EVIDENCE FOR DURABLE ANTI-TUMOR RESPONSES BY TAC-T CELLS IN PRECLINICAL MODELS OF SOLID TUMORS

Heather L. MacGregor[1], Duane Moogk[2], Stacey X. Xu[1], Carly F. Graham[2], Joanne A. Hammill[2], Swati Shetty[1], Kyle MacDonald[1], Ling Wang[1], Laura M. Shaver[1], Sailaja Pirati[1], Philbert Ip[1], Prabha Lal[1], Christopher W. Helsen[1], Jonathan L. Bramson[1,2], Sadhak Sengupta[1], Andreas G. Bader[1] 1. Triumvira Immunologics, Austin, TX, USA & Hamilton, ON, Canada 2. McMaster University, Hamilton, ON, Canada

Abstract

Background

T cell antigen coupler (TAC) is a chimeric receptor that redirects T cells (TAC-T) towards surface-expressed tumor antigens to create safe and durable anti-cancer immune responses. The TAC activates T cells by co-opting the endogenous T cell receptor machinery via a CD3ε-specific binding motif and a cytoplasmic co-receptor tail. TAC01-HER2, a first-in-class TAC-T product targeting HER2 (ERBB2), has entered a phase I/II clinical trial. Here, we have characterized the fate of TAC-T cells during anti-tumor responses in vitro and in vivo.

Materials and Methods

In vitro, HER2-specific TAC-T products were challenged with HER2-expressing and HER2-negative tumors cells. Kinetics of T cell proliferation, degranulation, activation, differentiation, and memory generation was assessed by flow cytometry. TAC-T products were subjected to multiple rounds of tumor cell exposure in vitro to test the durability of the T-cell-mediated immune response. Bioinformatic clustering analysis of flow cytometry data was performed to identify T cell populations and track them over time.T cell expansion in blood, tumor, bone marrow, and spleen were evaluated in vivo after primary xenograft tumor treatment and secondary tumor rechallenge. Tumor- and spleen-infiltrating or circulating T cells were phenotyped by flow cytometry after treatment with TAC-T cells.

Results

Co-culture studies revealed that TAC-T products become rapidly activated and degranulate upon contact with HER2-expressing, but not HER2-negative, cell lines. Activation coincided with rapid downregulation of the TAC receptor. A large proportion of the T cells expressed activation markers, and a majority of these also expressed degranulation markers, indicating ongoing cytotoxicity. In vitro and in vivo studies demonstrated a CD8-biased response characterized by a considerable expansion in the activated CD8 population enriched at the tumor site. Later, activation and differentiation markers returned to baseline concurrently with the re-emergence of surface TAC expression, initiating T cell proliferation. Importantly, central memory T cells were expanded, and stem-like cells were maintained, suggesting strong self-renewal potential. In vitro serial cytotoxicity assays showed that TAC-T products could repeatedly kill tumor cells up to 12 rounds over 40 days. In tumor rechallenge experiments, a single dose of TAC-T cells expanded to clear solid tumor xenografts and protected mice from a second tumor challenge 30 days post initial tumor clearance, indicating long-lasting T cell persistence.

Conclusion

The TAC-T product mounts an effective anti-tumor response in multiple preclinical models, comprising activated TAC-T cells that do not become terminally exhausted but are dominated by an activated CD8 response and supported by the expansion of a memory population, indicating robust self-renewal capacity.

TAC Science

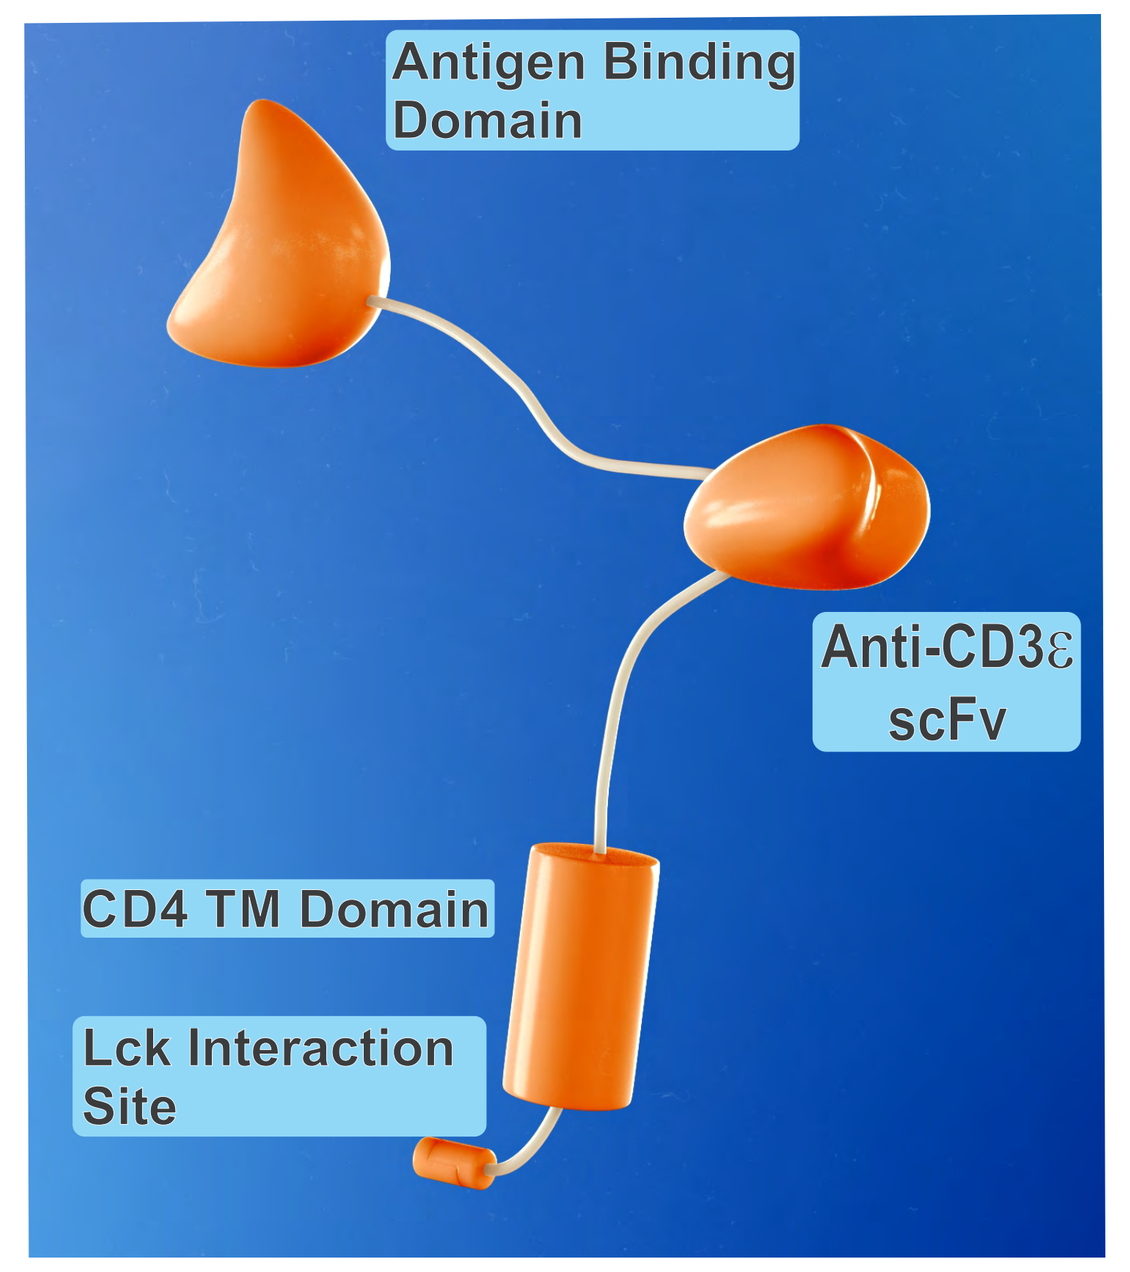
Antigen Binding Domain
Anti-CD3ε scFv
CD4 TM Domain
Lck Interaction Site

Key features of TAC technology:

- TAC functions independently of MHC
- TAC activates T cells via the endogenous TCR
- TAC incorporates the co-receptor and recruits the TCR complex, mimicking natural TCR activation

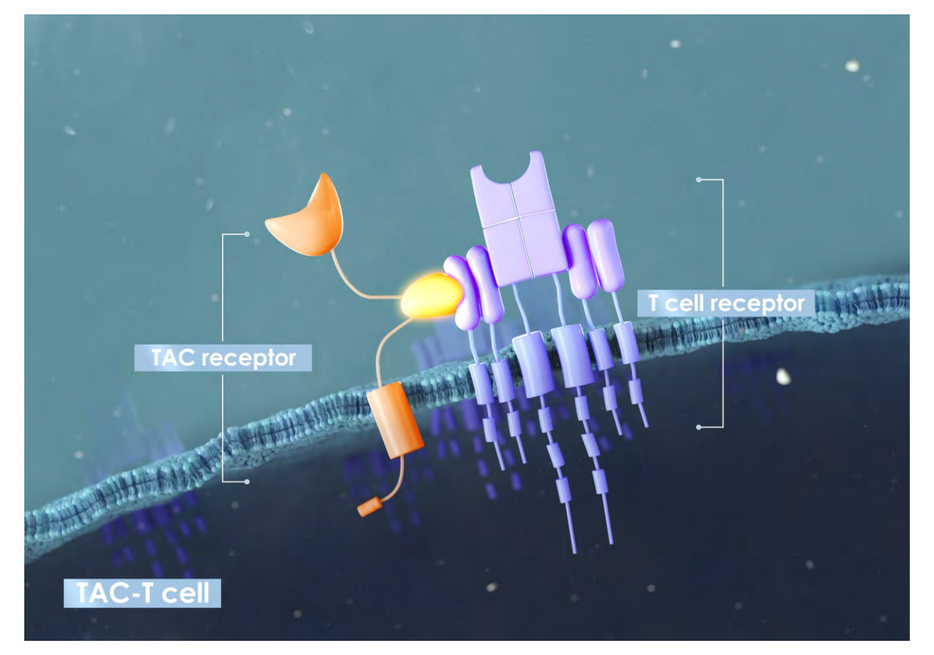
TAC receptor
T cell receptor
TAC-T cell

The membrane-bound TAC receptor interacts directly with the TCR-CD3 epsilon domain and...

