A PHASE I/II TRIAL INVESTIGATING SAFETY AND EFFICACY OF AUTOLOGOUS TAC T CELLS TARGETING HER2 IN RELAPSED OR REFRACTORY SOLID TUMORS (TACTIC-2)

Benjamin L. Schlechter, MD,[1] Daniel Olson, MD,[2] Samuel Saibil, MD,[3] Brooke Pieke,[2] Riemke Bouvier,[1] Mridula A. George, Md[6], Kelly Gruber[6] Nathan Ternus,[5] Maria Apostolopoulou,[5] Kara Moss,[5] Deyaa Adib, MD,[5] Ecaterina E. Dumbrava, MD,[4]

[1] Dana Farber Cancer Institute, Boston, MA, [2] University of Chicago, Chicago, IL, [3] Princess Margaret Cancer Centre, Toronto, ON, CA, [4] MD Anderson Cancer Center, Houston, TX, [5] Triumvira Immunologics, Austin, TX, [6] Rutgers Cancer Institute of New Jersey

INTRODUCTION

- The T cell antigen coupler (TAC) is a novel, proprietary chimeric receptor that facilitates the re-direction of T cells to tumor cells and activates T cells by co-opting the endogenous T cell receptor complex, with the goal to elicit a safe and durable anti-tumor response. In preclinical models of cancer, TAC-engineered T cells effectively eradicate tumor cells in vitro and in vivo without toxicities typically associated with engineered T cell products.
- TACTIC-2 (NCT04727151) is an open-label, multicenter phase I/II study that aims to establish safety, maximum tolerated dose (MTD), recommended phase 2 dose (RP2D), pharmacokinetic profile, and efficacy of TAC01-HER2 in patients with HER2-positive solid tumors by immunohistochemistry that are 1+, 2+, or 3+ (i.e. breast, lung, pancreatic, colorectal, gastric, endometrial, ovarian, and others) whom have progressed on prior anti-cancer therapies.
- We present a clinical update from Cohorts 1-3 (9 participants) that highlights safety and efficacy data; the study further elucidates potential therapeutic impact to patients with HER2 overexpressed solid tumors.

Significant Unmet Need Beyond HER2+ Breast Cancer[1]

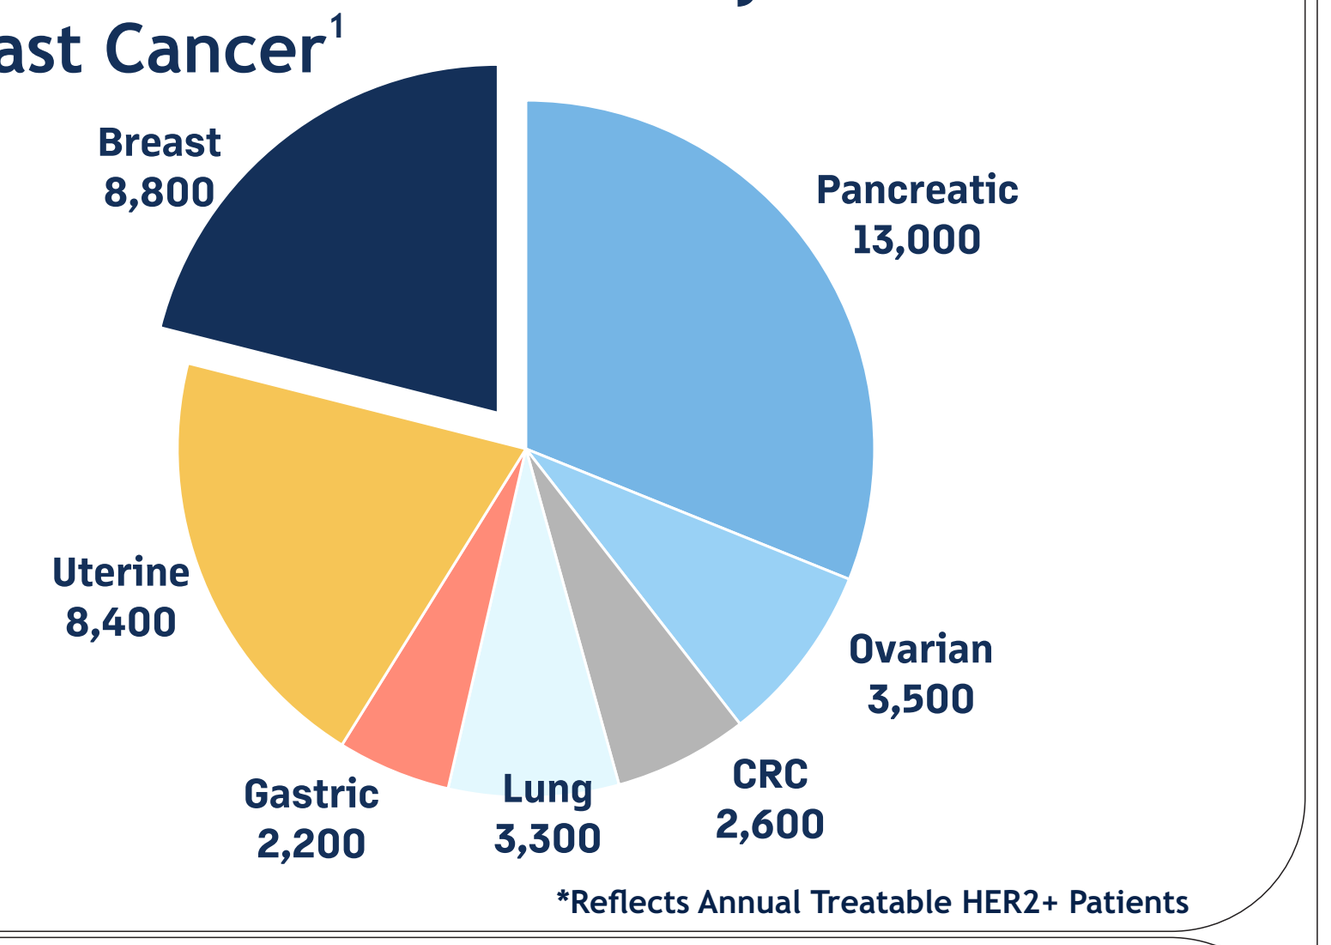
Breast
8,800
Pancreatic
13,000
Uterine
8,400
Ovarian
3,500
Gastric
2,200
Lung
3,300
CRC
2,600

