Development of Claudin 18.2-TAC T Cells for the Treatment of Gastric Cancer

Christopher W. Helsen, Tania Benatar, Thanyashanthi Nitya-Nootan, Heather McGregor, Philbert Ip, Prabha Lal, Stacey Xu, Laura Shaver, Suzy Prosser, Sadhak Sengupta, Andreas G. Bader
Triumvira Immunologics, 9433 Bee Caves Rd Building 1, Suite 240, Austin, TX 78733, USA (Headquarters), 270 Longwood Road South, Hamilton, Ontario L8P 0A6, Canada (Research Division)

ABSTRACT

Background

The T cell antigen coupler (TAC) is a novel, proprietary chimeric receptor that facilitates the re-direction of T cells to tumor cells and activates T cells by co-opting the endogenous T cell receptor complex with the goal to elicit a safe and durable anti-tumor response. In preclinical models of cancer, TAC-engineered T cells effectively eradicate tumor cells in vitro and in vivo without TAC-related toxicities. TAC01-HER2, a first-in-class TAC T product targeting HER2 (ERBB2), has recently entered a phase I/II clinical trial in patients with HER2-positive solid tumors. Here, we present the development of a new TAC T product targeting Claudin 18.2 (CLDN18.2) to treat gastric cancer. CLDN18.2 belongs to a family of Claudin tight junction proteins whose expression is naturally exclusive to normal stomach. In gastric cancer cells, however, protein expression patterns are perturbed, leading to tumor-selective surface expression of CLDN18.2. Thus, CLDN18.2 is a preferred antigen for the specific targeting of tumor cells via TAC T cells.

Materials and Methods

The functionality of the CLDN18.2-TAC receptor was characterized using a variety of in vitro and in vivo assays. In vitro assays were based on flow cytometric analysis of TAC surface staining and cytokine release. Cytotoxicity was assessed via luminescence-based co-culture assays and real-time microscopy. In vivo studies examined the anti-tumor effect of TAC-engineered T-cells against established CLDN18.2-expressing tumor xenografts.

Results

T cells virally transduced with the CLDN18.2-TAC transgene demonstrated satisfactory surface expression of the TAC receptor and showed increased cytokine production when activated by CLDN18.2 expressing target cells in vitro. Secretion of IL2, IFNγ and TNFα were comparable with cytokine levels produced by activated HER2-TAC T cells used as a positive control. In vitro cytotoxicity assays demonstrated a strong anti-CLDN18.2 response and killing of CLDN18.2 expressing target cell lines. No increases in cytokine levels and no cytotoxicity were observed in non-transduced T cells (NTD) and CLDN18.2-TAC T cells co-cultured with CLDN18.2-negative target cells, indicating that the T cell response is specific to the CLDN18.2 antigen. Intravenous administration of CLDN18.2-TAC T cells in mice carrying CLDN18.2-positive tumor xenografts led to a sustained anti-tumor response.

Conclusion

The in vitro and in vivo data confirm strong and specific activity of CLDN18.2-targeted TAC T cells against CLDN18.2-expressing cancer cells and highlight the versatility of the TAC platform for therapeutic applications in solid tumors.

TAC SCIENCE

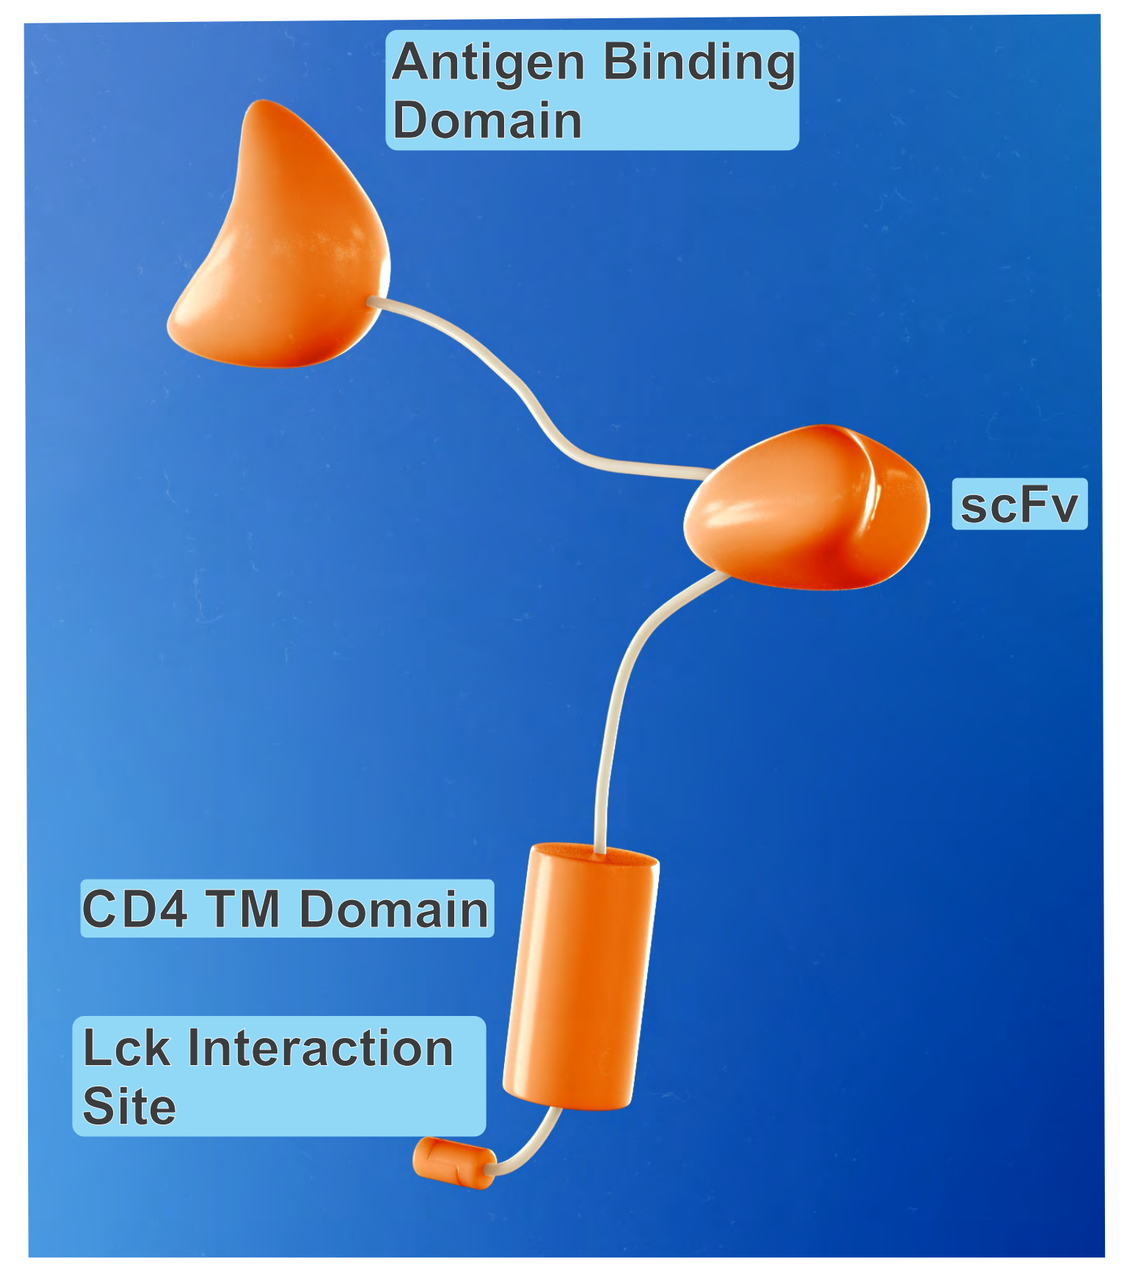
Antigen Binding Domain
scFv
CD4 TM Domain
Lck Interaction Site

Key features of TAC technology:

- TAC functions independently of MHC
- TAC requires endogenous TCR for T cell activation
- TAC incorporates the co-receptor and recruits the TCR complex, mimicking natural TCR activation

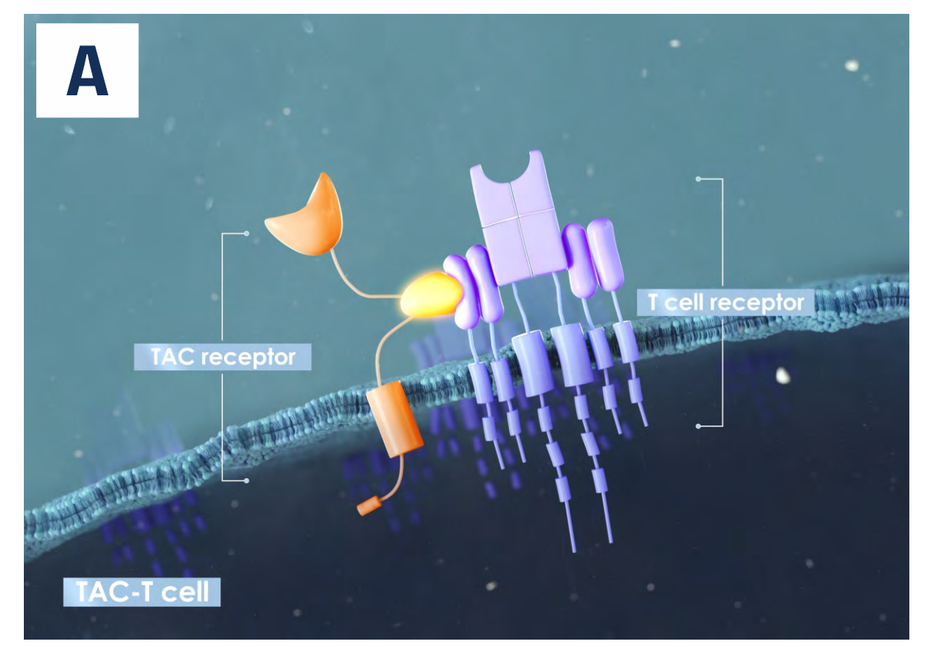
A
TAC receptor
T cell receptor
TAC-T cell

The TAC receptor interacts directly with the TCR-CD3 epsilon domain (A).