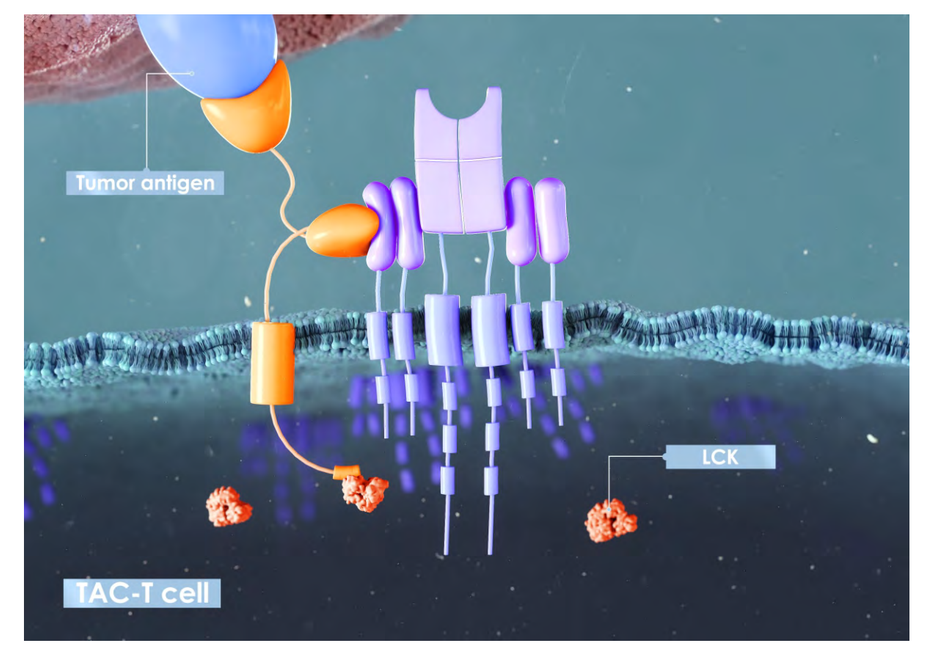
Tumor antigen
LCK
TAC-T cell

...binds directly the targeted tumor antigen. Clustering of TAC-TCR complexes leads to recruitment of kinases (Lck) via the cytoplasmic co-receptor domain and...

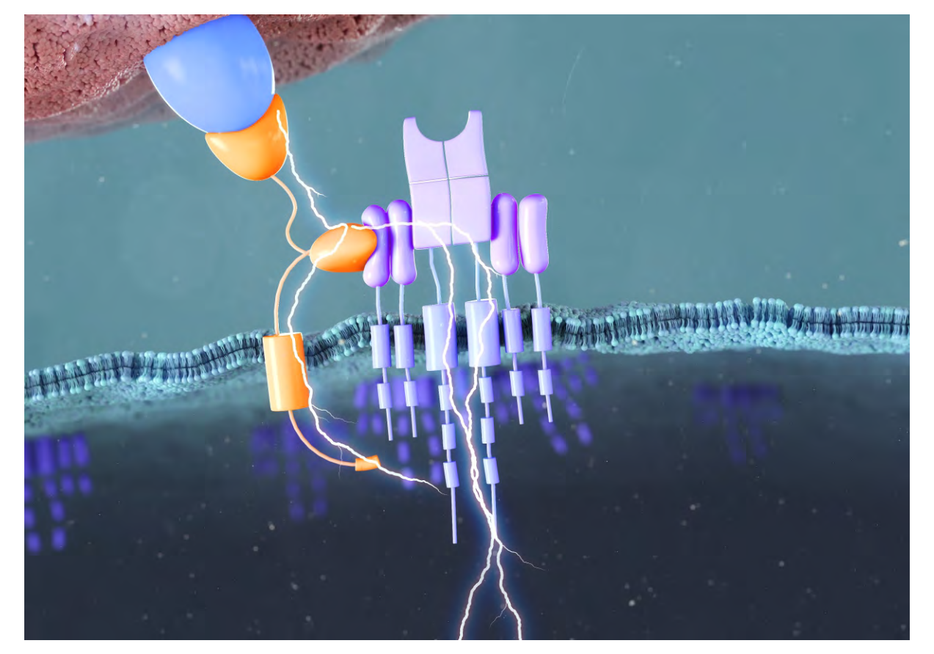

...initiates T cell activation via the endogenous CD3-TCR complex.

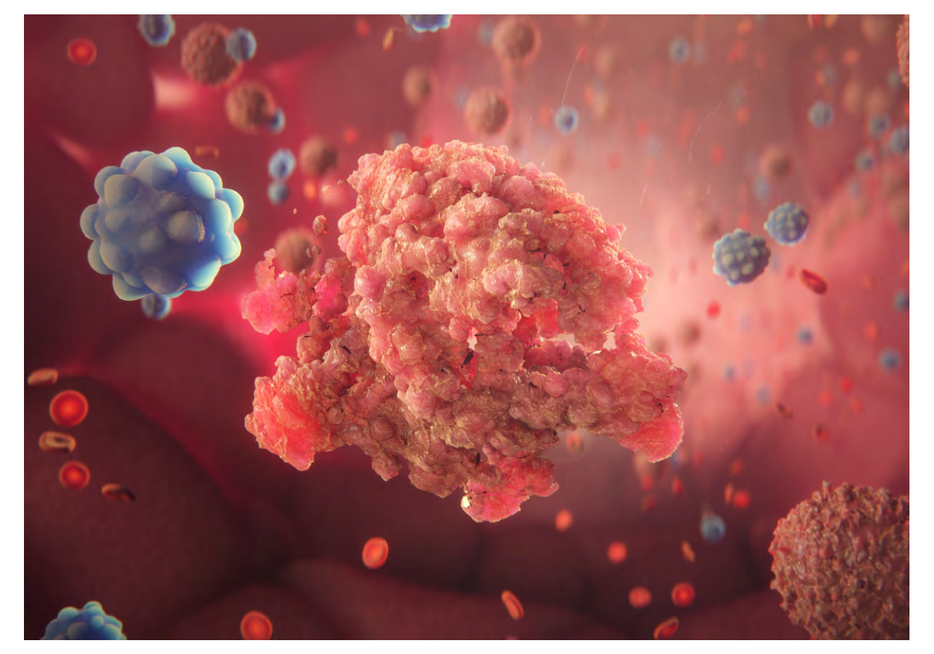

This results in effective cell lysis of multiple tumor cells during multiple killing events.