*Reflects Annual Treatable HER2+ Patients

TAC SCIENCE

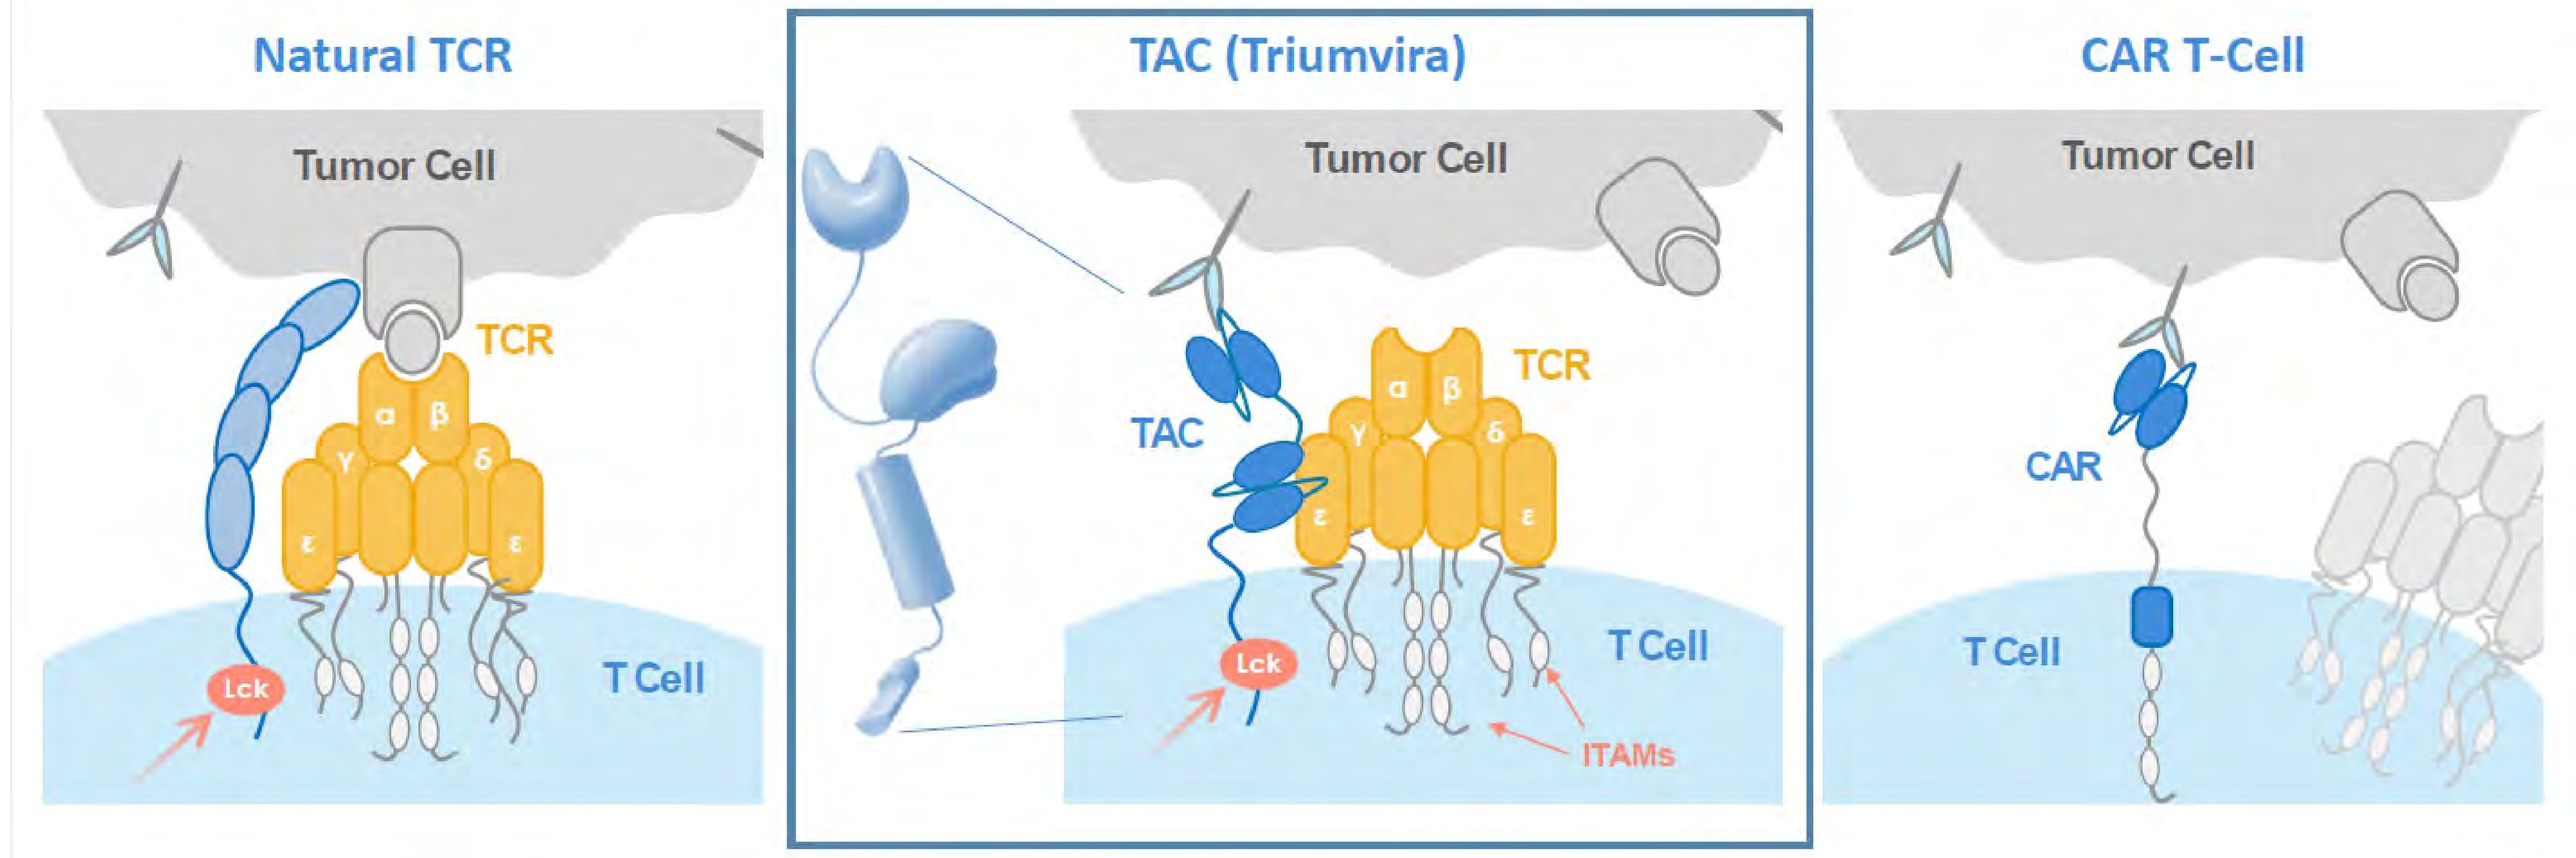
Natural TCR
Tumor Cell
TCR
Lck
T Cell
TAC (Triumvira)
Tumor Cell
TAC
TCR
Lck
T Cell
ITAMs
CAR T-Cell
Tumor Cell
CAR
T Cell

Watch a short animation to understand the TAC mechanism

TAC co-opts the natural TCR and provides the intracellular co-receptor function, mimicking normal TCR activation.