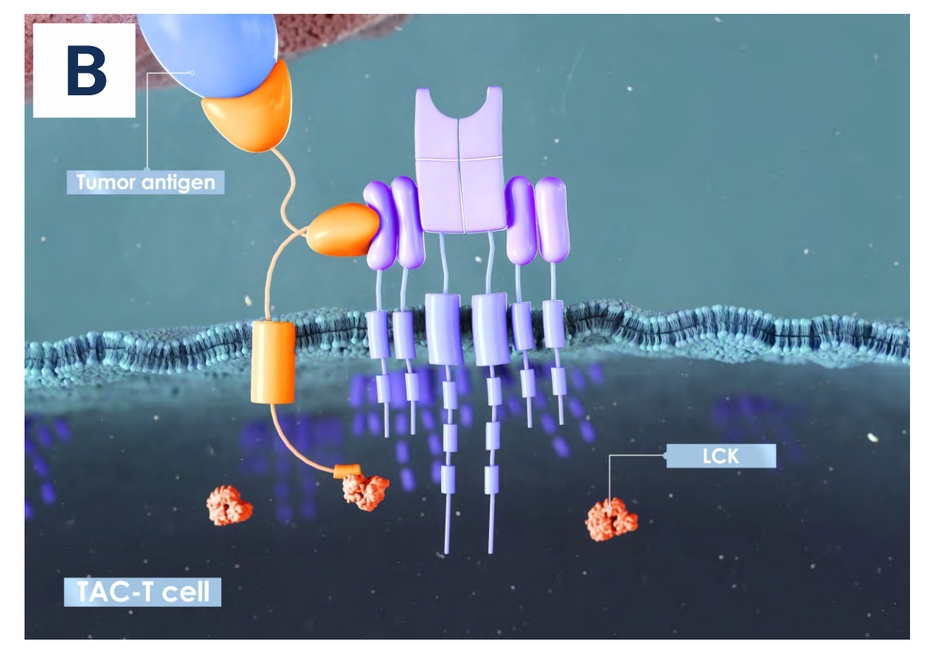
B
Tumor antigen
LCK
TAC-T cell

The TAC receptor also binds directly the tumor antigen. Initiating the first step in T cell activation (B) which then leads to the clustering of TAC-TCR complexes required for full T cell activation.

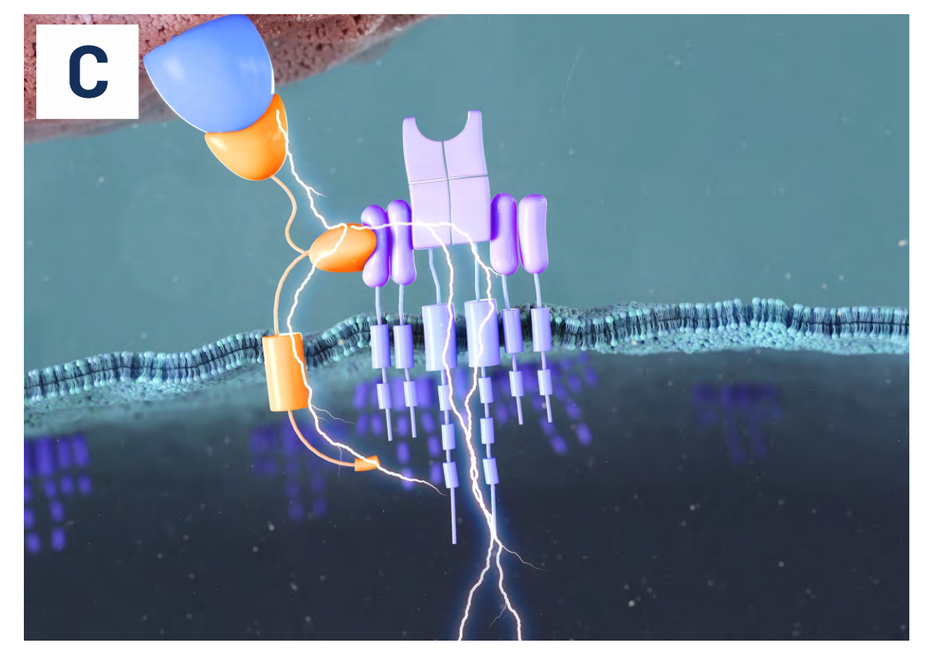
C

The TAC receptor then signals through the CD3-TCR complex (C).

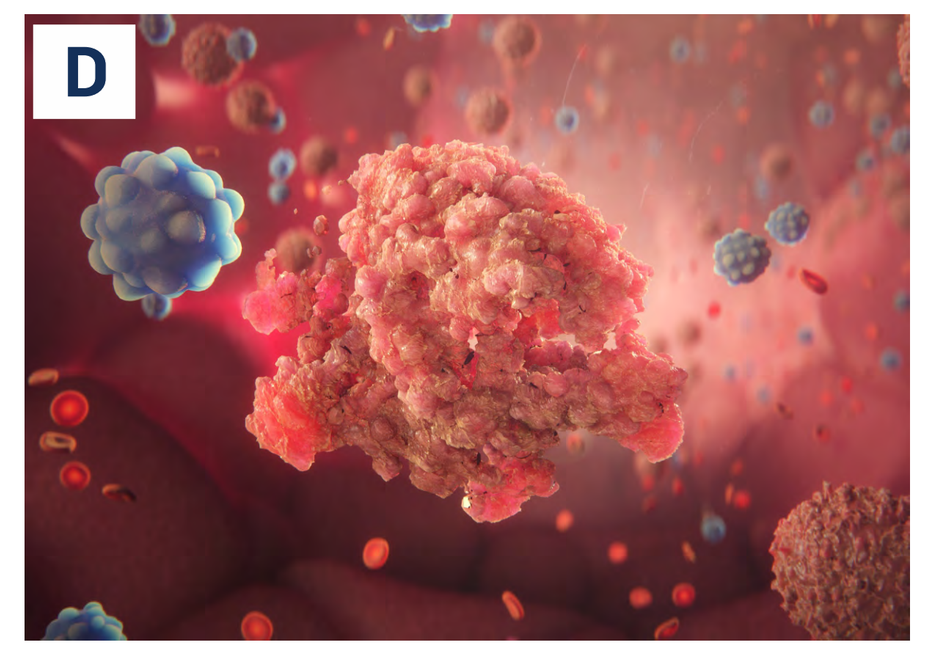
D

This ultimately results in tumor cell lysis (D).

triumvira

Watch a short animation to understand the TAC mechanism

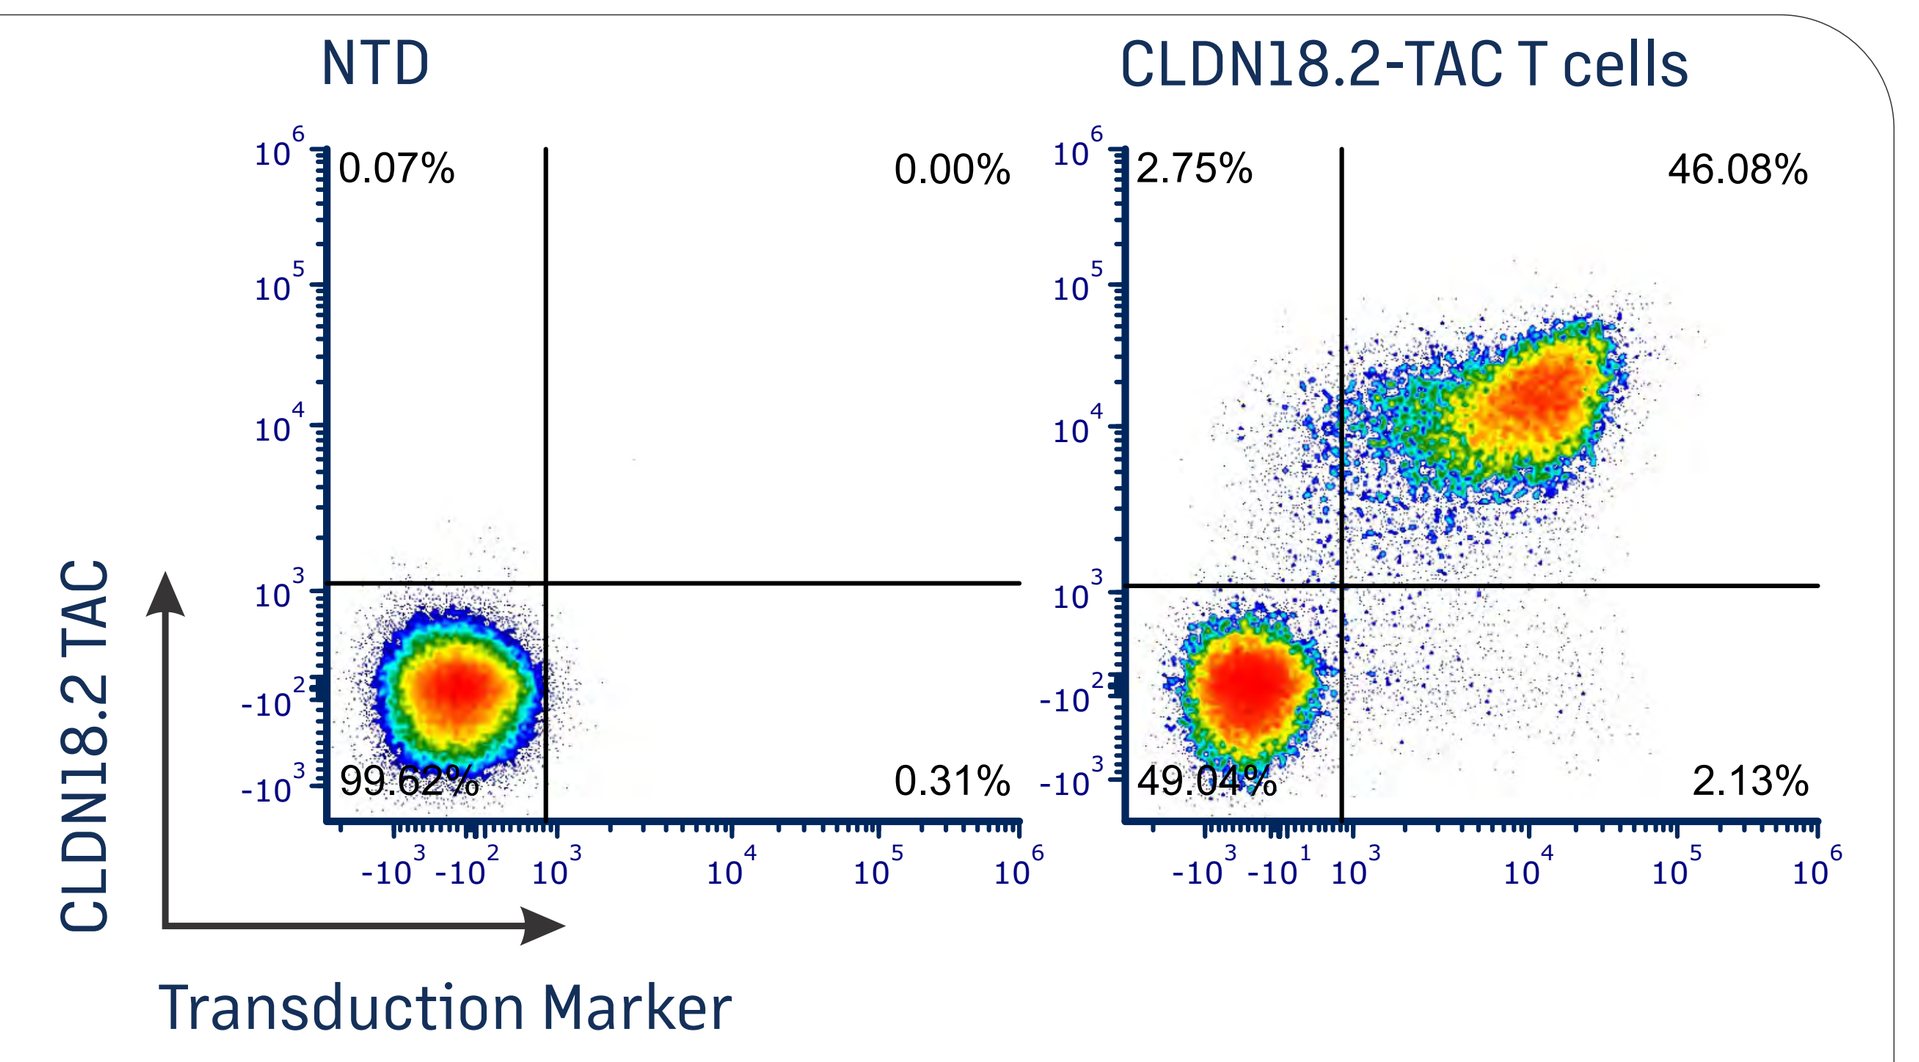
NTD
CLDN18.2-TAC T cells
0.07%
0.00%
99.62%
0.31%
2.75%
46.08%
49.04%
2.13%
CLDN18.2 TAC
Transduction Marker