Watch a short animation to understand the TAC mechanism

triumvira

Cocoon® T cell manufacturing enhances memory profile

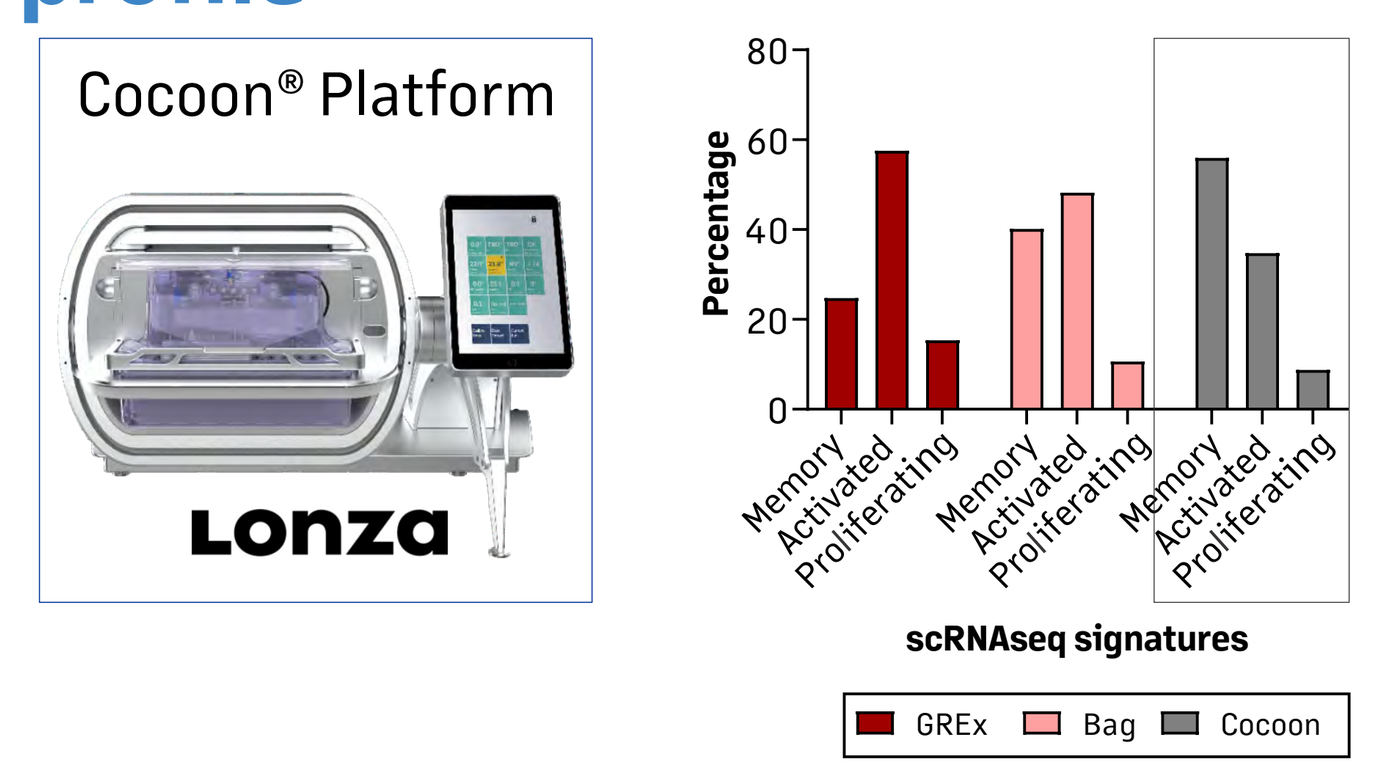
Cocoon® Platform
Lonza
Percentage
Memory
Activated
Proliferating
scRNAseq signatures
GREx
Bag
Cocoon

Figure 1: (Left) Lonza Cocoon® Platform; (Right) Bar graph representing memory (TCF7, SELL/CD62L, CCR7, CD27, LRRN3), activated (IFNG, S100A4, KLRK1, TTGB1, TNFRSF4), and proliferating (MKI67, TOP2A, HMGB2, HMGB3) gene signatures of TAC-T cells manufactured by different technologies, analyzed by scRNAseq studies.

Antigen-driven T cell activation in the absence of terminal T cell exhaustion

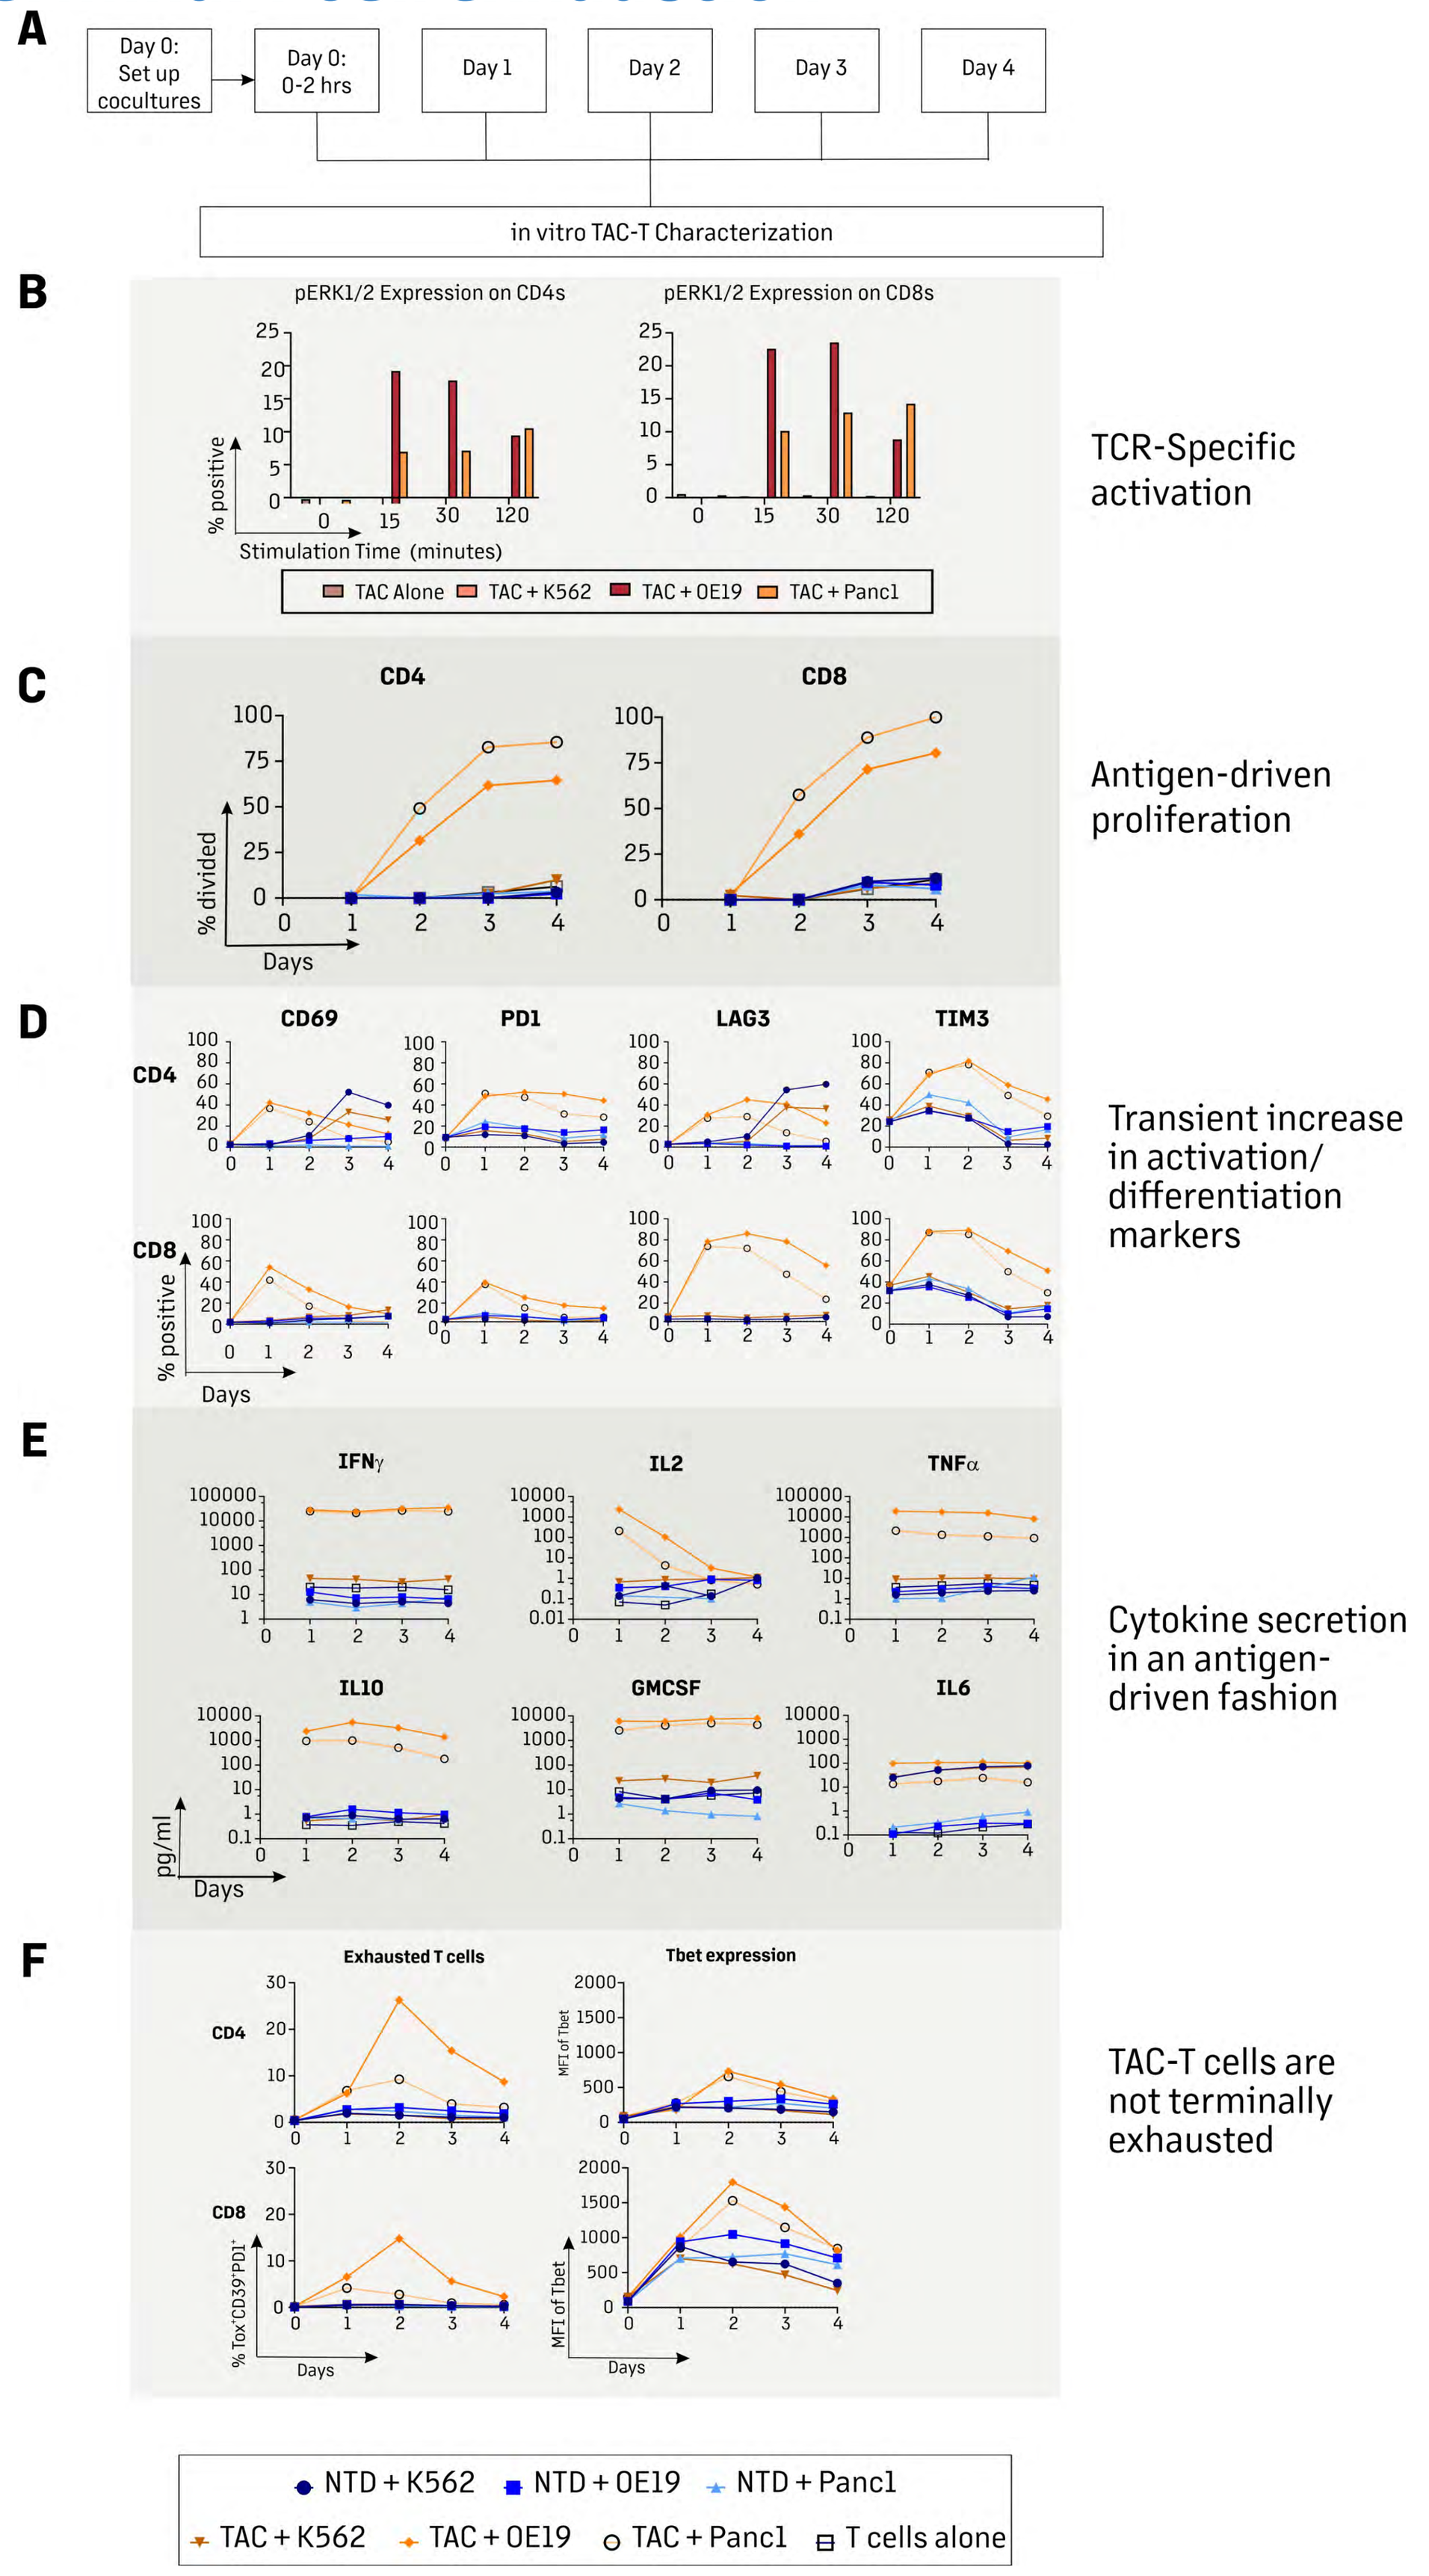
A
Day 0: Set up cocultures
Day 0: 0-2 hrs
Day 1
Day 2
Day 3
Day 4
in vitro TAC-T Characterization
B
pERK1/2 Expression on CD4s
pERK1/2 Expression on CD8s
% positive
Stimulation Time (minutes)
TAC Alone
TAC + K562
TAC + OE19
TAC + Panc1
TCR-Specific activation
C
CD4
CD8
% divided
Days
Antigen-driven proliferation
D
CD69
PD1
LAG3
TIM3
% positive
Transient increase in activation/ differentiation markers
E
IFNγ
IL2
TNFα
IL10
GMCSF
IL6
pg/ml
Cytokine secretion in an antigen-driven fashion
F
Exhausted T cells
Tbet expression
% Tox CD39 PD1
MFI of Tbet
TAC-T cells are not terminally exhausted
NTD + K562
NTD + OE19
NTD + Panc1
TAC + K562
TAC + OE19
TAC + Panc1
T cells alone