TRIAL DESIGN & MANUFACTURING

Phase I Dose Escalation				Phase II Dose Expansion		
DL 1 4 Patients Treated	DL 2 4 Patients Treated	DL 3 Patients Enrolled	DL 4 Enrolling	Group A: (1+, 2+ & 3+) HER2 Positive Breast Cancer; Planned Enrollment of 23 Patients; 2nd Line + Setting	Group B: (1+, 2+ & 3+) HER2 Positive non-Breast Cancer; Planned Enrollment of 35 Patients; 2nd Line + Setting	Group C: (1+, 2+ & 3+) HER2 Positive combination cohort with immune checkpoint inhibitor; Planned Enrollment of 35 Patients; 2nd Line + Setting
1-3 x 10^5 Cells/kg	6-8 x 10^5 Cells/kg	1-3 x 10^6 Cells/kg	6-8 x 10^6 Cells/kg			

Lonza Cocoon Eliminates Several Manufacturing Steps

Sample Collection → Sample Preparation → Selection (Optional) → Activation → Transduction → Expansion → Formulation & Release → Patient Administration

Fully Automated Manufacturing: 9 days; Vein-Vein Time: 21-24 Days

Lymphodepleting Chemotherapy:
3 consecutive days of fludarabine (Flu)* IV (30 mg/m2) and cyclophosphamide (Cy) IV (300 mg/m2) with/without Mesna IV
***due to national shortage of fludarabine, modified LDC given per institutional standard**

PHASE I TRIAL PROGRESS

Primary Endpoints	Secondary Endpoints
Safety: DLTs, MTD	Efficacy: ORR, DOR, PFS, OS, RP2D, Aes

Eligibility Criteria
Patients with advanced, metastatic, unresectable solid tumors which express HER2 after at least 2 lines of therapy, at least 1 measurable lesion per RECIST version 1.1, ECOG performance score 0-1, grade 1 or baseline for any prior treatment related toxicities.

Cohorts 1 & 2 + 1st patient in Cohort 3: Demographics and Tumor-Intrinsic Characteristis

Characteristic		Value	Characteristic		Value
Median Age (Range), by Year		64 (42-70)	Tumor Type (%)	Gastric	3 (33%)
Sex: Male/Female (%)	M	6 (66.6%)		Colorectal	3 (33%)
	F	3 (33.3%)		Gastroesophogeal Junction	2 (22%)
Race (%)	White	8 (89%)		Gall Bladder	1 (11%)
	Other	1 (11%)	Previous Anti-Cancer Therapy Median (Range)		4 (2-12)
ECOG PS (%)	0	5 (56%)	Previous Lines of HER2 Therapy Median (Range)		2 (1-9)
	1	4 (44%)			
HER2 Expression (%)	3+	8 (89%)	Previous HER2 Therapy Types (%)	Trastuzumab	14 (51.8%)
	2+/ISH+	1 (11%)		Trastuzumab Deruxtecan	3 (11.1%)
				Investigative	8 (29.6%)
				Pertuzumab	1 (3.7%)
				Tucatanib	1 (3.7%)