CLDN18.2-TAC Expression on Surface of T Cells

T cells were engineered using a lentivirus co-expressing the mStrawberry red fluorescence protein as transduction marker (see above). CLDN18.2-TAC showed good surface expression and T cells were easily engineered.

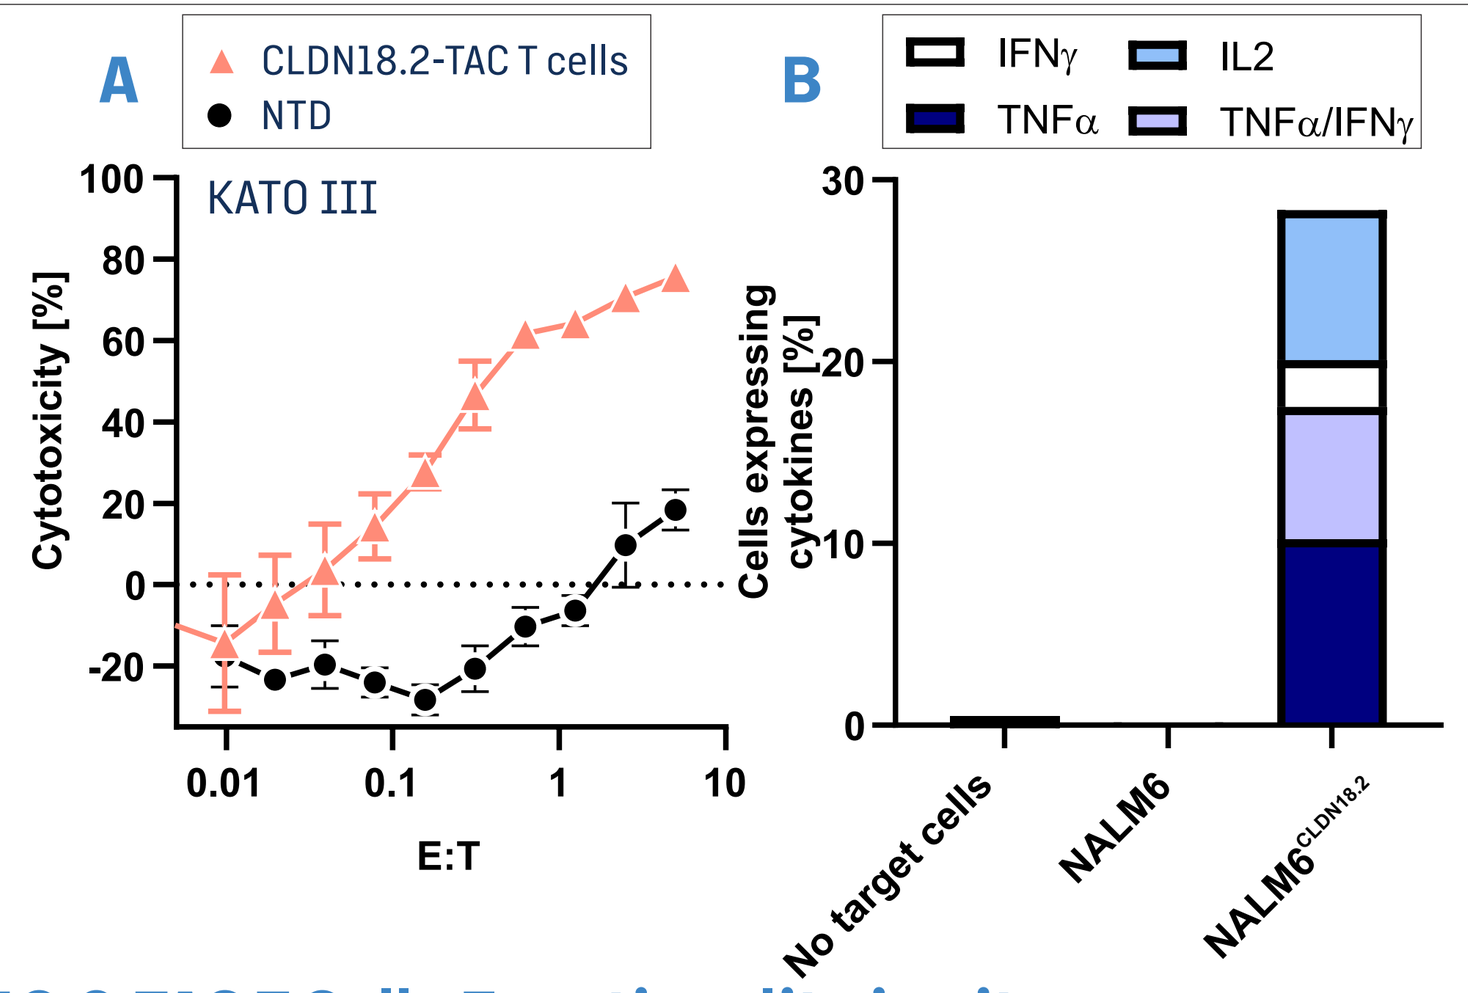
A
CLDN18.2-TAC T cells
NTD
KATO III
Cytotoxicity [%]
E:T
B
IFNγ
IL2
TNFα
TNFα/IFNγ
Cells expressing cytokines [%]
No target cells
NALM6
NALM6CLDN18.2

CLDN18.2-TAC T Cells Functionality in vitro

A. KATO III cells, naturally expressing Claudin 18.2, were engineered to express enhanced Luciferase (eLuc). Following a 30 h co-culture KATO III^{eLuc} viability was assessed. CLDN18.2-TAC T cells but not non-transduced control T cells (NTD) were able to efficiently engage and kill KATO III^{eLuc} cells.

B. CLDN18.2-TAC T cells were co-cultured for 4 h with NALM6$^{CLDN18.2}$, NALM6 or in absence of target cells, stained for IFNγ, TNFα, and IL2, and analysed by flow cytometry. CLDN18.2-TAC T cells showed strong cytokine production in the presence but not in the absence of CLDN18.2-expressing target cells. There was no indication of auto-activation.