Figure 2: Antigen-driven T cell activation in absence of terminal T cell exhaustion. (**A**) Design of Experiment: Flow cytometric analysis of T cells after 0 minutes to 4 days of culture with tumor and control cells; (**B**) bar graph representing pERK1/2 levels in T cells; Line graphs representing (**C**) antigen-driven T cell proliferation measured by CTV® dilution, (**D**) surface expression of T cell activation markers. (**E**) Intracellular cytokine levels, and (**F**) terminal exhaustion markers and intracellular T-bet levels.

TSCM cell fraction persists after stimulation

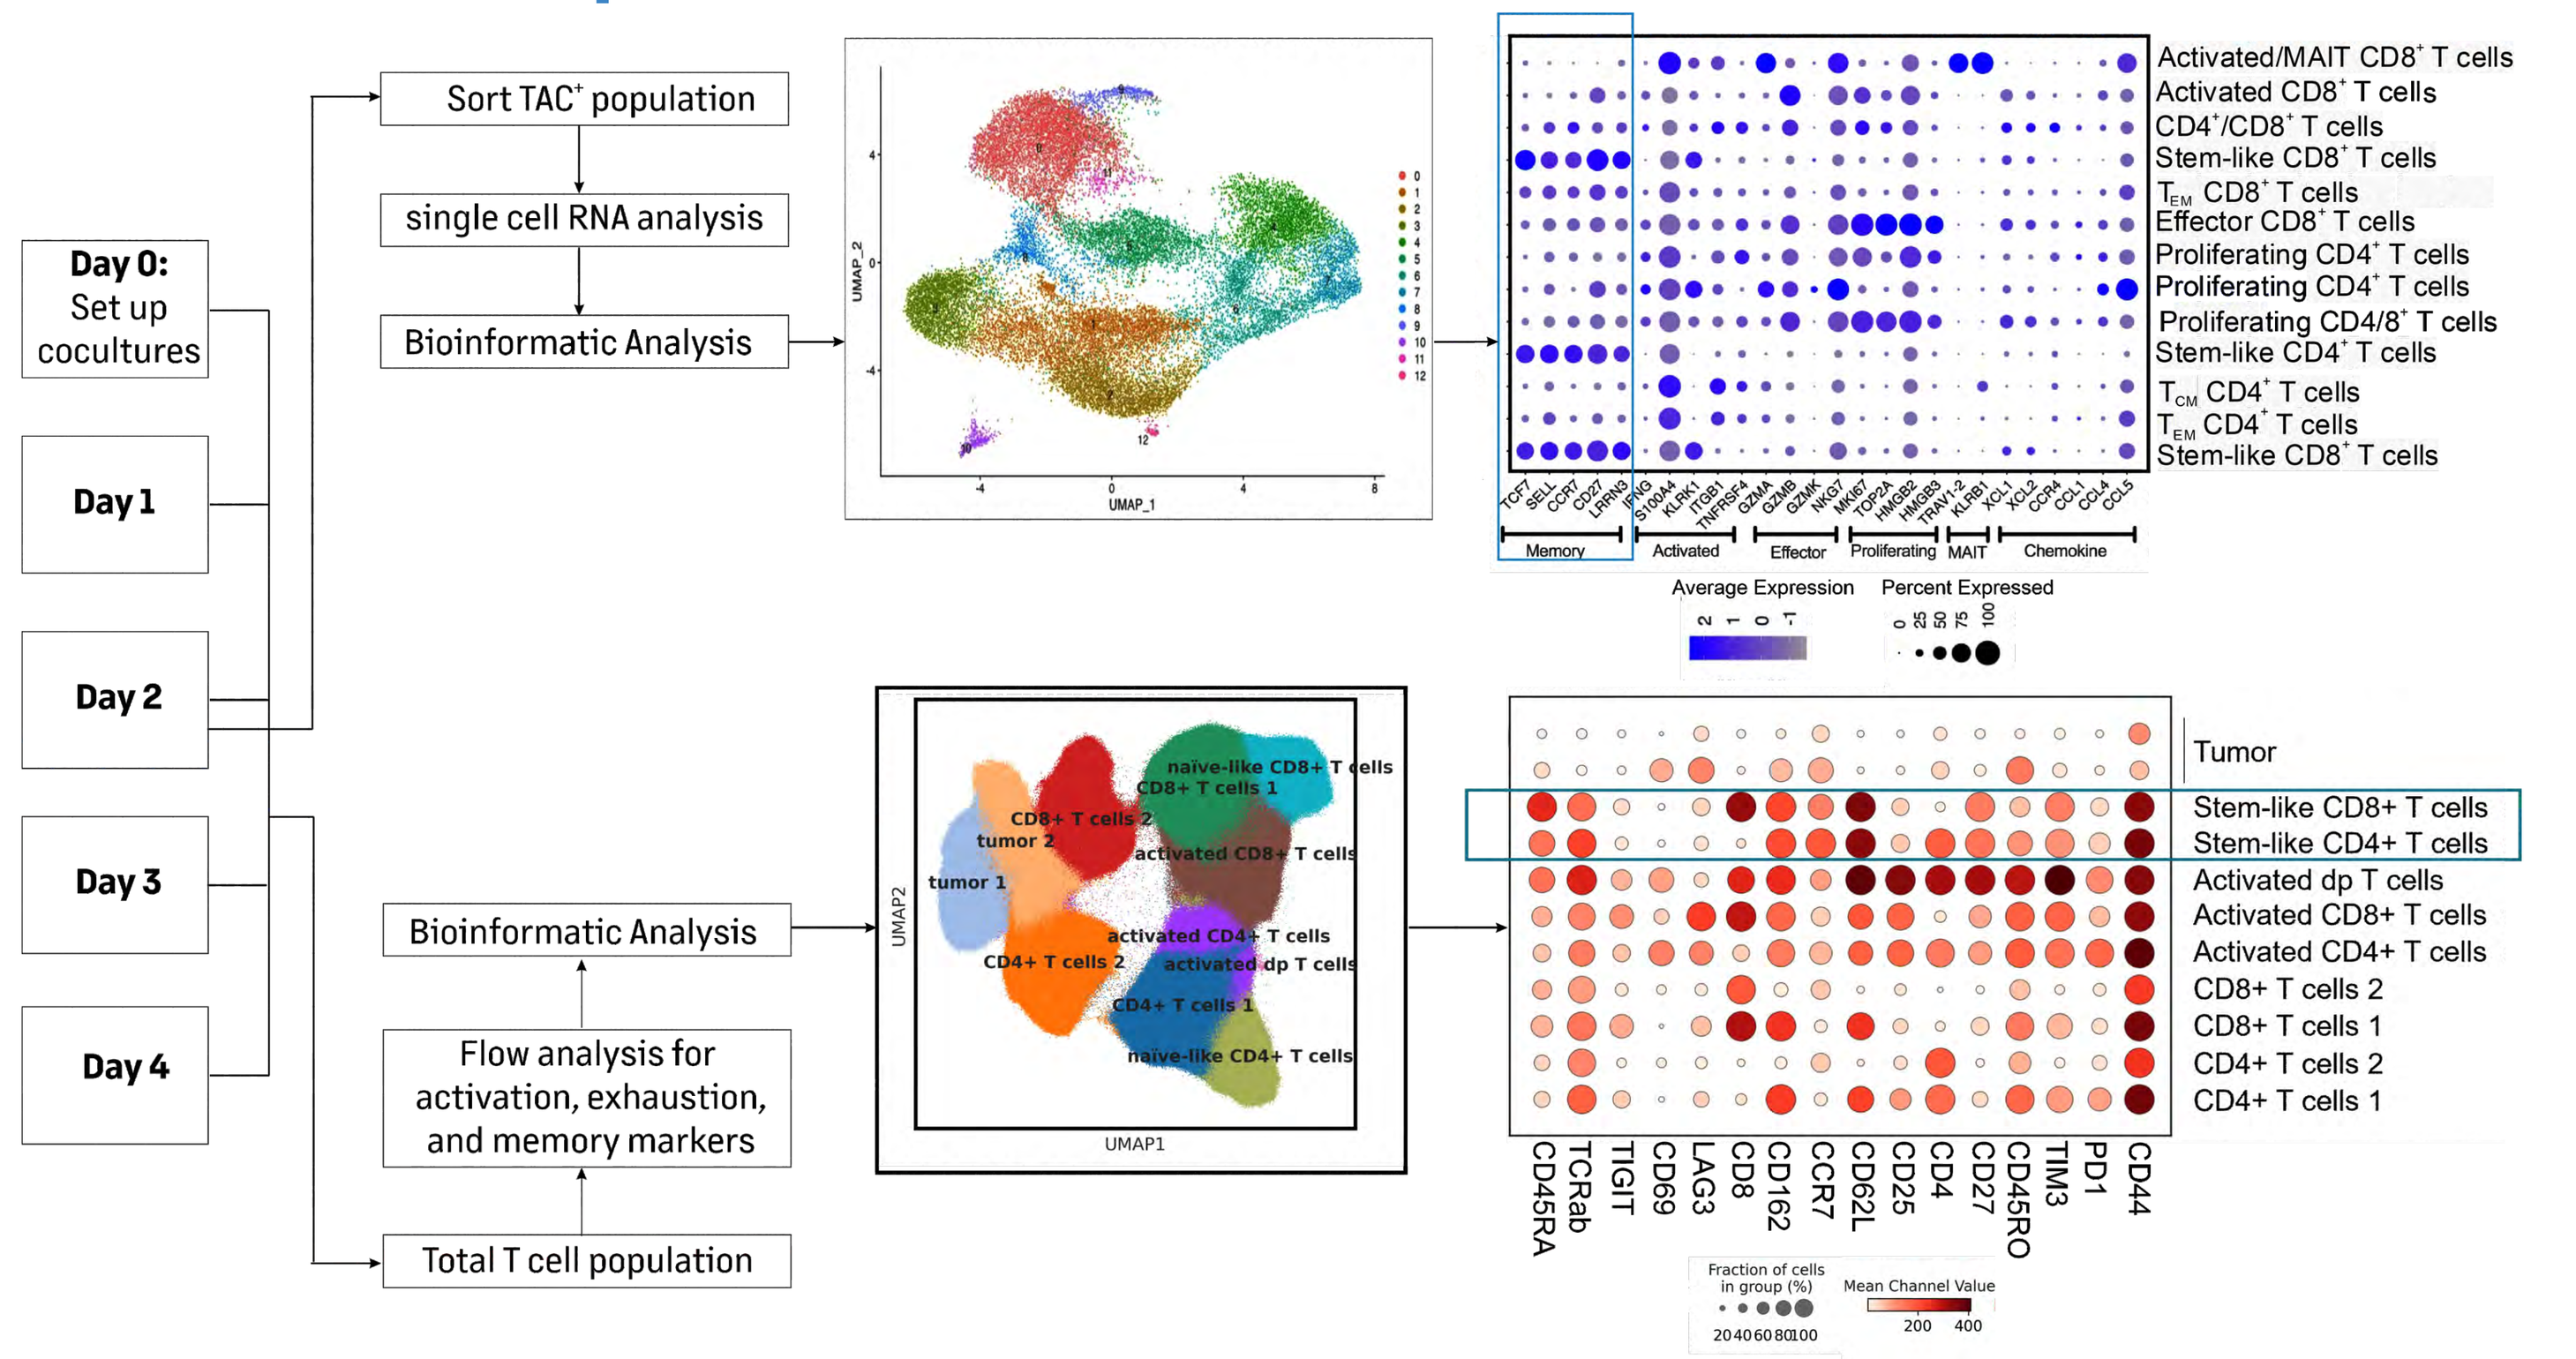
Day 0: Set up cocultures
Day 1
Day 2
Day 3
Day 4
Sort TAC+ population
single cell RNA analysis
Bioinformatic Analysis
Flow analysis for activation, exhaustion, and memory markers
Total T cell population
UMAP_1
UMAP_2
Activated/MAIT CD8+ T cells
Activated CD8+ T cells
CD4+/CD8+ T cells
Stem-like CD8+ T cells
Effector CD8+ T cells
Proliferating CD4+ T cells
Proliferating CD4/8+ T cells
Stem-like CD4+ T cells
Memory
Activated
Effector
Proliferating
MAIT
Chemokine
Average Expression
Percent Expressed
Tumor
Stem-like CD8+ T cells
Stem-like CD4+ T cells
Activated dp T cells
Activated CD8+ T cells
Activated CD4+ T cells
CD8+ T cells 2
CD8+ T cells 1
CD4+ T cells 2
CD4+ T cells 1
CD45RA
TCRab
TIGIT
CD69
LAG3
CD8
CD162
CCR7
CD62L
CD25
CD4
CD27
CD45RO
TIM3
PD1
CD44
Fraction of cells in group (%)
Mean Channel Value

Figure 3: Persistent TSCM fraction during TAC-T target engagement. (Top) Bioinformatic analysis of TAC-T scRNA signature clusters and dot plots from 48 hr co-cultures with tumor cells. (Bottom) T cell phenotype clusters and dot plot from 17-color complex flow cytometric analysis of cells after 0-4 days of coculture with tumor cells.