PHASE I SAFETY DATA

Summary of Adverse Events by Incidence

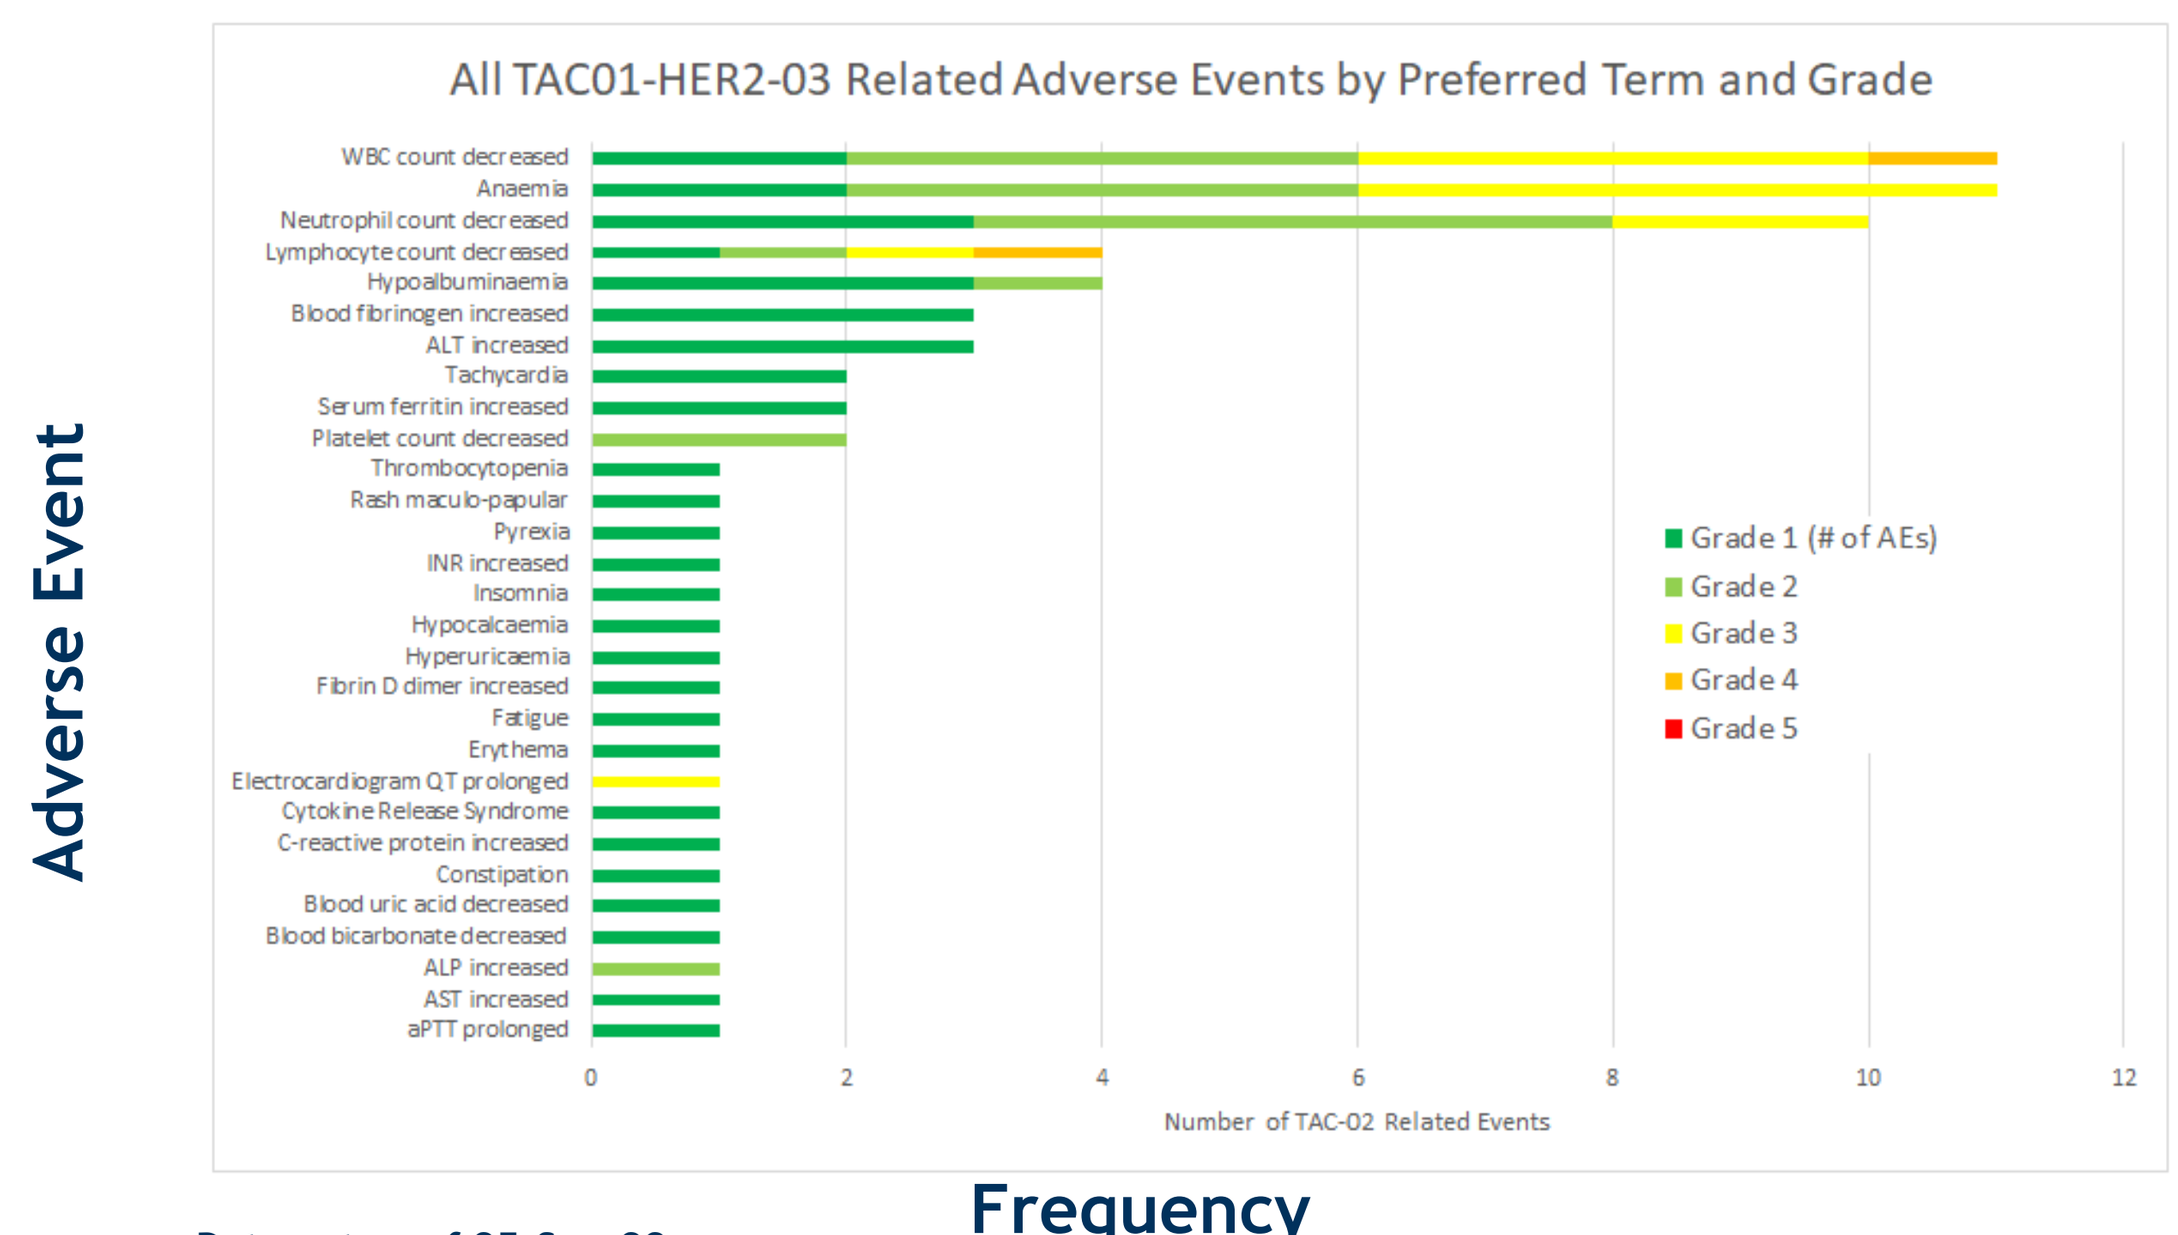
All TAC01-HER2-03 Related Adverse Events by Preferred Term and Grade
Adverse Event
WBC count decreased
Anaemia
Neutrophil count decreased
Lymphocyte count decreased
Hypoalbuminaemia
Blood fibrinogen increased
ALT increased
Tachycardia
Serum ferritin increased
Platelet count decreased
Thrombocytopenia
Rash maculo-papular
Pyrexia
INR increased
Insomnia
Hypocalcaemia
Hyperuricaemia
Fibrin D dimer increased
Fatigue
Erythema
Electrocardiogram QT prolonged
Cytokine Release Syndrome
C-reactive protein increased
Constipation
Blood uric acid decreased
Blood bicarbonate decreased
ALP increased
AST increased
aPTT prolonged
Grade 1 (# of AEs)
Grade 2
Grade 3
Grade 4
Grade 5
0
2
4
6
8
10
12
Number of TAC-02 Related Events
Frequency

Data cut as of 25-Sep-22

Safety Summary

One CRS grade 1 event (with pyrexia as the only symptom) at DL3 resolved within 3 days.

