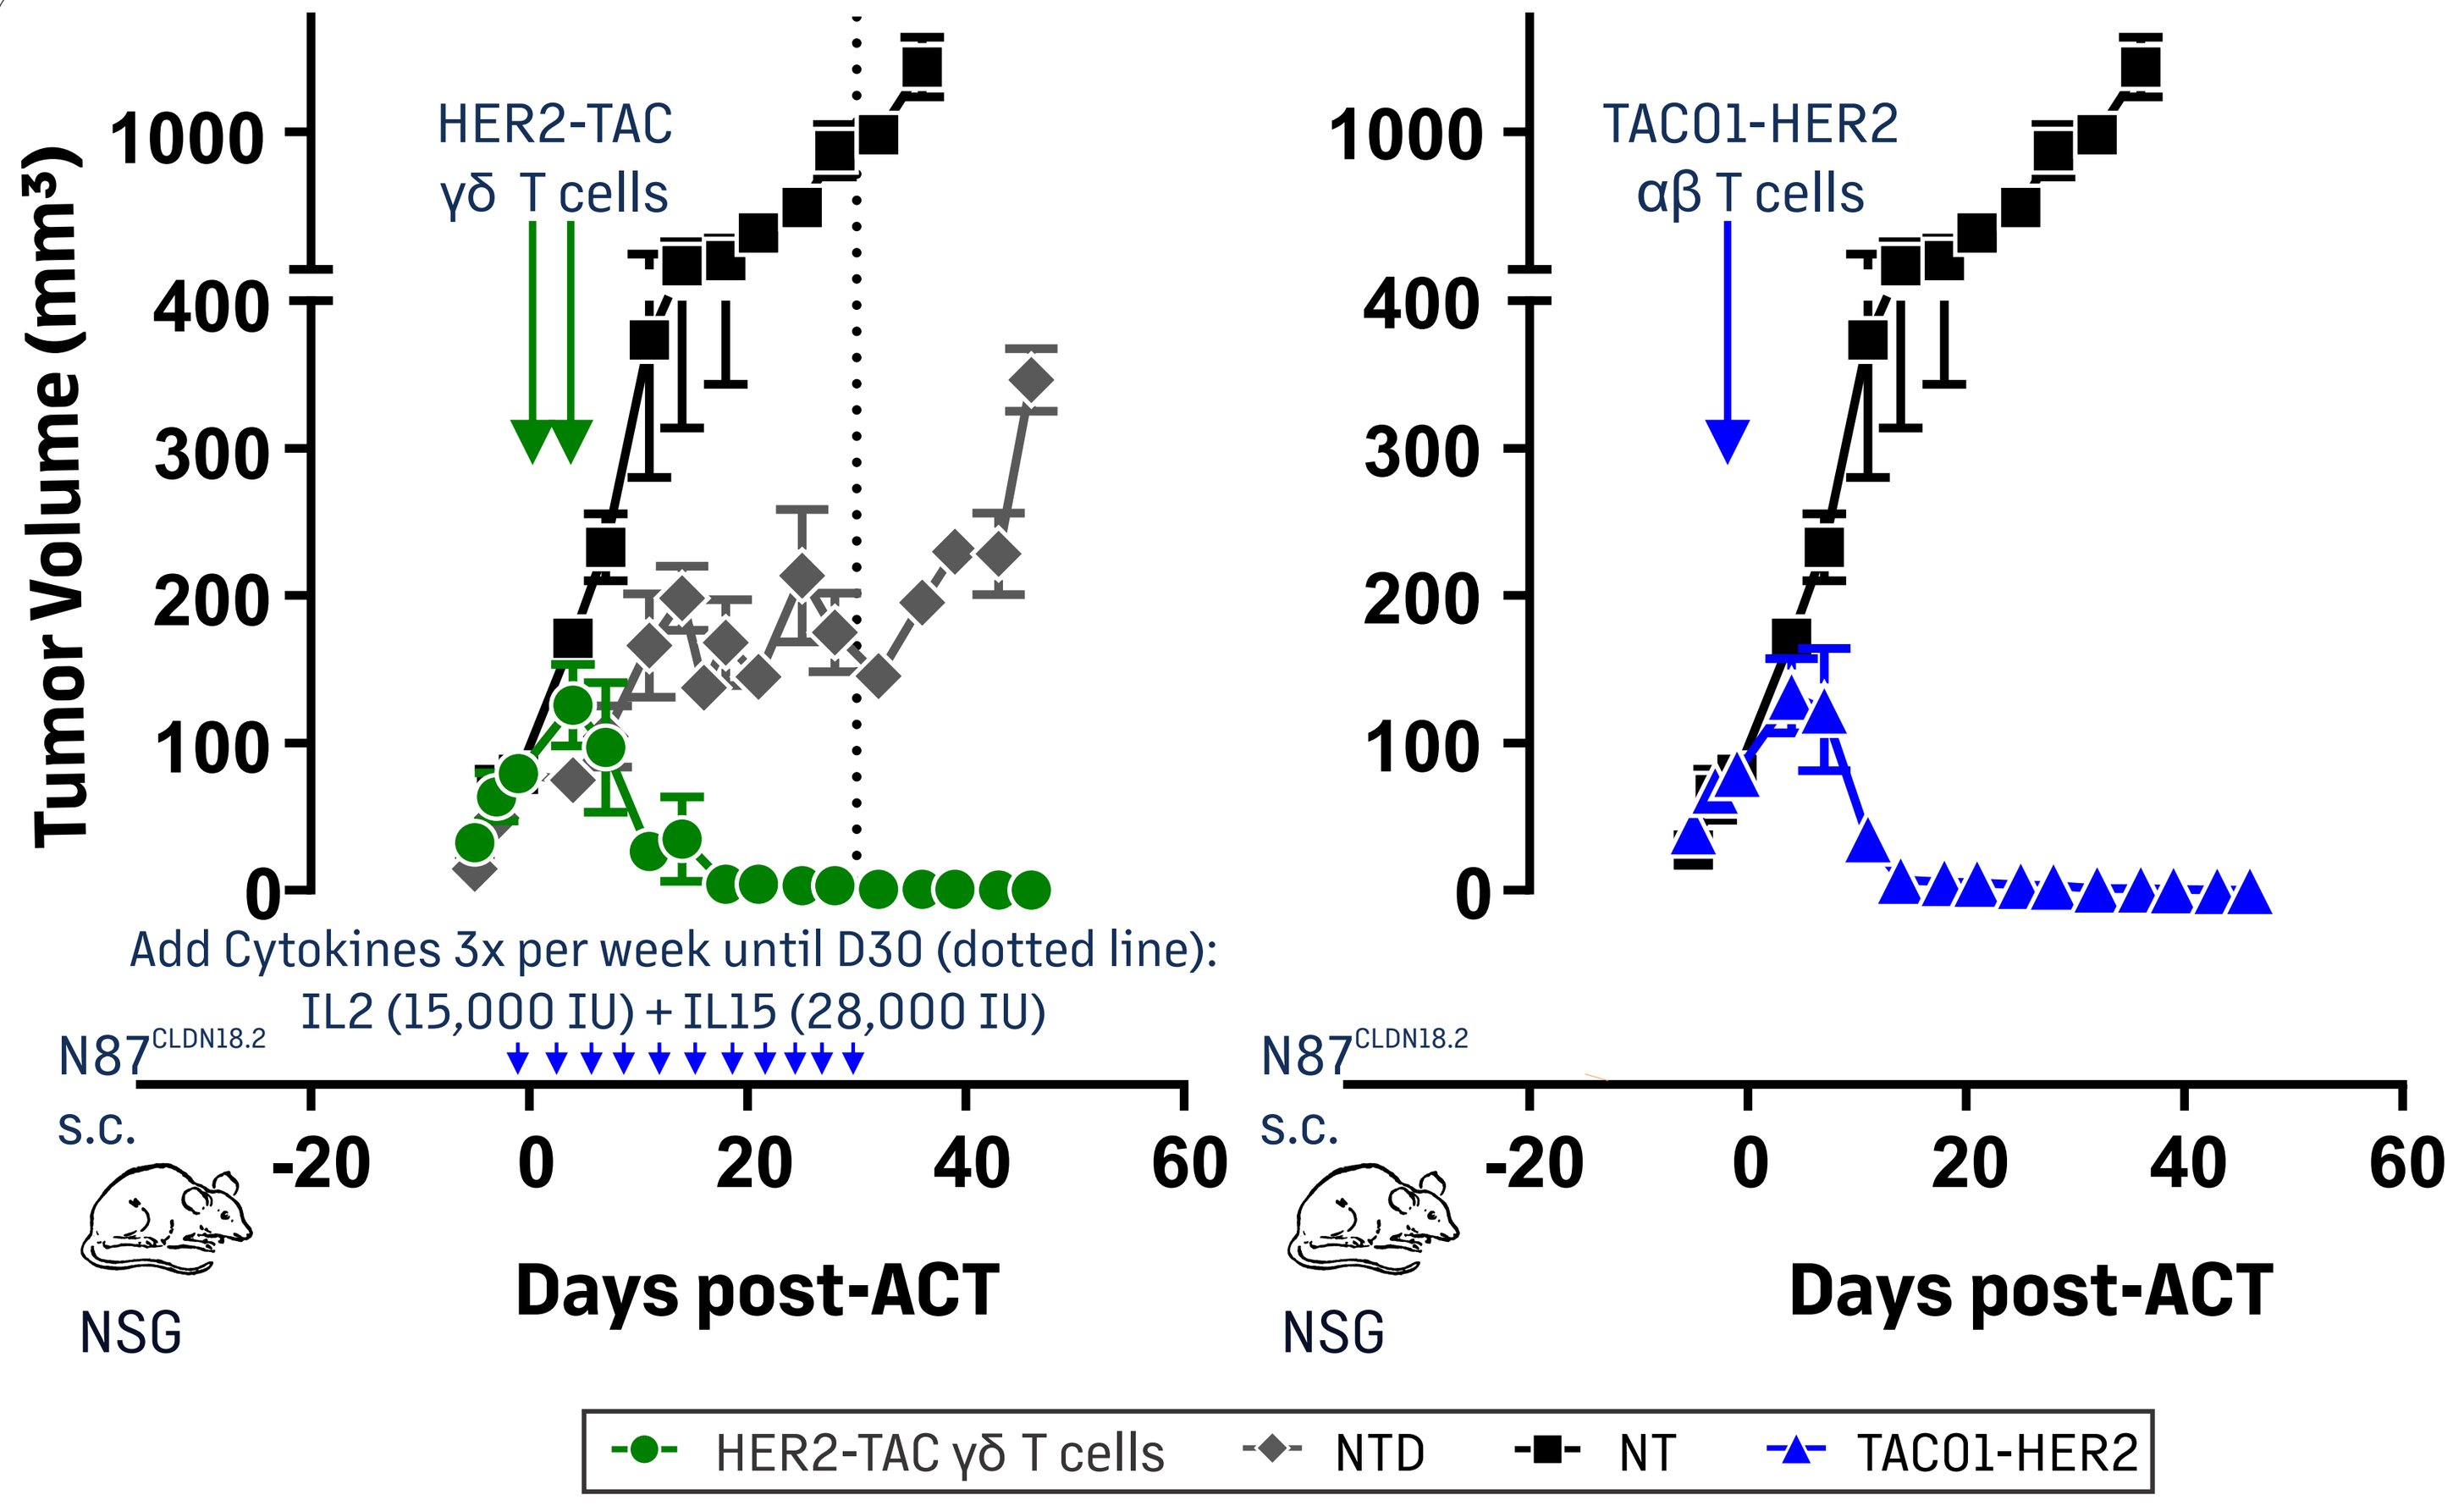
Tumor Volume (mm³)
HER2-TAC γδ T cells
TAC01-HER2 αβ T cells
Add Cytokines 3x per week until D30 (dotted line):
IL2 (15,000 IU) + IL15 (28,000 IU)
N87$^{CLDN18.2}$
s.c.
NSG
Days post-ACT
HER2-TAC γδ T cells
NTD
NT
TAC01-HER2

HER2-TAC γδ T Cells Demonstrate Robust Efficacy Against Solid Tumors In Vivo

NSG mice inoculated with N87$^{CLDN18.2}$ tumor cells were treated with 12 x 10^6 HER2-TAC γδ T cells or non-transduced (NTD) control as a split dose administered 24-hours apart (D0 and D1 post-ACT). TAC01-HER2 αβ T cells (positive control) were administered as a single dose of 6 x 10^6 on D0. A no treatment (NT) group was included as the negative control. Mean tumor volumes are shown ± SEM.

Summary

- **HER2-TAC γδ T cells are selectively activated in the presence of HER2-positive tumor cells**
- **HER2-TAC γδ T cells display strong cytotoxicity towards HER2-positive tumor cells in co-culture assays**
- **HER2-TAC γδ T cells effectively eradicate HER2-expressing tumors in vivo**