Tumor-specific activation and persistent circulating TSCM cells in xenograft tumor model

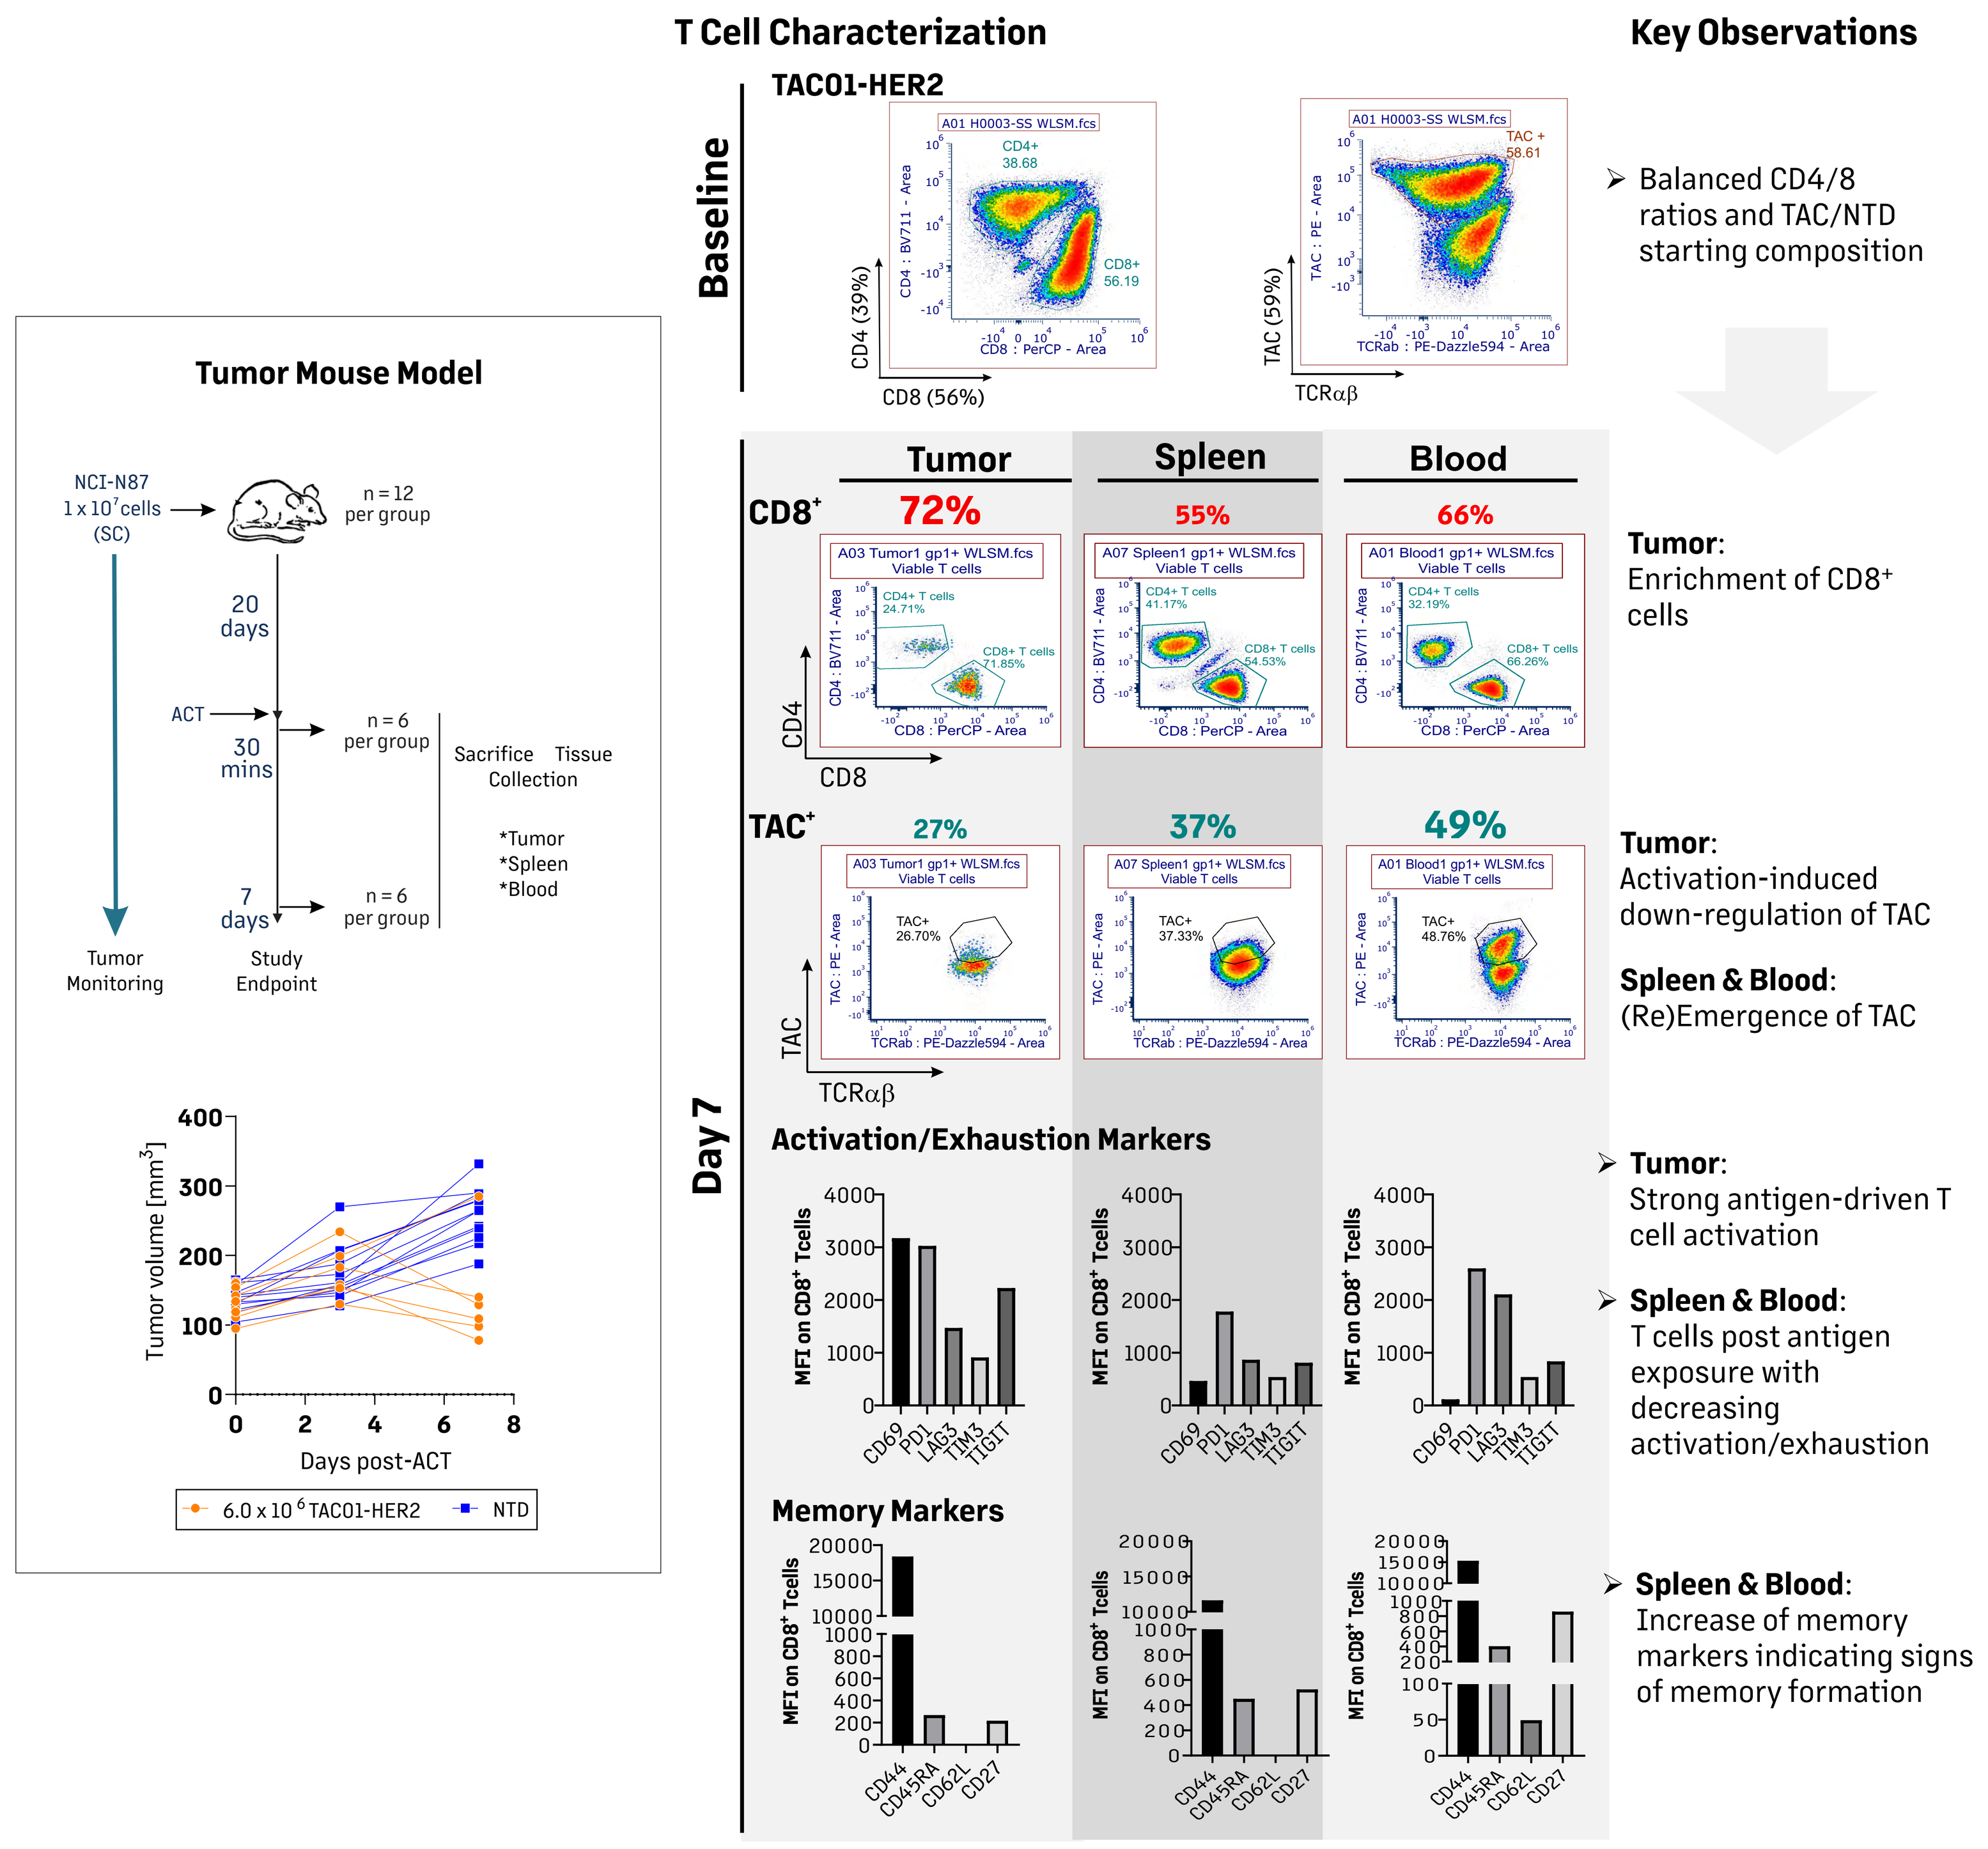
Tumor Mouse Model
NCI-N87 1 x 10^7 cells (SC)
n = 12 per group
20 days
ACT
30 mins
n = 6 per group
7 days
n = 6 per group
Sacrifice Tissue Collection
*Tumor
*Spleen
*Blood
Tumor Monitoring
Study Endpoint
Tumor volume [mm3]
Days post-ACT
6.0 x 10^6 TAC01-HER2
NTD
T Cell Characterization
TAC01-HER2
Baseline
CD4 (39%)
CD8 (56%)
TAC (59%)
TCRαβ
Day 7
Tumor
Spleen
Blood
CD8+
72%
55%
66%
TAC+
27%
37%
49%
Activation/Exhaustion Markers
MFI on CD8+ Tcells
CD69
PD1
LAG3
TIM3
TIGIT
Memory Markers
CD44
CD45RA
CD62L
CD27
Key Observations
Balanced CD4/8 ratios and TAC/NTD starting composition
Tumor: Enrichment of CD8+ cells
Tumor: Activation-induced down-regulation of TAC
Spleen & Blood: (Re)Emergence of TAC
Tumor: Strong antigen-driven T cell activation
Spleen & Blood: T cells post antigen exposure with decreasing activation/exhaustion
Spleen & Blood: Increase of memory markers indicating signs of memory formation