No Observed Immune Effector Cell-Associated Neurotoxicity (ICANS) through the first patient at DL 3

Most grade 2, 3 events were expected and related to LD chemotherapy.

CHANGES IN TUMOR MEASUREMENTS ACROSS ALL DOSE LEVELS

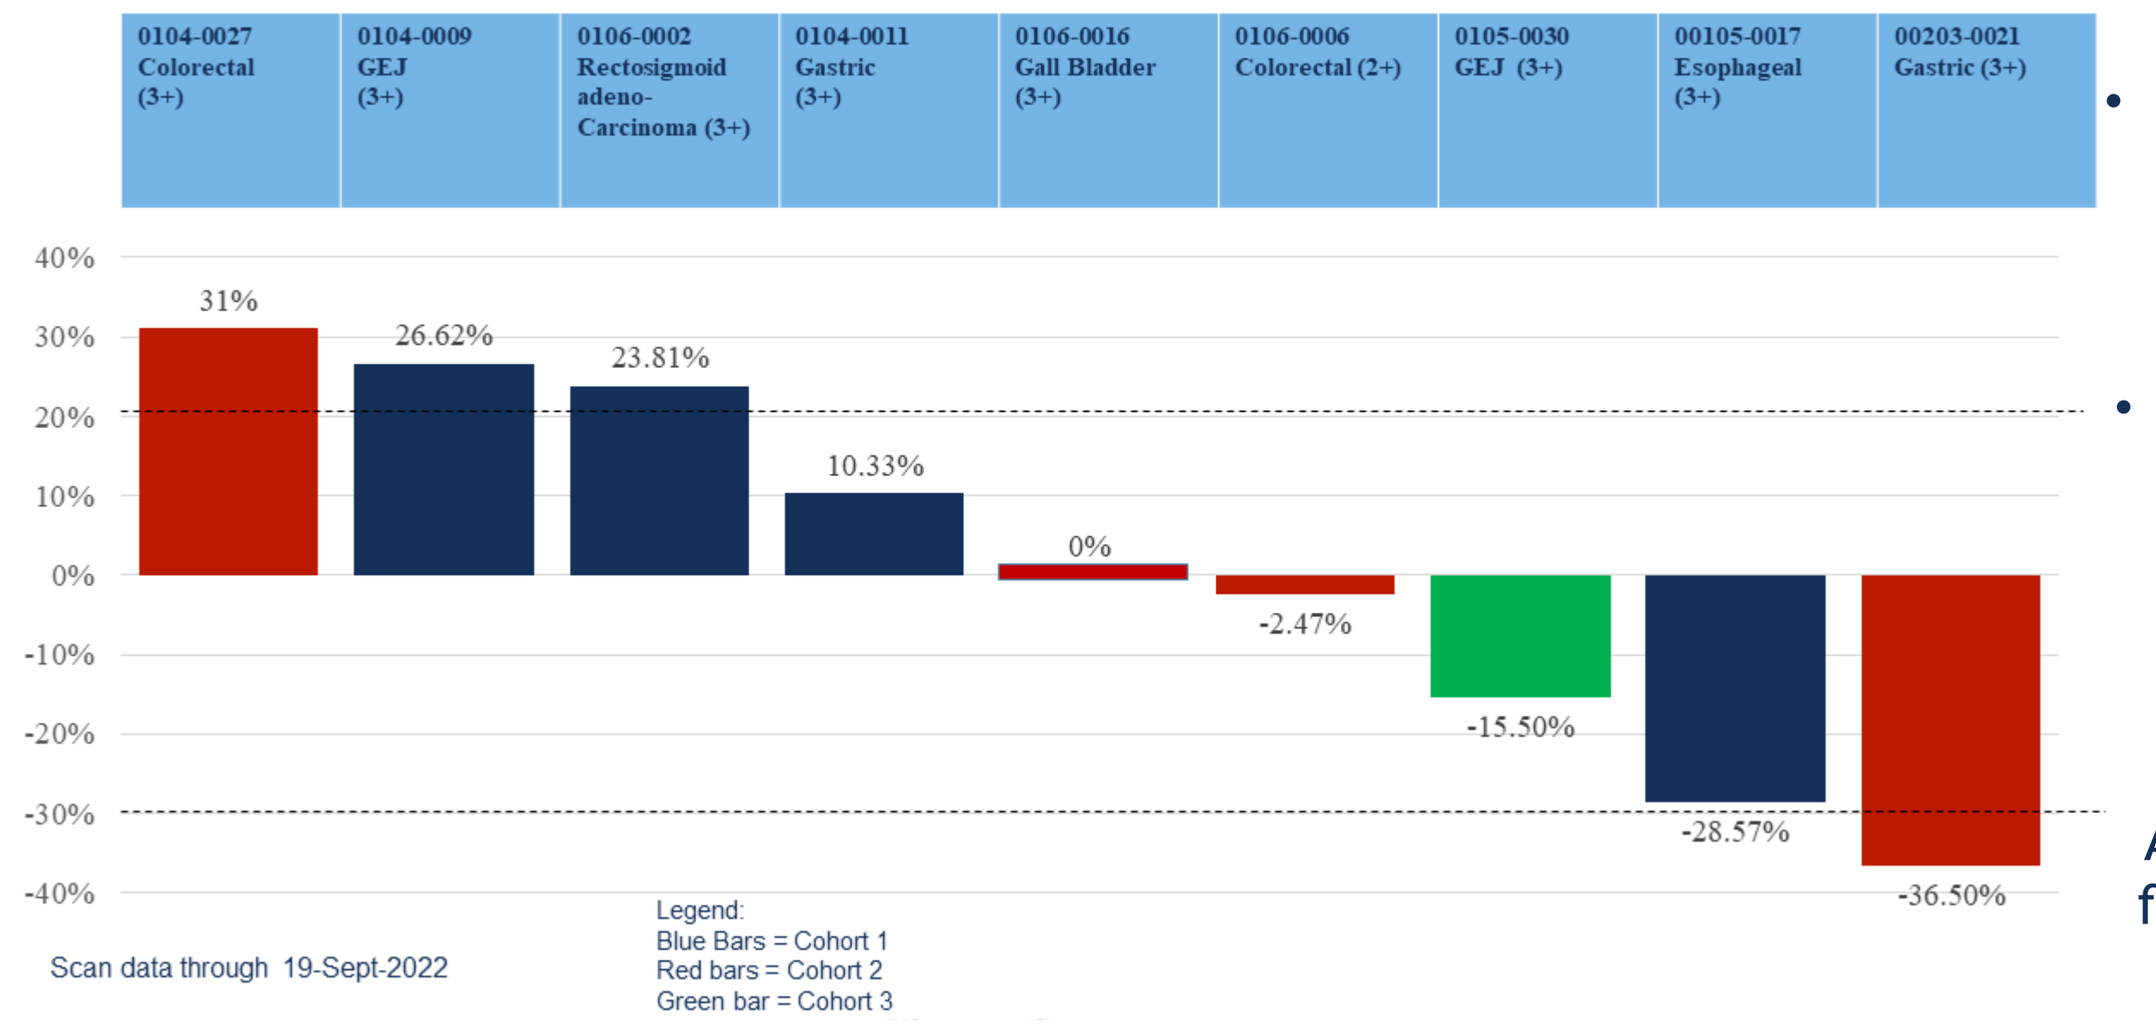
0104-0027 Colorectal (3+)
0104-0009 GEJ (3+)
0106-0002 Rectosigmoid adeno-Carcinoma (3+)
0104-0011 Gastric (3+)
0106-0016 Gall Bladder (3+)
0106-0006 Colorectal (2+)
0105-0030 GEJ (3+)
00105-0017 Esophageal (3+)
00203-0021 Gastric (3+)
40%
30%
20%
10%
0%
-10%
-20%
-30%
-40%
31%
26.62%
23.81%
10.33%
0%
-2.47%
-15.50%
-28.57%
-36.50%
Scan data through 19-Sept-2022
Legend:
Blue Bars = Cohort 1
Red bars = Cohort 2
Green bar = Cohort 3