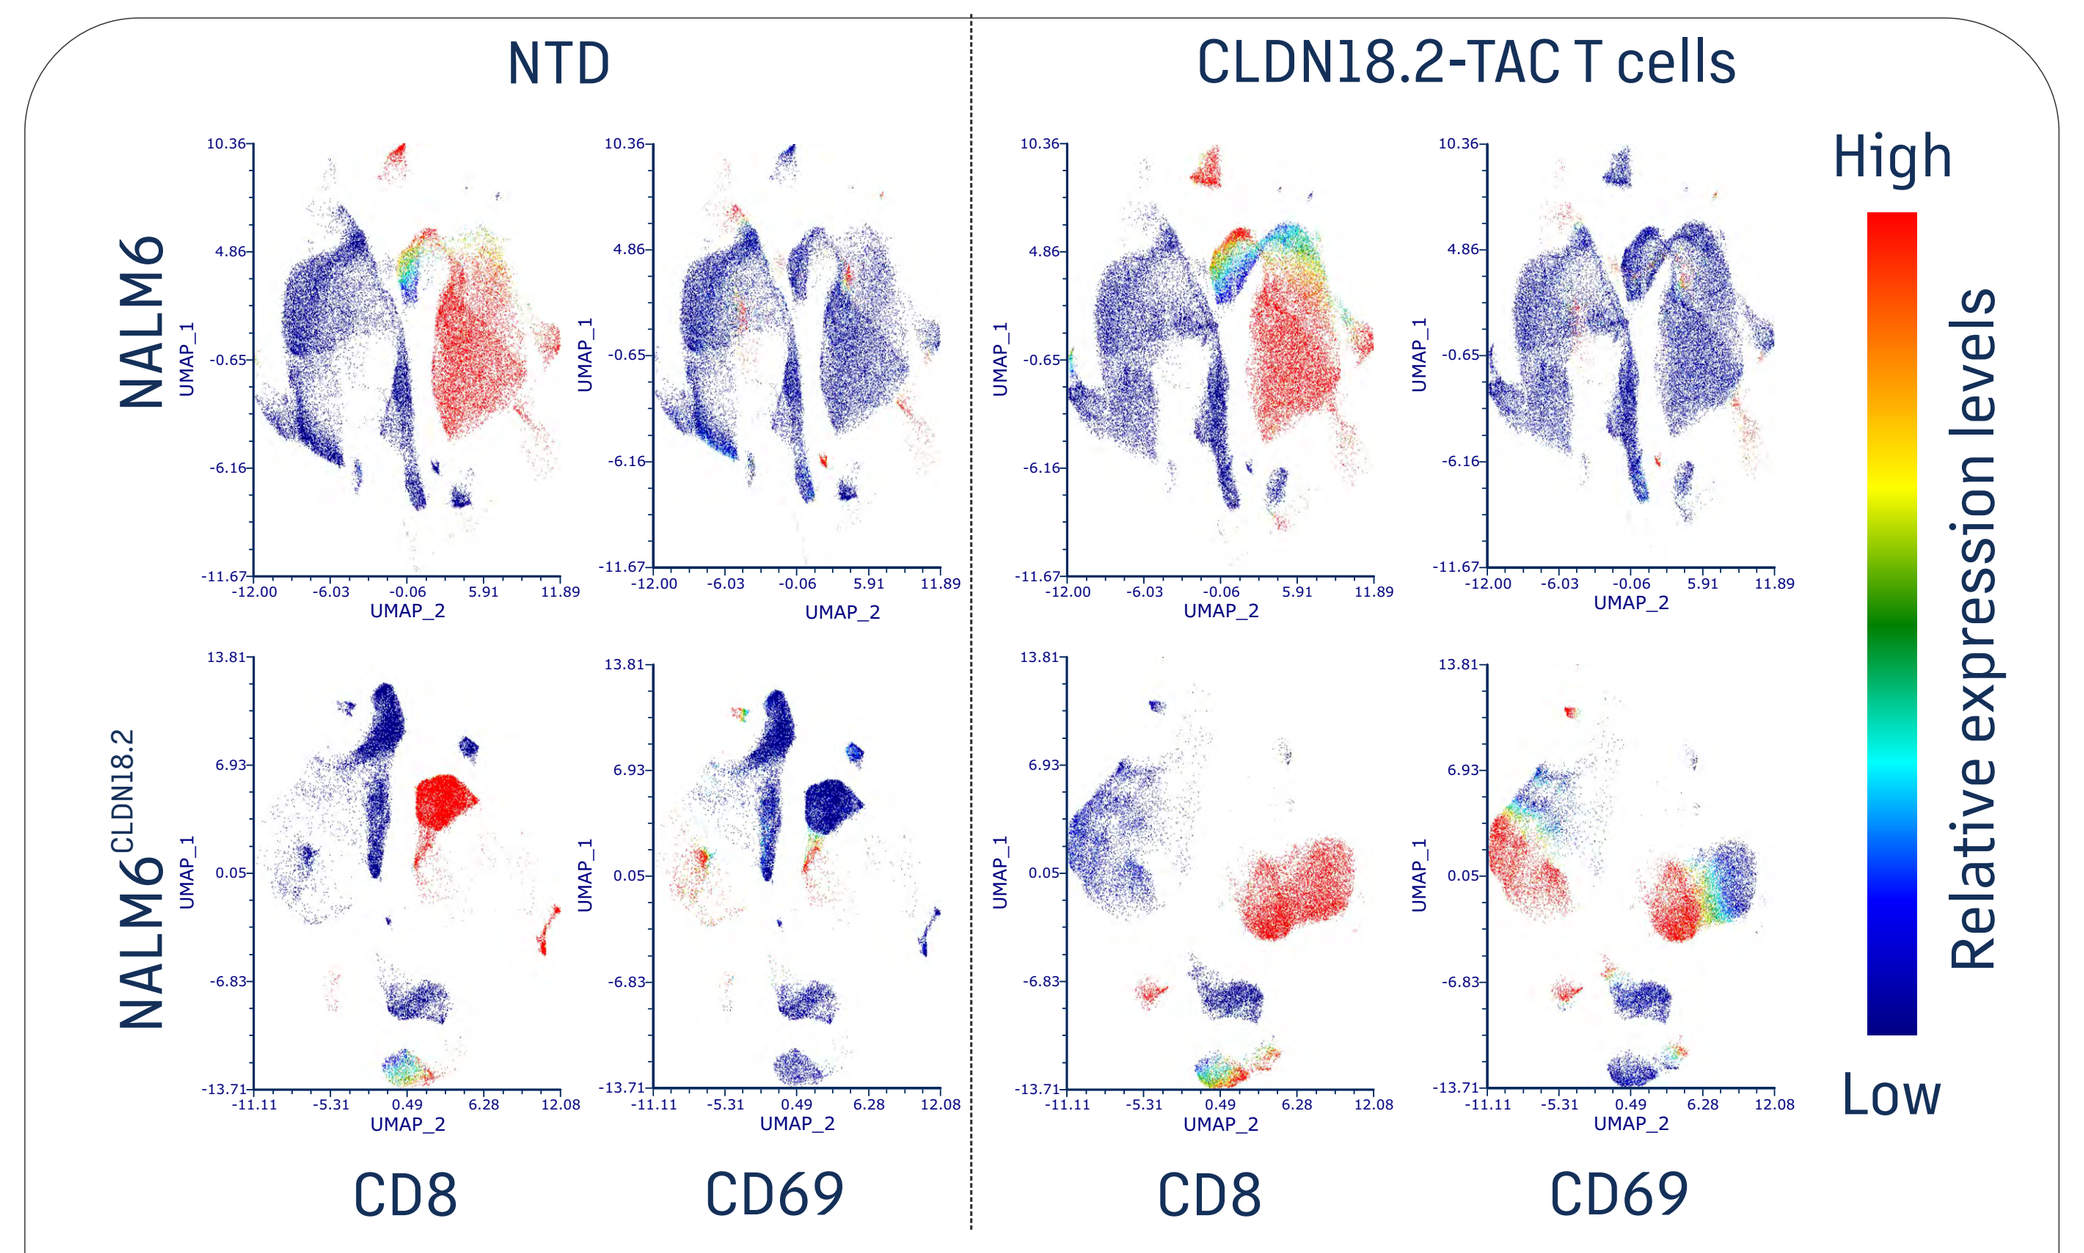
NTD
CLDN18.2-TAC T cells
NALM6
NALM6CLDN18.2
CD8
CD69
CD8
CD69
High
Low
Relative expression levels

CLDN18.2-TAC T Cells Phenotype

CLDN18.2-TAC T cells or NTD control cells were analysed with 16 surface markers. Cells were analysed and clustered using the UMAP algorithm. Expression of CD8 and CD69 is shown, with colors indicating the relative abundance of the illustrated markers.

This preliminary data set shows similar patterns of expression of all surface markers analysed on CLDN18.2-TAC T cells and NTD cells cultured with NALM6 cells. However, when cultured with CLDN18.2-expressing NALM6 cells, CLDN18.2-TAC T cells (but not NTD cells) become activated as indicated by upregulation of CD69. CLDN18.2-TAC T cells retain similar memory phenotypes to the NTD controls even post activation (data not shown). Together, this data demonstrates that CLDN18.2-TAC T cells do not exhibit tonic signalling and are activated only in the presence of antigen.