Figure 4: Tumor-specific activation and persistent circulating TSCM fraction during in vivo tumor rejection. (Left) Design of in vivo experiment and line graph representing in vivo tumor growth. (Right) Key in vivo T cell phenotype in descending order: baseline CD4/CD8 ratio and surface TAC expression; CD4/CD8 ratio and surface TAC expression on T cells harvested from tumor, spleen, and blood 7 days after adoptive cell therapy (ACT); bar graph representing activation/exhaustion markers; bar graph representing memory markers.

triumvira

Durable anti-tumor responses

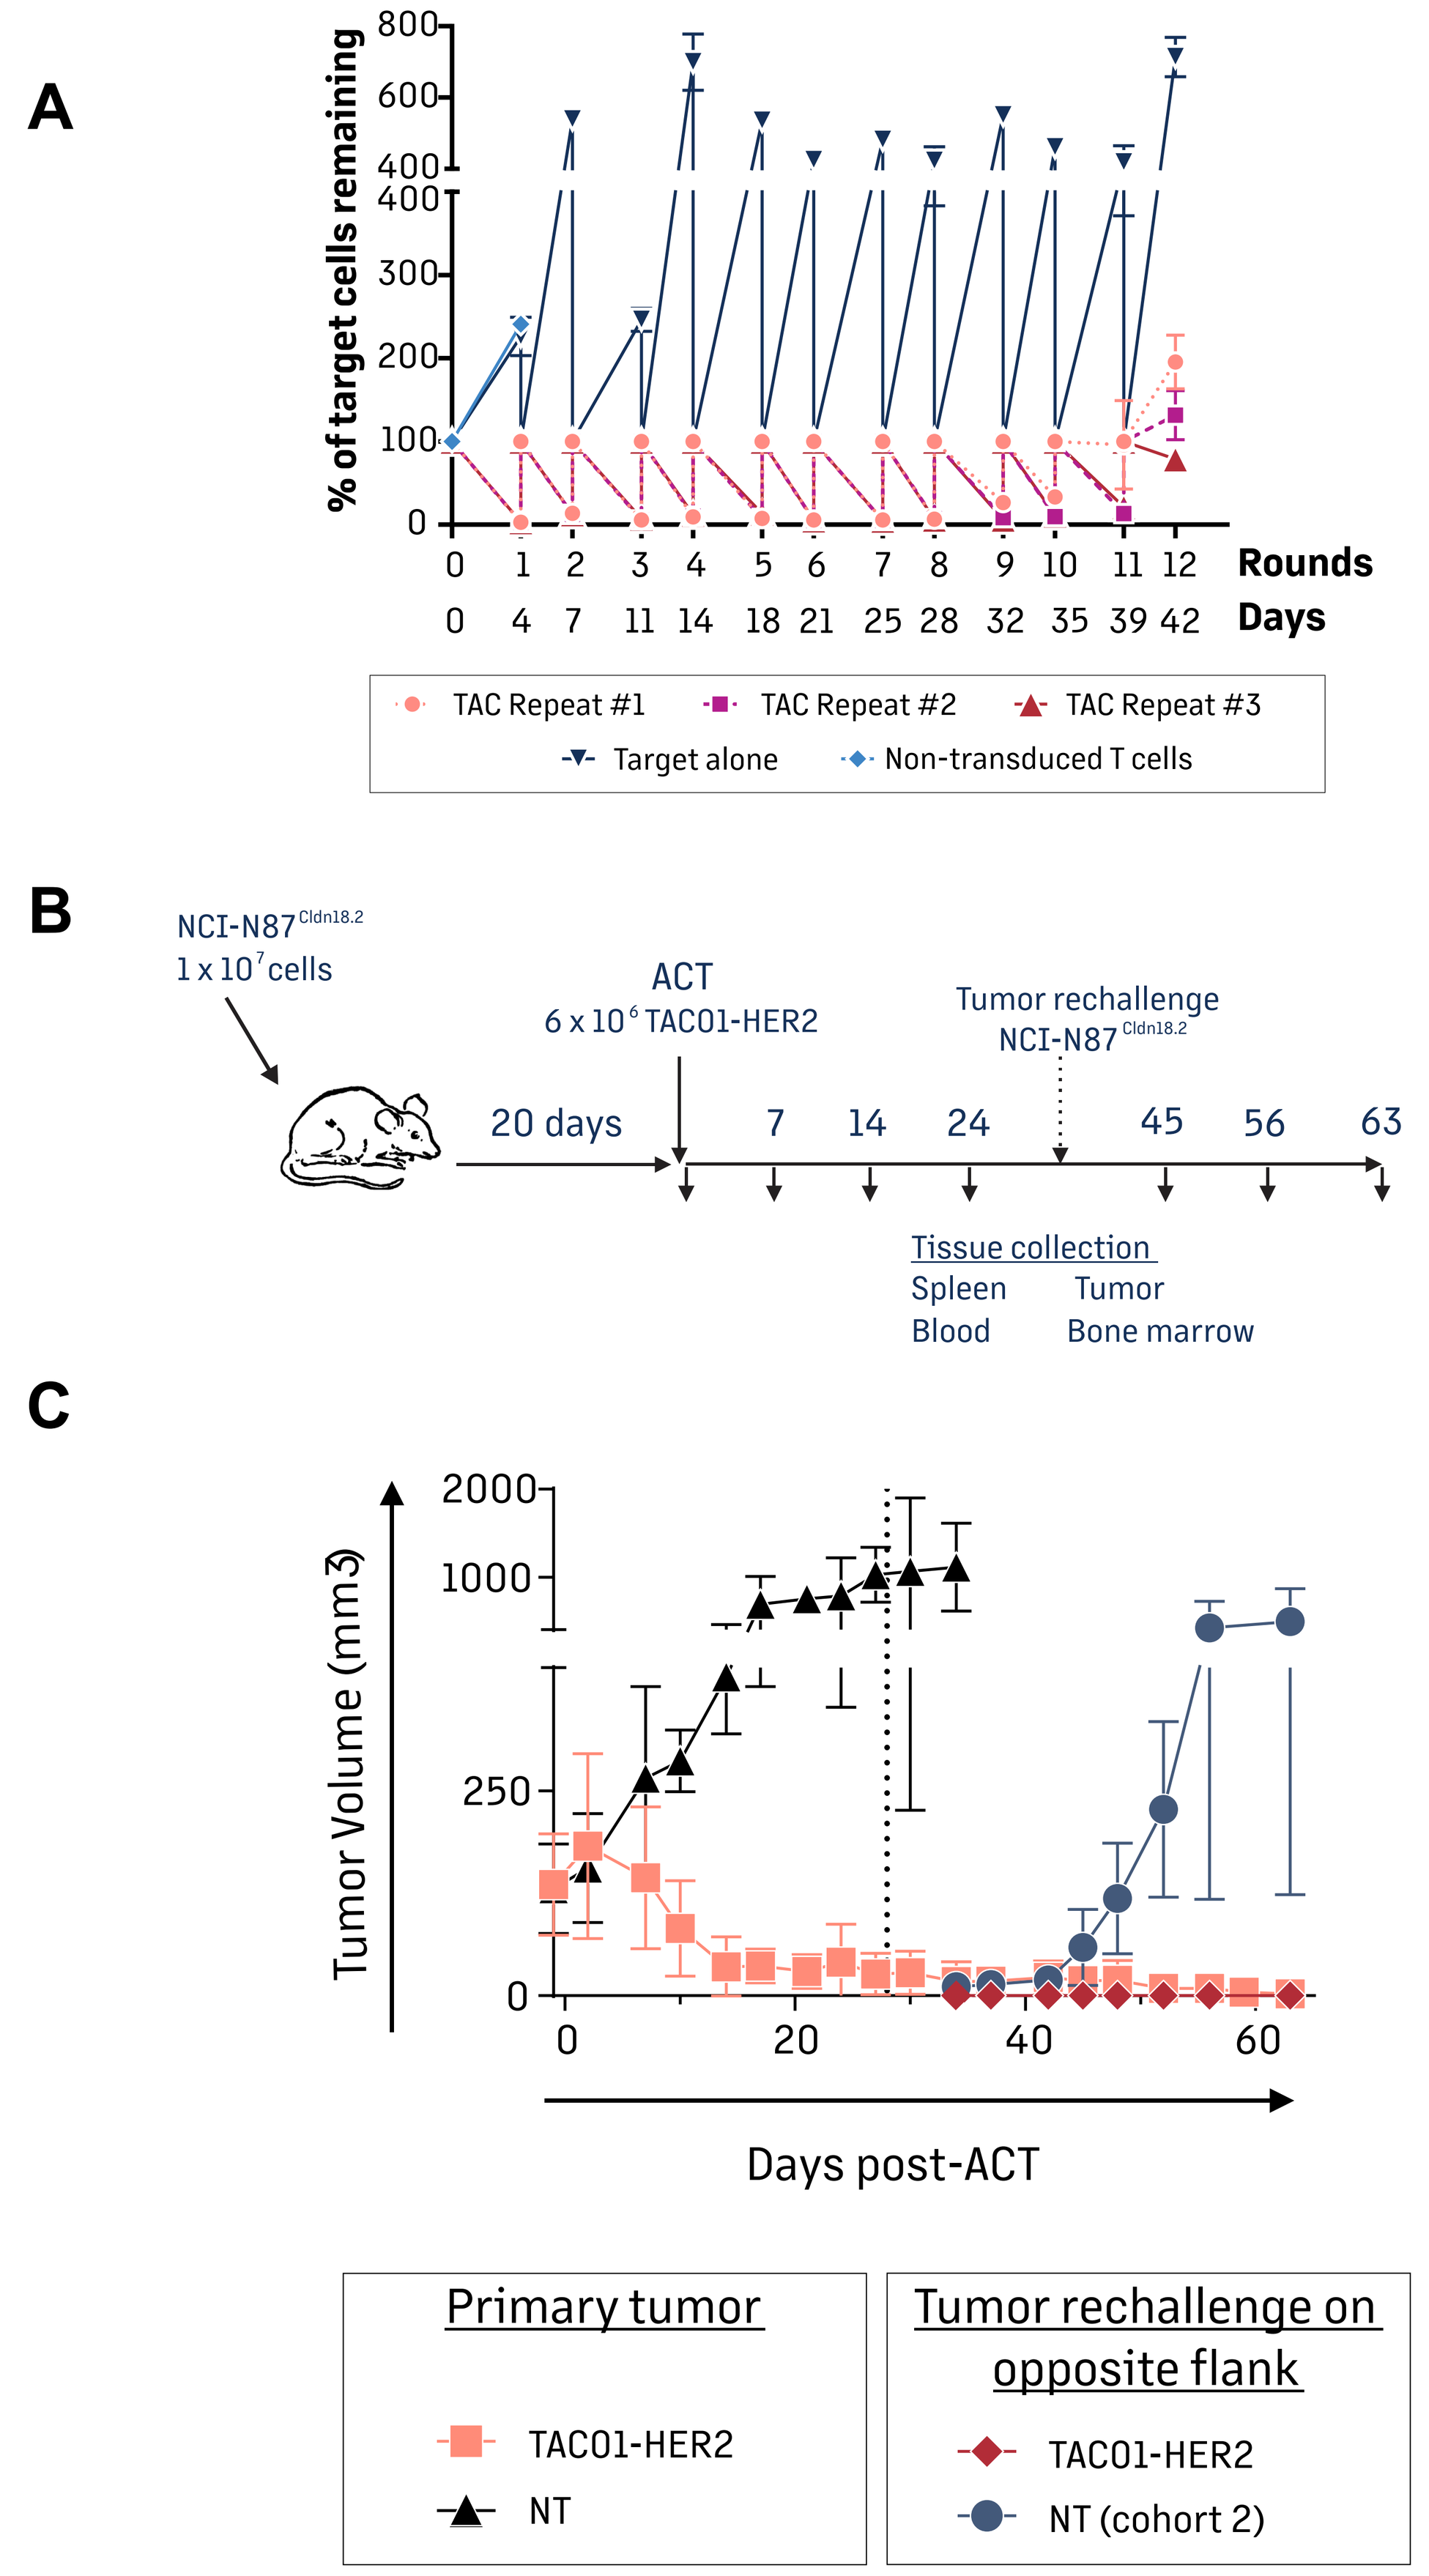
A
% of target cells remaining
Rounds
Days
TAC Repeat #1
TAC Repeat #2
TAC Repeat #3
Target alone
Non-transduced T cells
B
NCI-N87 Cldn18.2
1 x 10^7 cells
ACT
6 x 10^6 TAC01-HER2
Tumor rechallenge
NCI-N87 Cldn18.2
20 days
Tissue collection
Spleen
Blood
Tumor
Bone marrow
C
Tumor Volume (mm3)
Days post-ACT
Primary tumor
TAC01-HER2
NT
Tumor rechallenge on opposite flank
TAC01-HER2
NT (cohort 2)

Figure 5: TAC-T cells elicit durable anti-tumor responses. (**A**) Line graph depicting the survival curve of tumor cells following exposure to TAC-T cells in an in vitro repeat killing assay; (**B**) Design of in vivo tumor rechallenge experiment in MHC I/II-DKO xenograft model; (**C**) Line graph representing in vivo tumor growth.

Summary

- TAC-T products are marked by a significant fraction of **stem-like memory cells (TSCM), enhanced by Cocoon® technology**
- **TSCM cells persist** during TAC-T cell target engagement
- Tumor-specific TAC-T activation leads to **persistent TSCM cell fraction** during tumor rejection in vivo
- TAC-T cells elicit **durable anti-tumor responses** in vitro and in vivo