Cohort 1
- **Suboptimal entry DL,** however **1st signal of clinical activity,** showing 1 pt. with mixed response

Cohort 2
- **Original DL1**
- 1 pt. with PR and 2 pts. with SD (zero change from baseline)

Cohort 3
- **2 pts. with stable disease**
- 1 pt. with unconfirmed progressive disease

Follow-Up
All patients are still in 2-year follow-up per protocol, except for two patients who died due to PD in cohort 1, and another who withdrew consent in Cohort 2.

Disease control rate of 75% at DL2, a very low dose of TAC T cell count in heavily pretreated pts. with aggressive malignancies

TUMOR ASSESSMENT: FIRST PATIENT RESPONSE

Partial Response Observed in Patient 0203-0021
42 year old male with IHC 3+, HER2+ stage IVb metastatic gastric adenocarcinoma. Previously treated with 2 lines of HER2-directed targeted therapy + chemotherapy & palliative radiation. The patient also received bridging therapy which consisted of 3 cycles of chemotherapy.

Stable Disease noted in both patients 0106-0016 (3+ HER2) and 0106-0006 (2+ HER2). Remarkably, no significant change in tumor size from baseline was observed in both patients.

RECIST 1.1 Tumor Response Assessments (Measurable Disease)

Baseline	Day 29	% change
20 mm	12.7mm	-36.5%

Gastrohepatic Lymph Node in Patient 0203-0021*

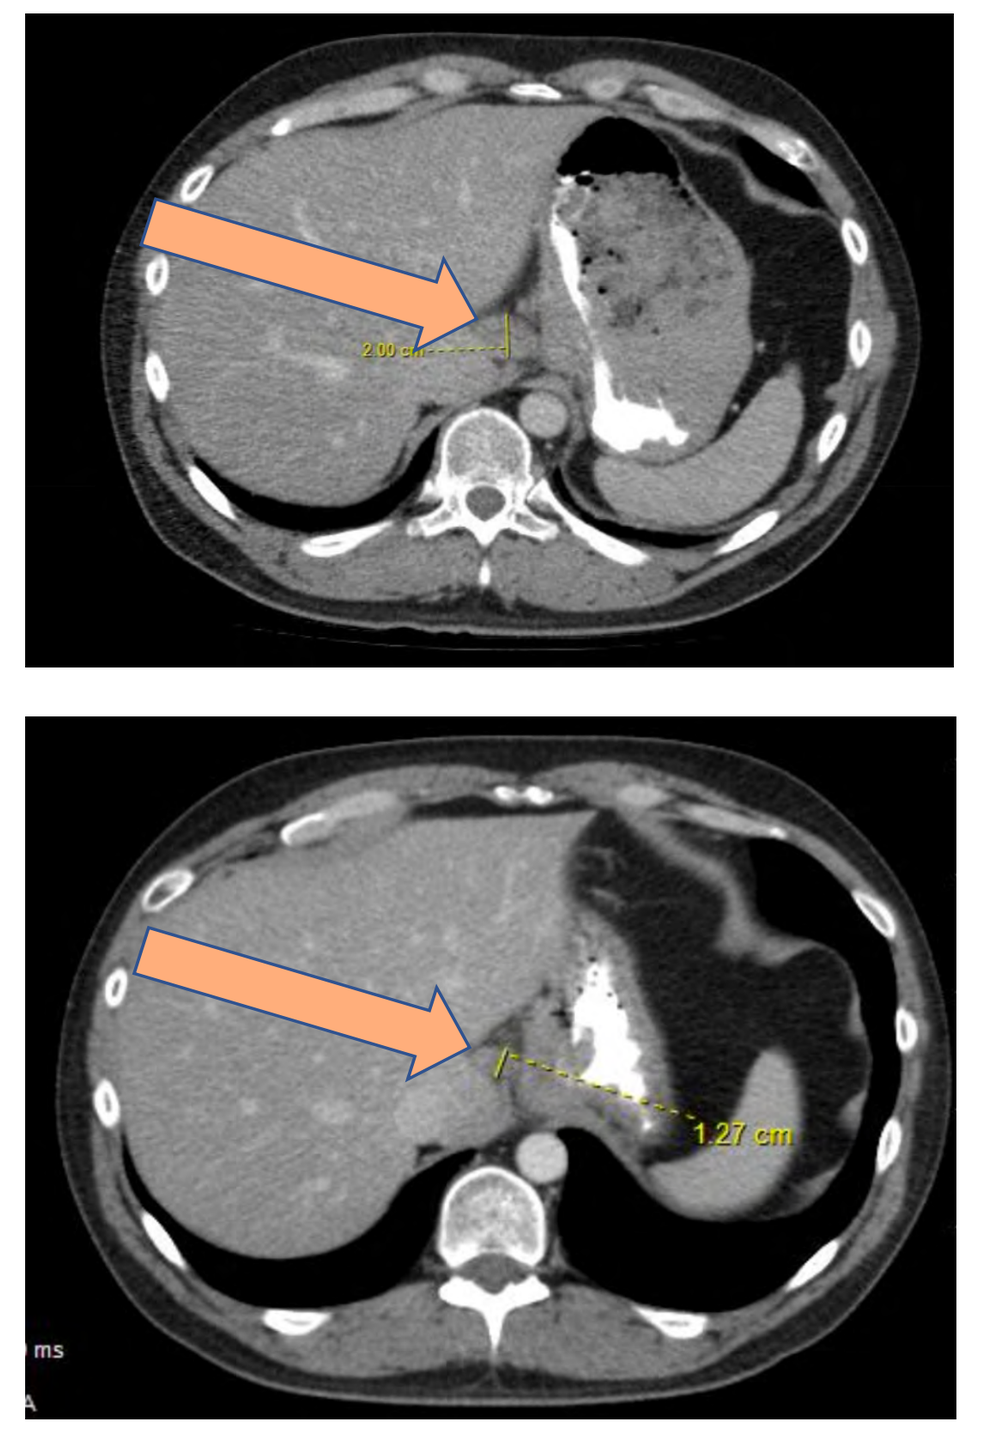
Baseline
Day 0
(2.00 cm)
Day 28
(1.27 cm)
1.27 cm