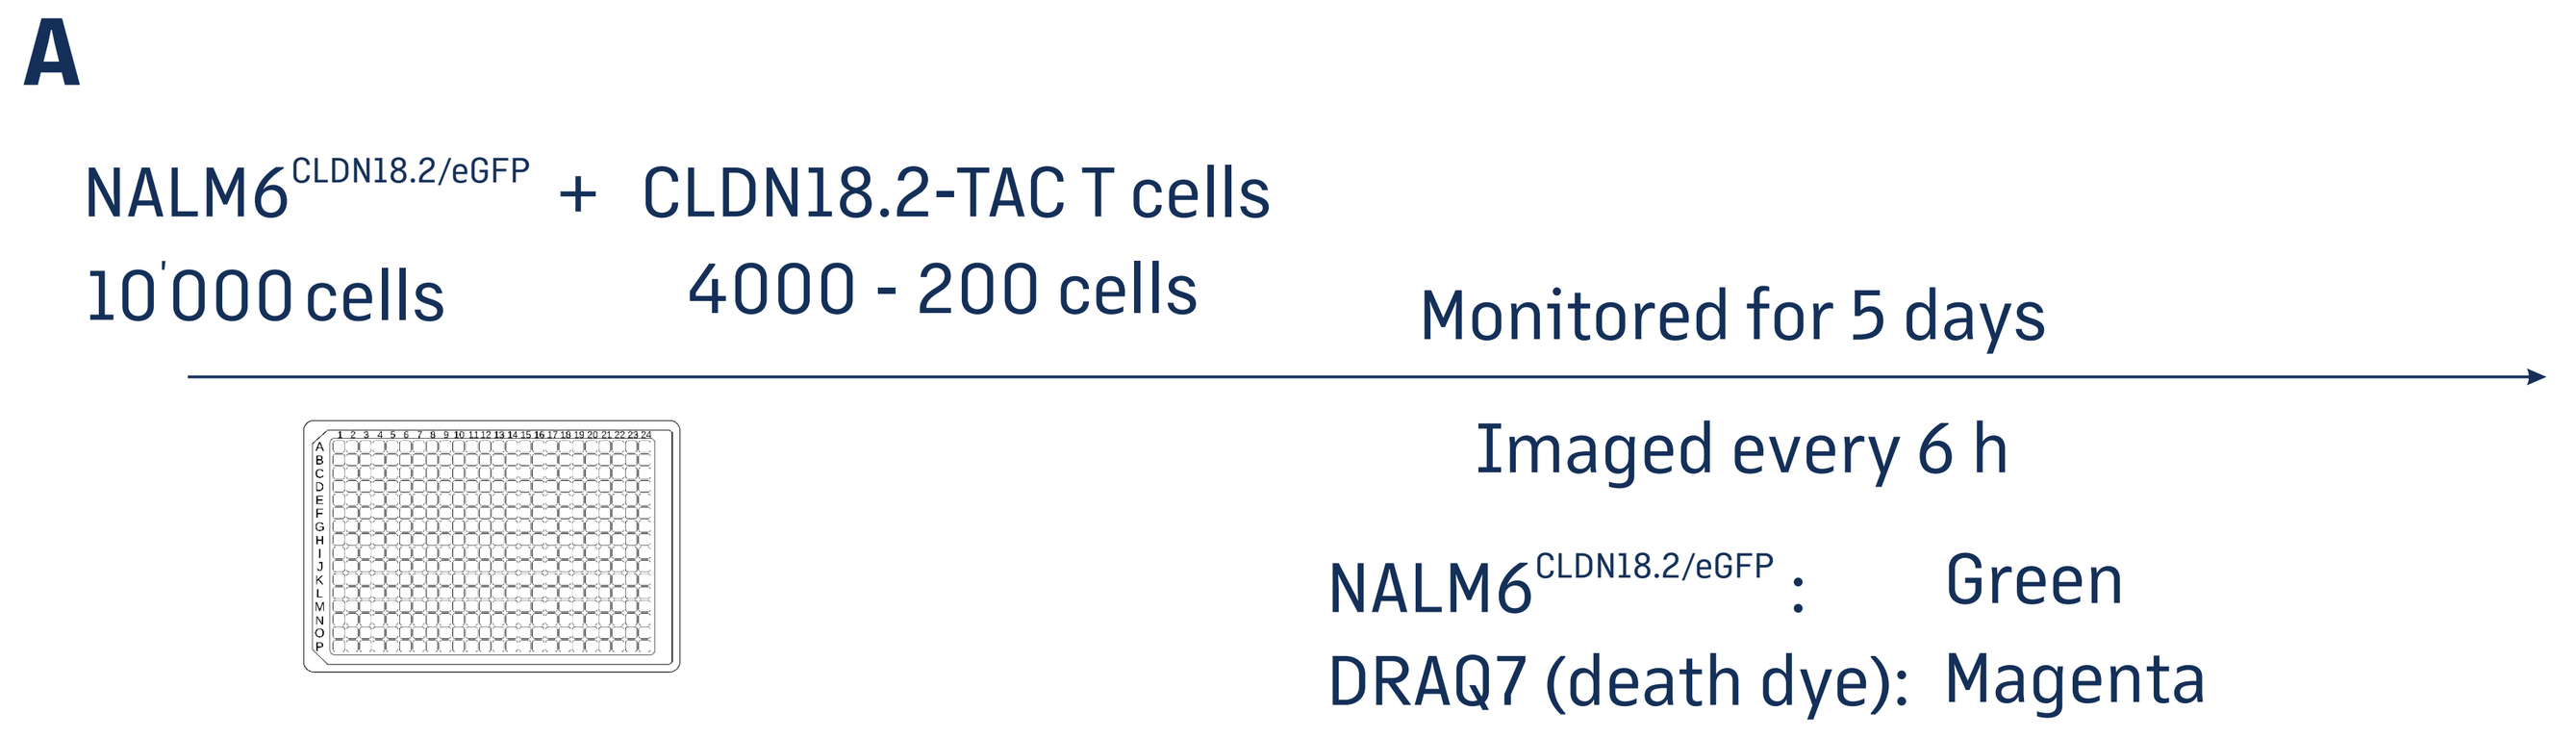
A
NALM6CLDN18.2/eGFP + CLDN18.2-TAC T cells
10'000 cells
4000 - 200 cells
Monitored for 5 days
Imaged every 6 h
NALM6CLDN18.2/eGFP: Green
DRAQ7 (death dye): Magenta

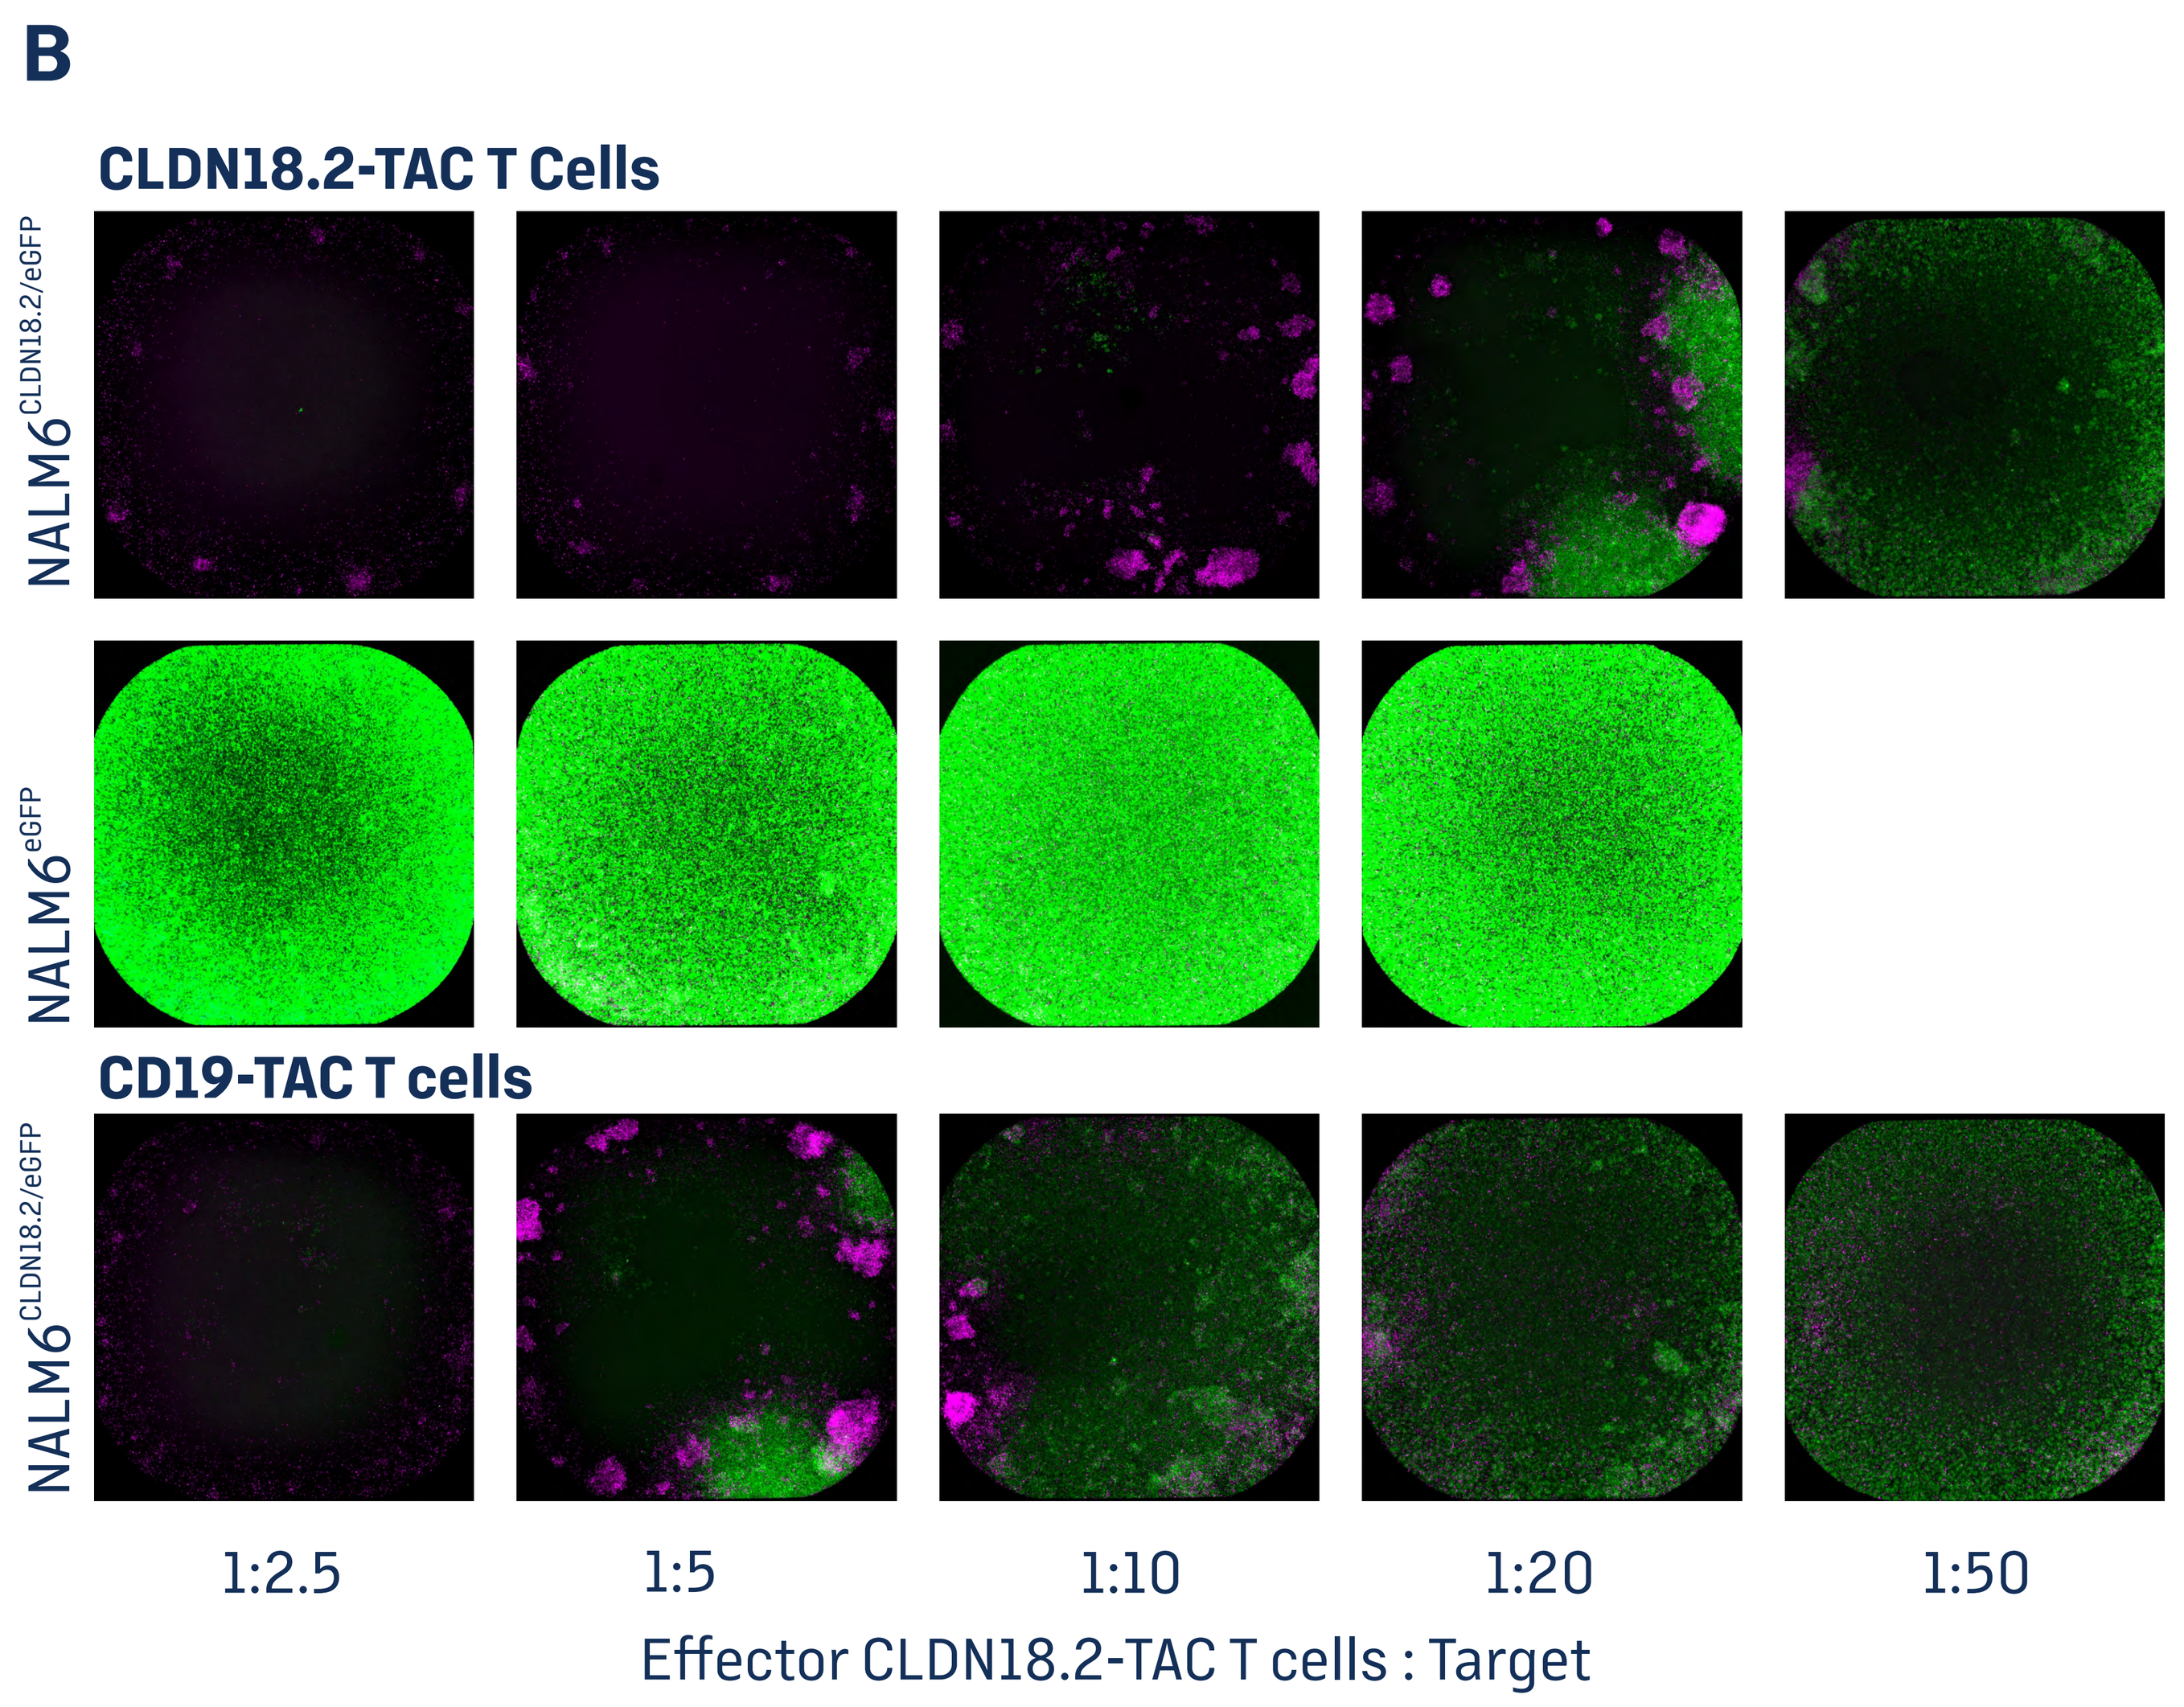
B
CLDN18.2-TAC T Cells
NALM6CLDN18.2/eGFP
NALM6eGFP
CD19-TAC T cells
NALM6CLDN18.2/eGFP
1:2.5
1:5
1:10
1:20
1:50
Effector CLDN18.2-TAC T cells : Target

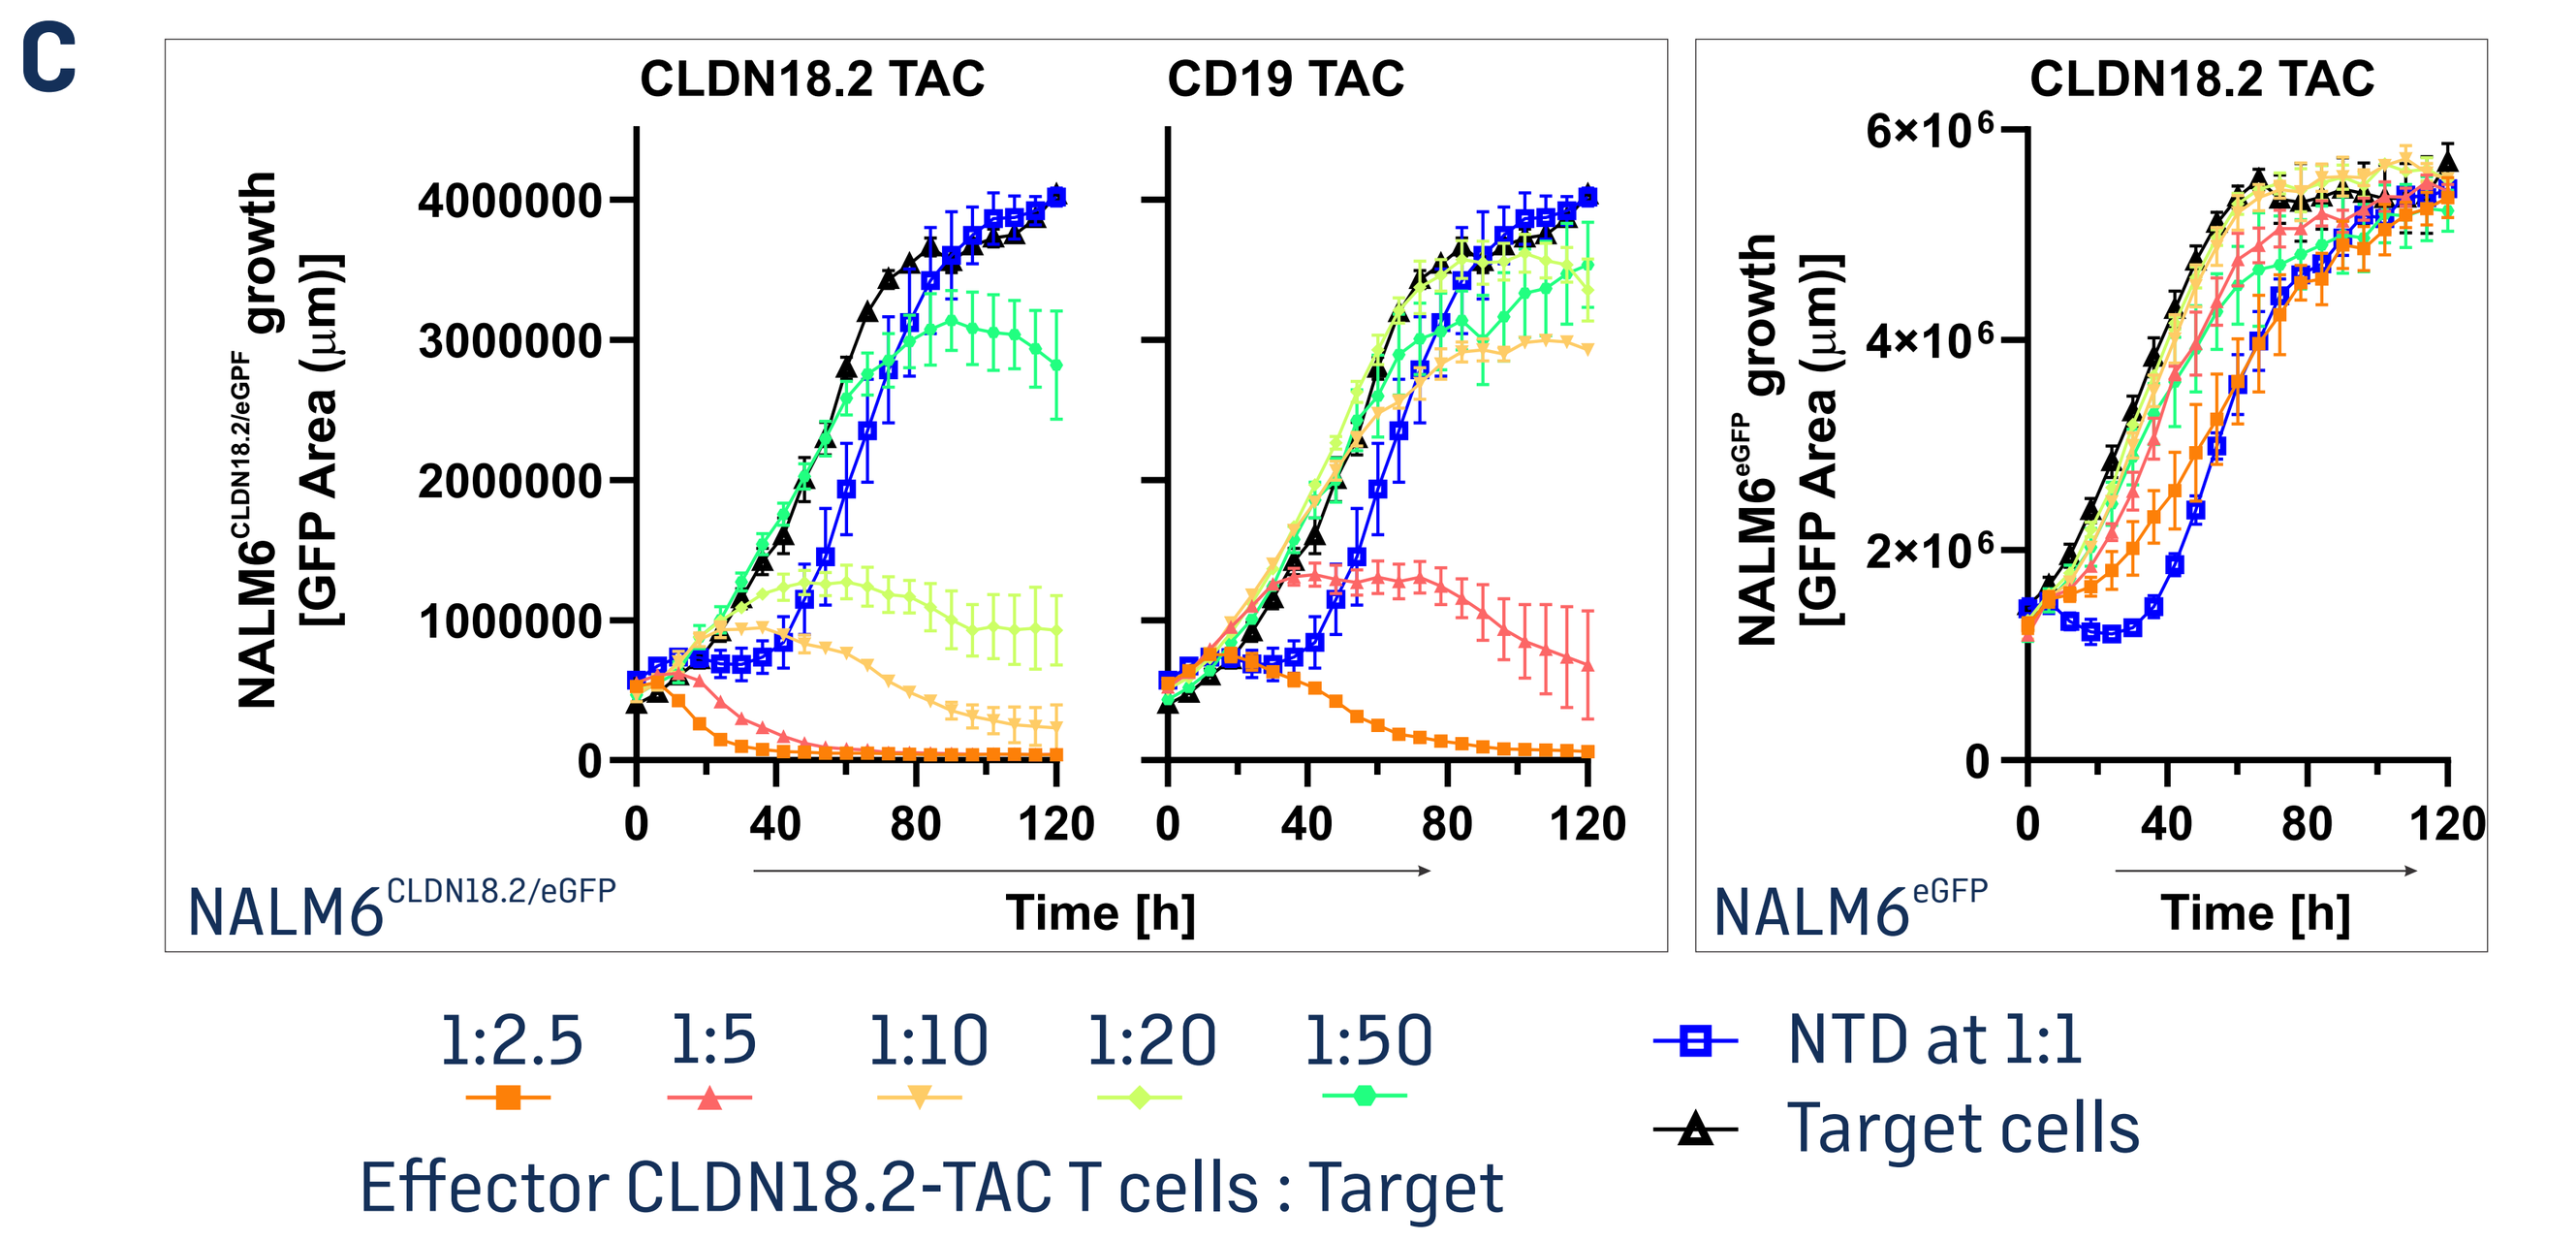
C
CLDN18.2 TAC
CD19 TAC
NALM6CLDN18.2/eGFP growth [GFP Area (μm)]
Time [h]
NALM6CLDN18.2/eGFP
CLDN18.2 TAC
NALM6eGFP growth [GFP Area (μm)]
NALM6eGFP
1:2.5
1:5
1:10
1:20
1:50
Effector CLDN18.2-TAC T cells : Target
NTD at 1:1
Target cells