Gastrohepatic Node Reduction / **Perioportal Mass Reduction**

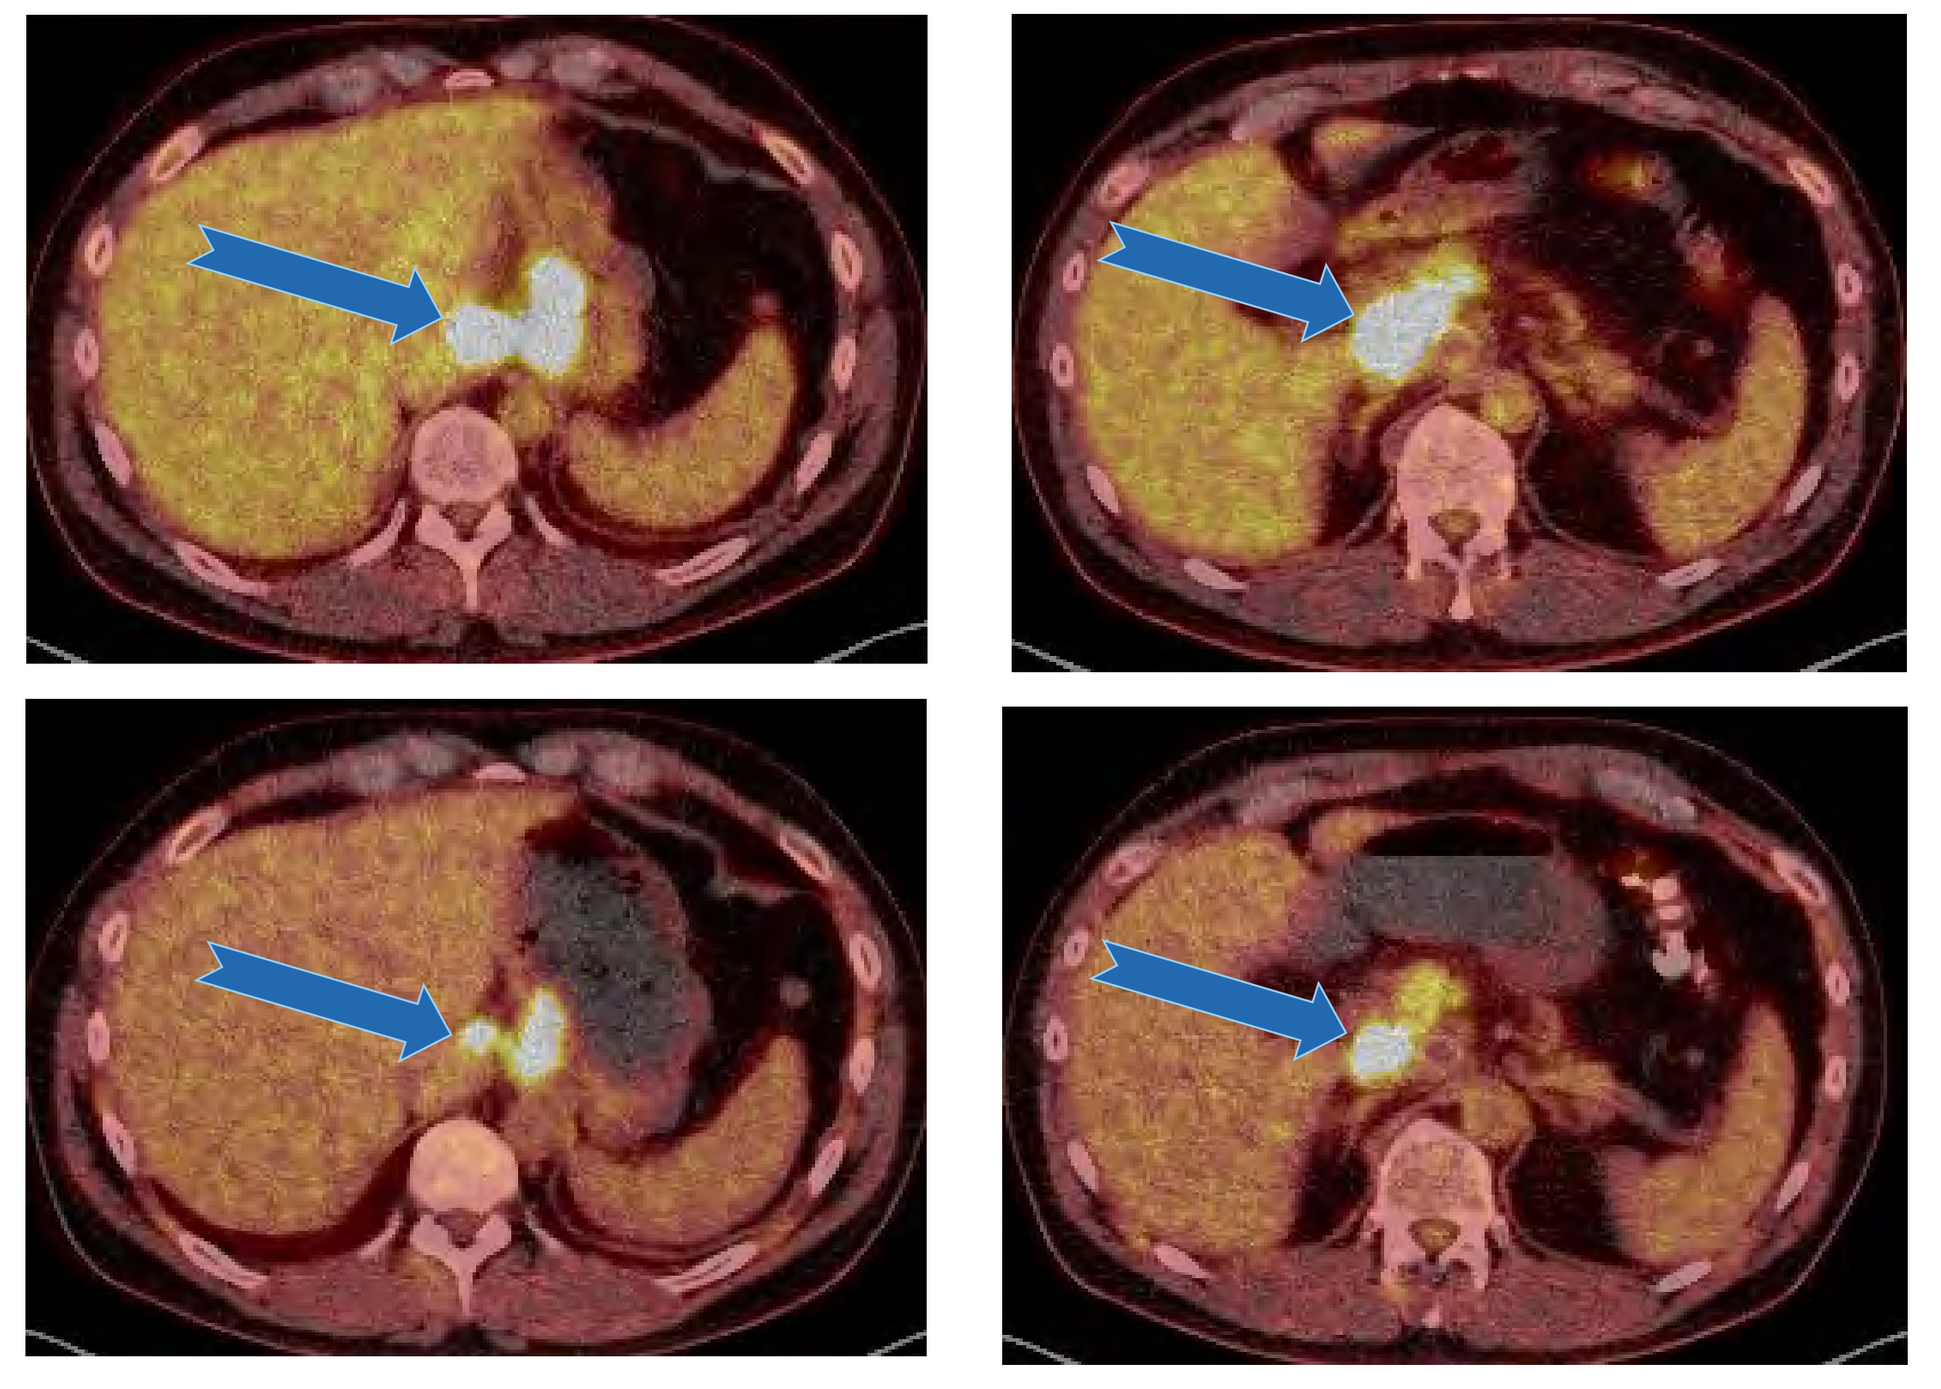
Baseline
Day 0
Day 29

PET Scan Results (Evaluable disease)

Tumor	Size Pre	Size Post	SUVmax pre TAC-T	SUVmax post TAC-T
Right lower paraoesophageal node at the level of T10	-	-	6.5	3.2
Gastrohepatic ligament lymph nodes	2.4 x 2.3 cm	1.9 x 1.2 cm	20.5	15
Portacaval nodal mass*	5.1 x 4.0 cm	4.8 x 3.4 cm	22.1	29
Left common iliac lymph node	No gross interval change per radiologist		19.3	23.1

Lymph Node Reduction
There has been overall interval decreased size of previously noted metabolically active lymph nodes associated with the mass, however with persistent intense metabolic activity in most of them.

*Despite SUVmax increase the lesions shows more extensive photopenic areas, representing cystic/necrotic change->cancer cell death.
** There was also a stable cystic/necrotic node->cancer cell death.

SUMMARY & CONCLUSIONS

SAFETY

Interim results from the Phase I TACTIC-02 study suggest that TAC-01 HER2 is safe and well tolerated, supported by the absence of DLTs and events of special interest except for one grade 1 CRS at Dl3, which quickly resolved with minor intervention.

EFFICACY

Demonstrated early signals of clinical activity, highlighting a partial response in a stage IVb gastric cancer patient and a disease control rate of 75% at DL2.

TRIAL PROGRESS: Phase I trial is ongoing and on track to be completed in Jan 2023. Enrollment completed at DL3 & patients consented to secure timely enrollment on DL4.
Phase II enrollment begins in Q1 2023.

Disclosures: Dr. Benjamin L. Schlechter has no conflicts of interest to report.
Sponsorship: This Phase I/II Clinical Trial has been fully funded by Triumvira Immunologics Inc.
Contact: dadib@triumvira.com, benjamin_schlechter@dfci.harvard.edu

Sources: 1. Siegel R, et. al. Cancer Statistics 2022. A Cancer Journal for Clinicians. 2022;72(1):7-33 & Triumvira Internal Analysis and Assumptions
Acknowledgments: Clinical Trial Sites and Apheresis Unit staff: Princess Margaret Cancer Centre, The University of Chicago Medical Center, Dana Farber Cancer Institute, MD Anderson Cancer Center as well as the patients and their families.
Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.