Real-time in vitro Cytotoxicity

A. CLDN18.2-TAC T cells or non-transduced control T cells (NTD) were co-cultured with NALM6$^{CLDN18.2/eGFP}$, NALM6^{eGFP} or target cells without T cells for 5 days. Each 384 well was imaged every 6 h. Tumor cells are shown in green, and cell death is shown in magenta (DRAQ7 dye). All reactions were set up as triplicates.

B. Final images of different E:T ratios after 5 days of co-culture. CLND18.2-TAC T cells when co-cultured with NALM6^{eGFP} did not control tumor cell growth. However, when co-cultured with NALM6$^{CLDN18.2/eGFP}$ growth of cancer cells was controlled at a E:T of 1:20 (500 TAC T cells : 10'000 target cells). By comparison, the CD19- TAC construct, known to be highly efficacious in vivo, controlled tumor cell growth at a ratio of 1:5 (2000 TAC T cells : 10'000 target cells).

C. Quantitative analysis of tumor cell growth as a function of eGFP area. Tumor cells alone (black) grew exponentially. NTD (blue) used at a high E:T of 1:1 showed a brief delay in NALM6^{eGFP}/NALM6$^{CLDN18.2/eGFP}$ but did not control tumor cell growth. CLDN18.2-TAC T cells did not control NALM6^{eGFP} proliferation beyond the NTD control. However, when co-cultured with NALM6$^{CLDN18.2/eGFP}$, significant tumor cell control was achieved superior to the CD19-TAC T product.

triumvira

In Vivo Evaluation of CLDN18.2-TAC T cells

NSG mice were inoculated with 5 x 10^5 NALM6$^{CLDN18.2/eLuc}$ tumor cells. Four days after engraftment mice were treated with a single dose of CLDN18.2-TAC T cells. Non-treated (NT) mice and mice treated with non-transduced T cells (NTD) were used as negative controls. Mice were dosed with 4 x 10^6 TAC T cells or NTD to match the total T cell dose used for TAC T cells. Total luminescence was measured weekly, and the resulting total flux (photons/second) as the sum of the dorsal and ventral reads is shown.

Complete tumor rejection was achieved in all animals treated with CLDN18.2-TAC T cells. Negative controls did not show any tumor control. No TAC associated toxicity was observed.

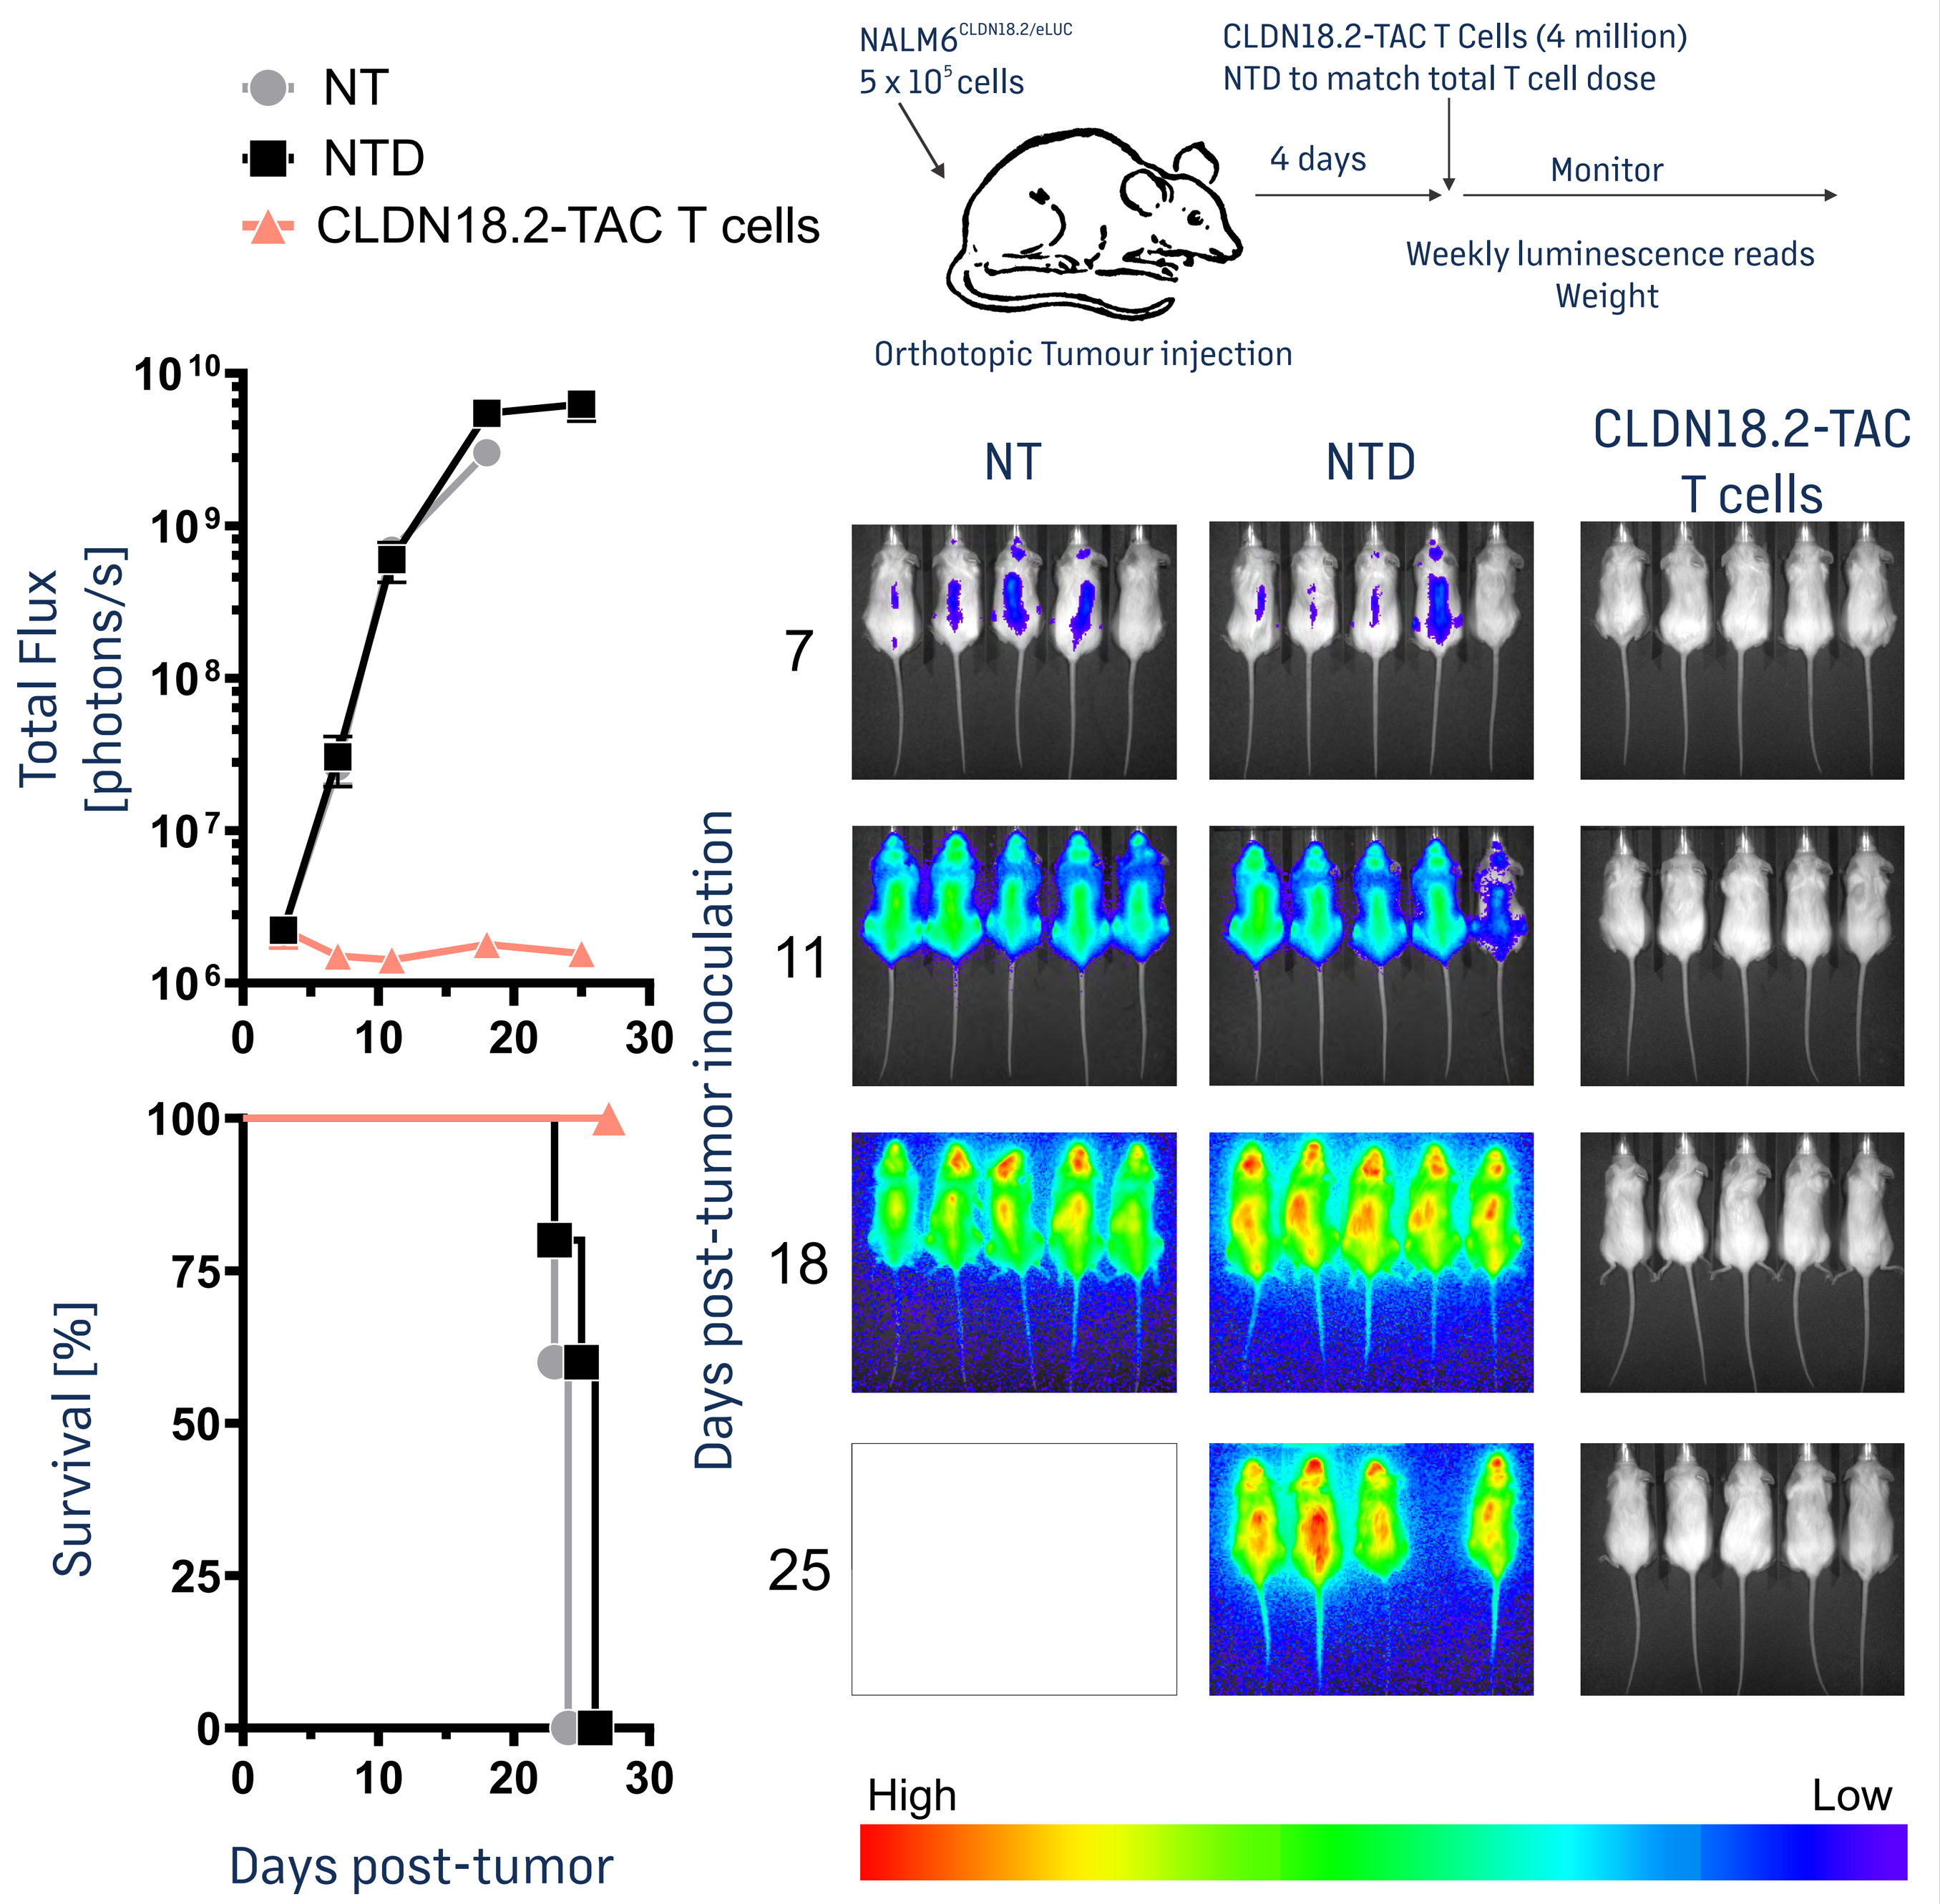
NT
NTD
CLDN18.2-TAC T cells
NALM6CLDN18.2/eLUC
5 x 10^5 cells
CLDN18.2-TAC T Cells (4 million)
NTD to match total T cell dose
4 days
Monitor
Weekly luminescence reads
Weight
Orthotopic Tumour injection
Total Flux [photons/s]
Survival [%]
Days post-tumor
Days post-tumor inoculation
NT
NTD
CLDN18.2-TAC T cells
7
11
18
25
High
Low

Summary

- **Activation of CLDN18.2-TAC T cells is specific to target cells with endogenous (KATO III) and ectopic Claudin 18.2 expression (NALM6$^{CLDN18.2/eGFP}$)**
- **CLDN18.2-TAC T cells do not show signs of auto-activation or elevated exhaustion markers post manufacturing**
- **CLDN18.2-TAC T cells effectively eradicate Claudin 18.2-expressing tumor cells in vitro and in vivo**